The Healing Power of Nature

The Use of Fragmented Intact Fish Skin in Patients Promoting Closure in Difficult to Heal Wounds in Cancer Patients

Arjun Joshi, Alyssa Shrestha, Shyamali Singhal MD PhD
Mountain View Surgery, El Camino Hospital

INTRODUCTION

Surgical wounds in patients who have been treated with chemotherapy and radiation are notoriously difficult to heal primarily and even more difficult to heal with secondary intention. In this clinical case study, we present with one TNBC patient with a non healing wound due to radiation in a heavily pretreated patient with chemotherapy and targeted therapy. She was treated with debridement and wound vac prior to placement of fragmented FSG. The FSG was prepared based on the manufacturer's instructions. Only simple secondary dressings were needed to secure the graft. We placed the FSG at weekly intervals for 4 weeks. Within 4 weeks, the wound epithelialized.

METHODS

The wound was kept clean and covered since January 2018. The following products were used:

- **Santyl** - Chemical debridement. Dime size application used daily.
- **NPWT (KCI) Black Foam** - cut to size and was applied to the wound. It was used as per the manufacturer's directions with a pressure of 125mm Hg changed twice a week.
- **Antimicrobial Wound Matrix (Puraply)** - It was cut to size placed on the wound bed, a nonstick adaptic dressing was placed.
- **NPWT** - was placed over the wound matrix,
- **FSG (Kerecis)** - The slurry was made and the wound covered. Dressings were applied as per manufacturer's guidelines.
- **Xeroform Dressing** - placed on to cover the wound and changed daily.

CASE

A 69-year-old female who presented:

Left breast cancer - diagnosed 2009, triple negative, involving pectoralis major (T4N2M0)

- Neoadjuvant Taxotere x 3, FEC x4
- Left modified radical mastectomy residual malignancy 2 cm, close margin at sternal border, refused XRT
- Surveillance until 2013

New Right breast cancer and ulcer - presented on 1/2018 at the left sternal border. 2 x2 cm

- Gemcitabine/Cisplatinum finished 7/18, wound care xeroform dressing

1/2019 - presented with enlarging sternal border ulcer

- Abraxane and Azetulimab

9/2019 - Right mastectomy, excision of left sternal border lesion with primary closure. Patient refused more extensive surgery. Left sternal border lesion margins positive for tumor.

Radiation to chest wall, supraclav and boost to the sternal border.

Risk Factors for Poor Wound Healing:

- Cancer

- Poor nutrition

- Steroid usage for premedication during chemotherapy

- **Pertinent positives:** infection, age, medications, nutrition, radiation with a boost to the ulcer bed
- **Pertinent negatives**: no smoking

WOUND HISTORY

She presented to the wound clinic on 4/21 with an enlarging wound at the left sternal border.

She was treated with local debridement and Santyl as she refused NWPT.

- **7/16/21** - Biopsy of bone and tissue showed no evidence of malignancy.
- **8/27/21** - NPWT therapy initiated, over several months including a trial of Puraply. The wound had granulation tissue.
- **2/25/22** - Granulation tissue healthy.
- **3/3/22** - Kerecis Fish was applied for 3 weeks.
- **6/3/22** - Last day of wound care treatment.

RESULTS

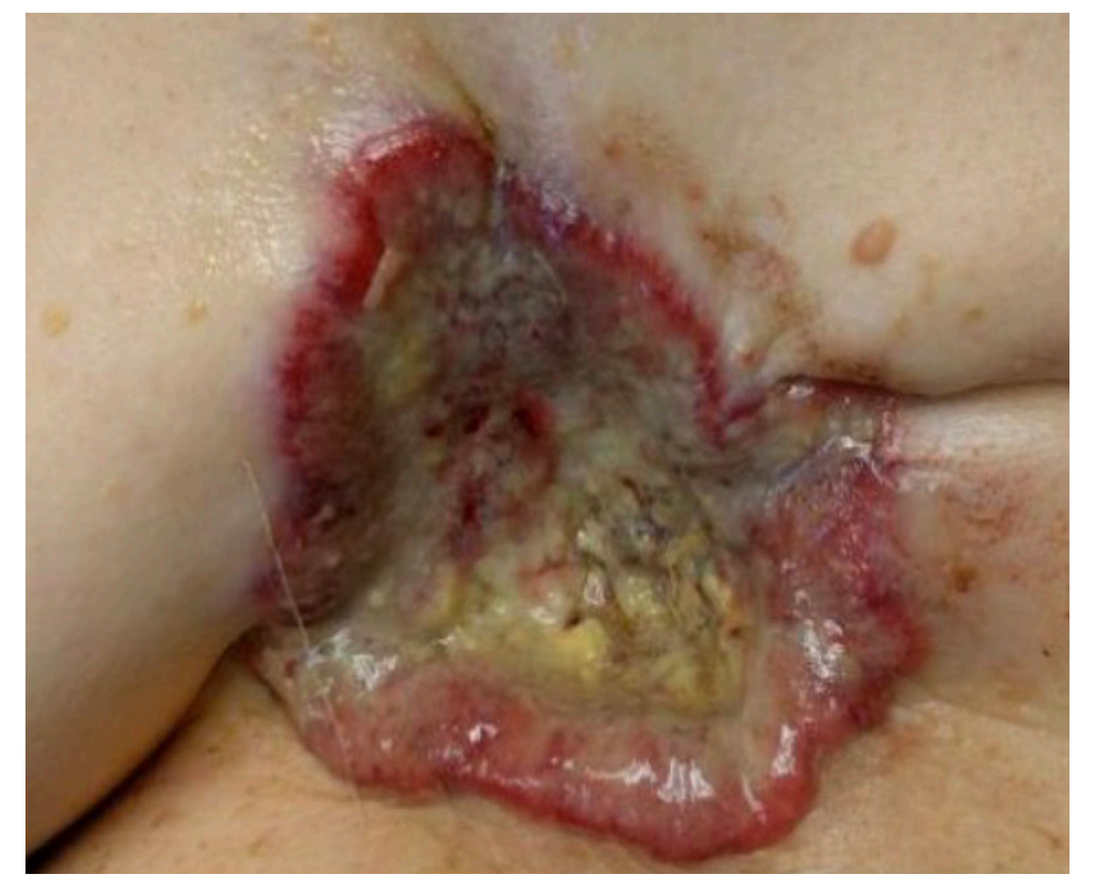

2/2/2018 - Patient presents with a 6x6 cm wound at the left sternal border.

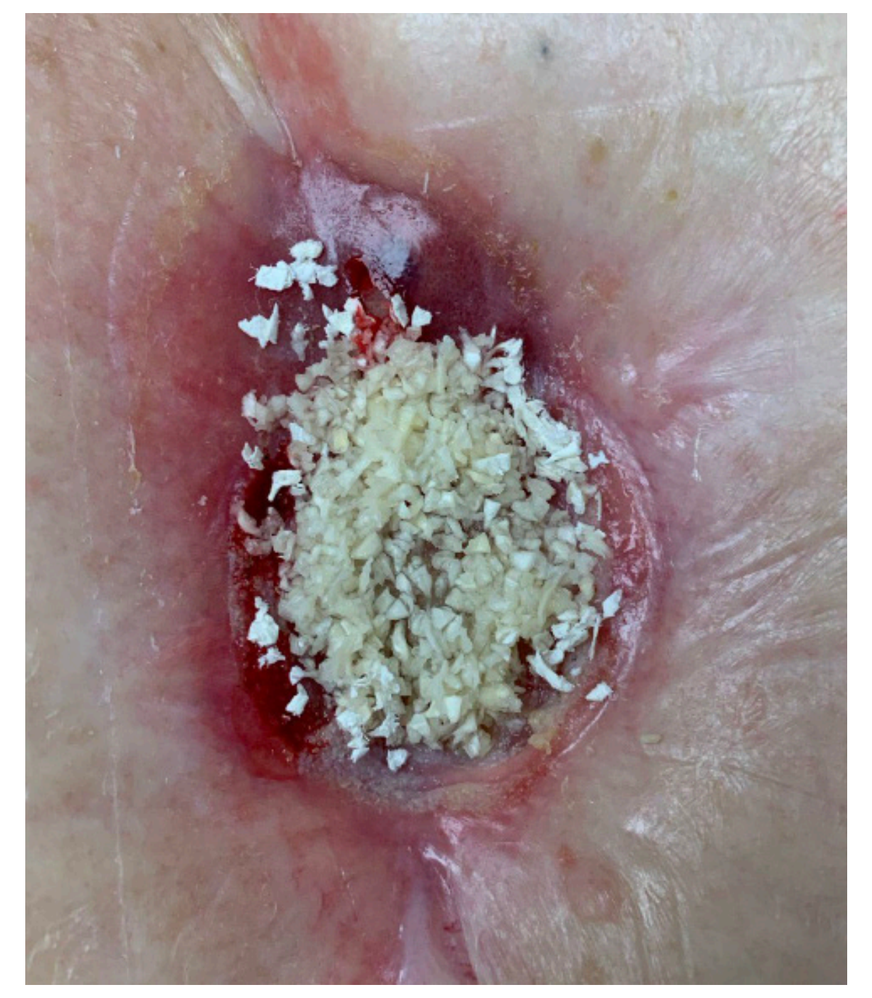

3/3/2022

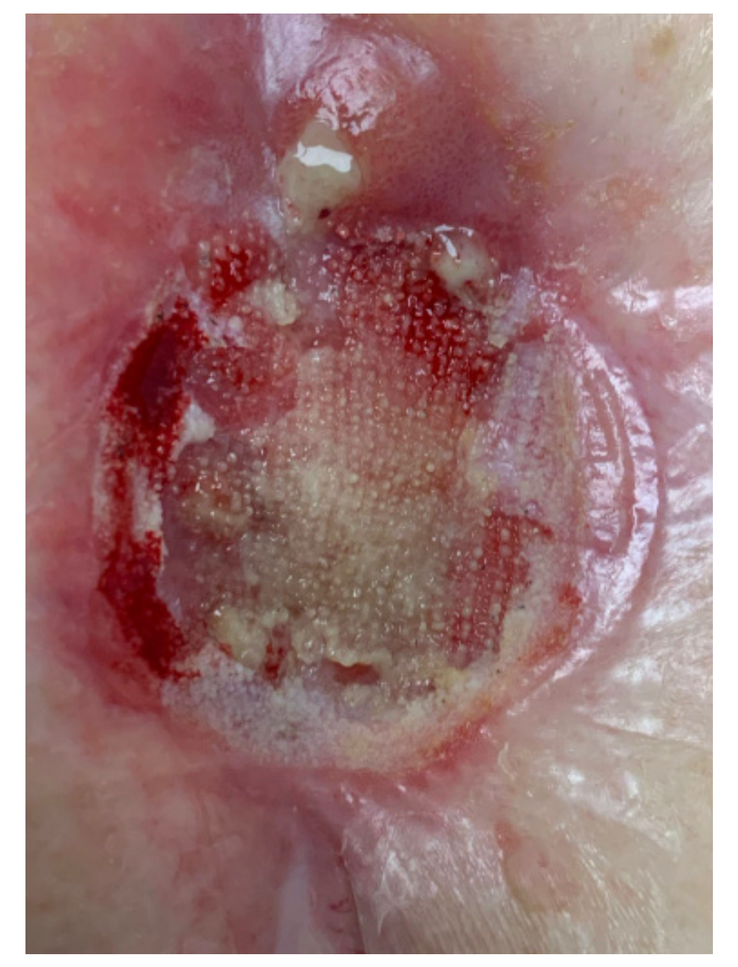

3/10/2022

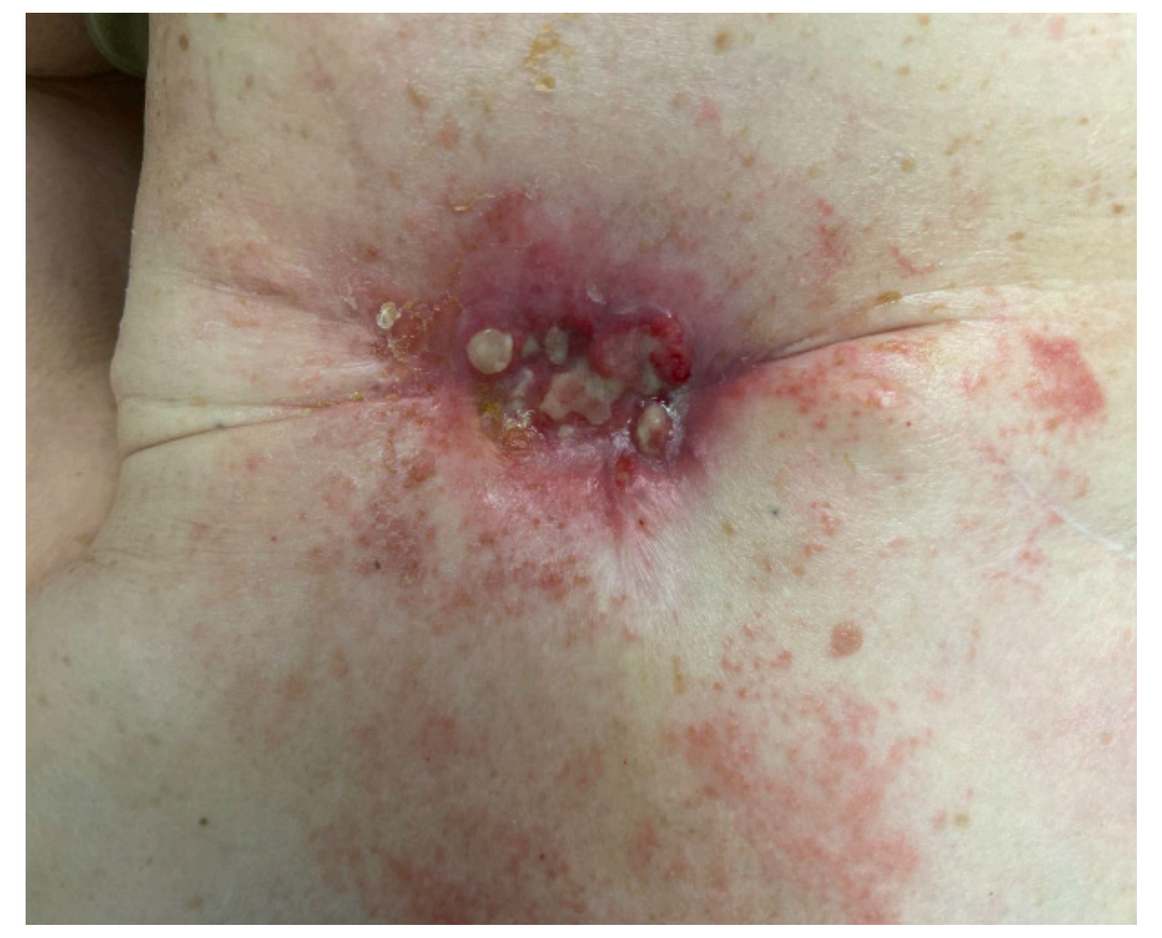

6/3/2022 - Wound is mostly closed.

CONCLUSION

69 y female with TNBC locally advanced with bilateral breast cancer. In the area of the left sternal border, the residual malignancy had eroded to the bone. She had refused chest wall resection. Attempt at local resection left sternal border was made with primary closure and there was residual disease and positive margins. She underwent radiation including a boost and continued chemotherapy. Her presentation to the wound center showed breakdown of the left medial edge of the sternum in radiated tissue. Biopsy conducted on 7/22 showed no evidence of malignancy. Wound care was initiated with NPWT for almost 1 year followed by puraply/NPWT. Once granulation tissue was evident, then FSG was applied with excellent epithelialization. In a wound that was not expected to heal, it has healed with secondary intention. Our patient had many risk factors for poor wound healing as mentioned. A combination of NPWT and wound matrix developed a bed of granulation tissue. Once the bed of granulation tissue was present, the FSG was applied with excellent results. The FSG is one of the newer xenograft materials that promote wound healing. In direct comparisons to other amniotic membranes, FSG leads to accelerated closure of the wound. Our case study demonstrates that a combination of NPWT, wound matrix and FSG can lead to epithelialization of a complex wound.