The Healing Power of Nature

The use of fish skin graft* in patients with periosteum/bone exposed post scalp lesion removal for definitive wound closure

Courtney Aldridge PA-C and Ryan P. O'Quinn, MD
South Texas Skin Cancer Center, San Antonio, TX

INTRODUCTION

Exposure of the periosteum and calvarium during malignant lesion excision of the scalp can limit reconstruction options for successful wound closure. Wounds with exposed periosteum/bone typically require that patients be taken to the operating room to have the bone burred to induce bleeding so a split thickness skin graft can be applied. A hallmark of fragmented intact fish skin grafts (FSG) is their ability to fill irregular tissue defects after surgical procedures or trauma. FSG can be helpful for healing avascular surfaces (such as exposed bone). FSG can be used to fill these lesions where periosteum/bone is exposed and accomplish complete healing by secondary intention, eliminating the need for operating room procedures.

METHODS

In this clinical case study, two patients presented with periosteum/bone exposure following skin cancer excision using Mohs procedure. Traditional treatment would have warranted surgical reconstruction by plastic surgical techniques (such as split thickness skin grafting) as both patients had minimal tissue mobility. Fragmented FSG was prepared based on the manufacturer's instructions. Only simple secondary wound dressings were needed to secure the graft.

.

*Kerecis™, Kerecis, Isafjordur, Iceland

CASE # 1:

Patient History: 92 year old frail female with dementia and anemia

Wound History: Acute surgical wound following Mohs surgical excision of squamous cell carcinoma (SCC) resulting in a 5.0 x 4.5 cm wound with exposed bone (calvarium)

Kerecis Applications: 4 Applications of fragmented fish skin graft

Patient Outcomes: Complete healing with no complications and excellent cosmetic outcome with a 1.7 x 1.5 cm flat flush scar

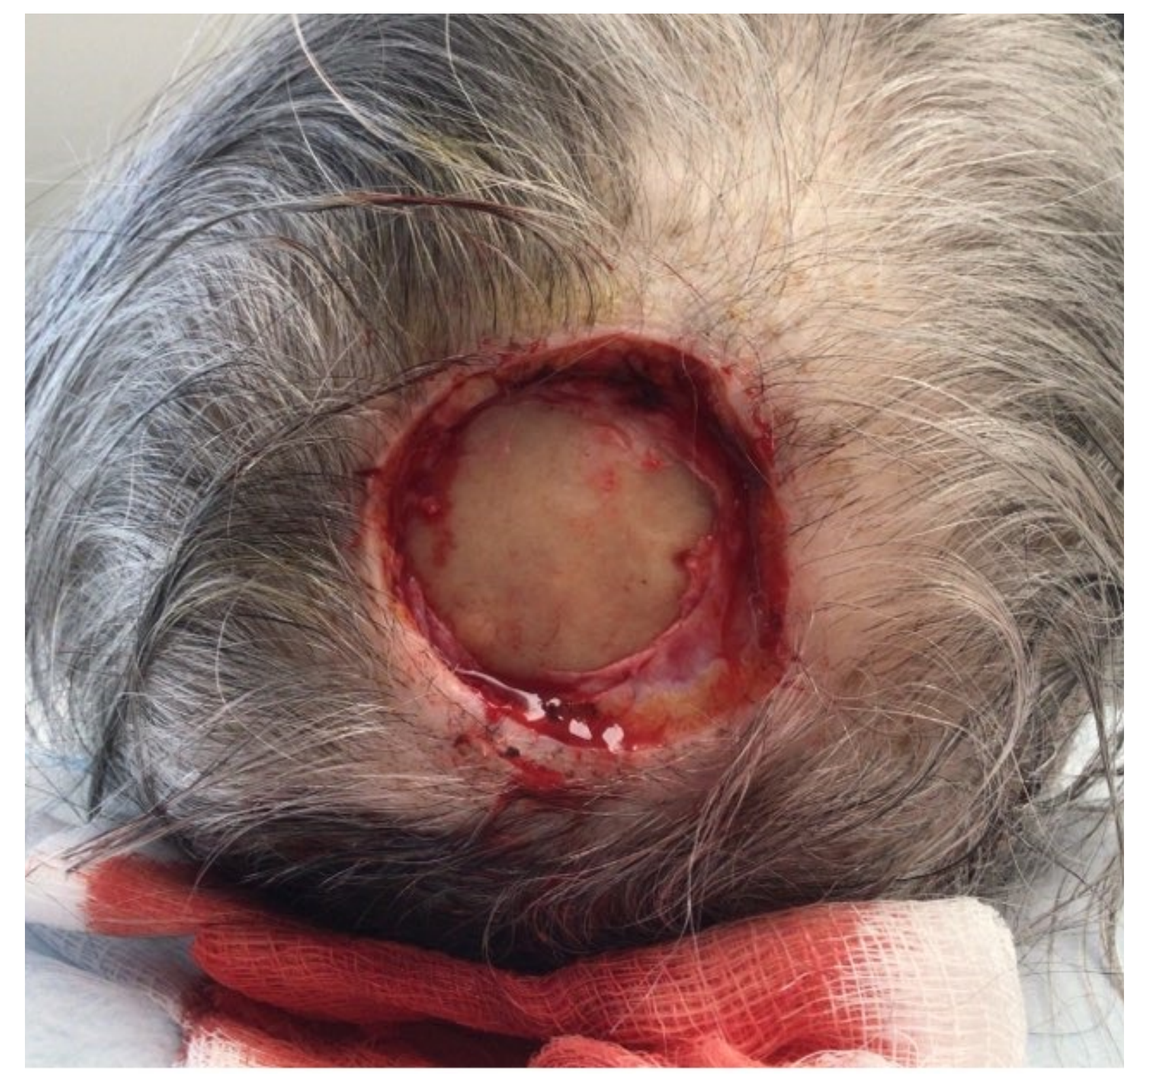

5.0 x 4.5 cm wound to the level of the bone (calvarium) following Mohs surgical excision of a squamous cell carcinoma (SCC)

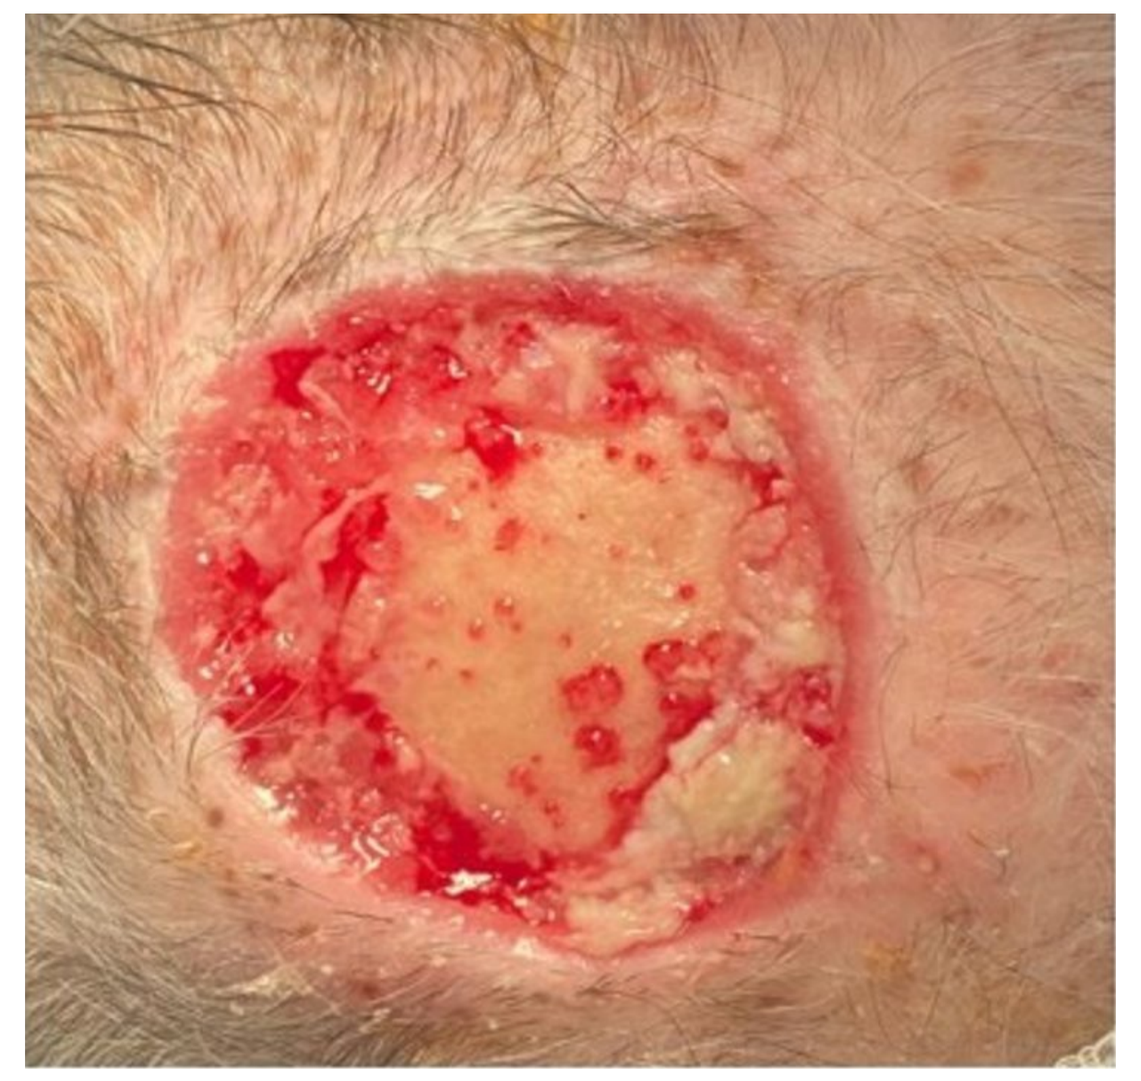

Islands of granulation tissue budding from calvarium with minimal inflammation

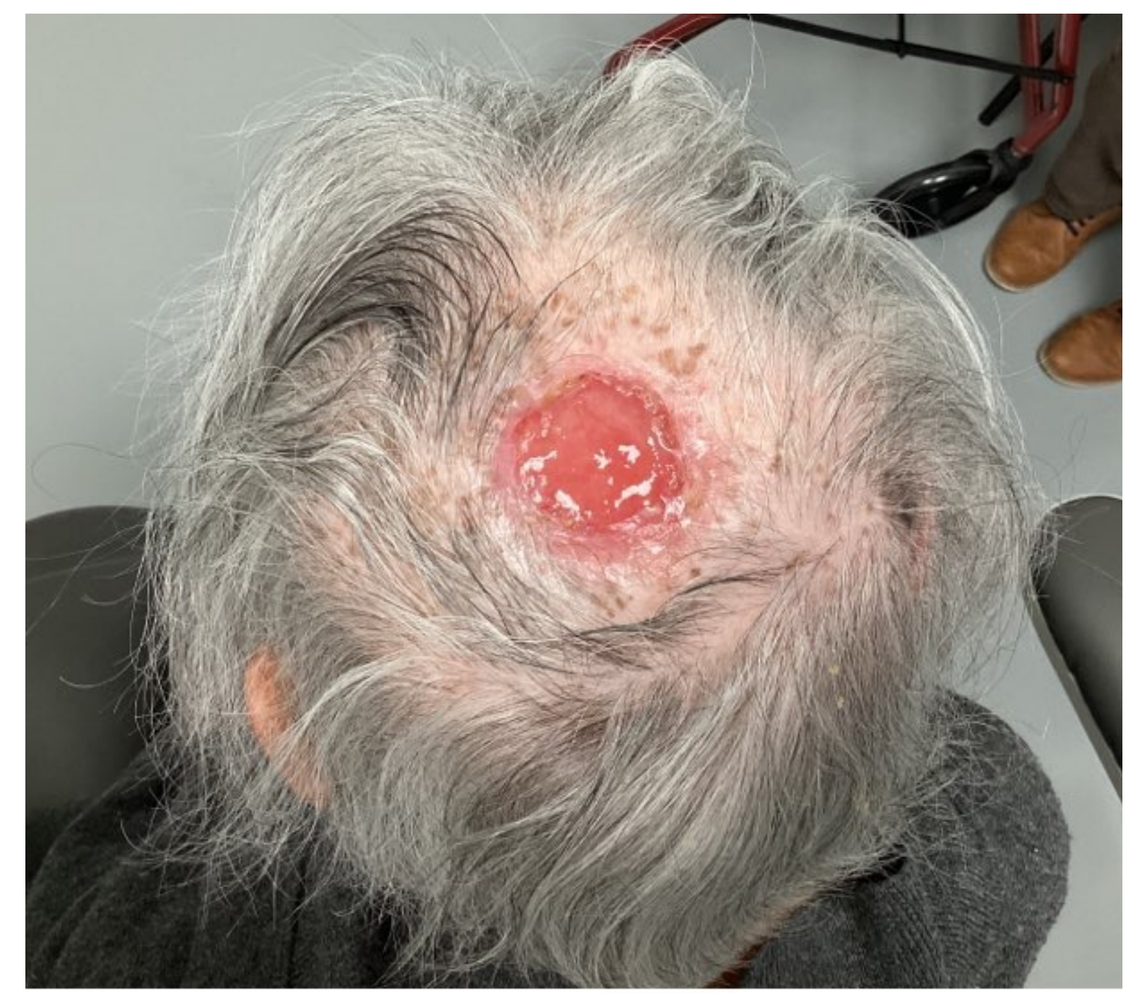

Complete coverage of calvarium with robust granulation tissue

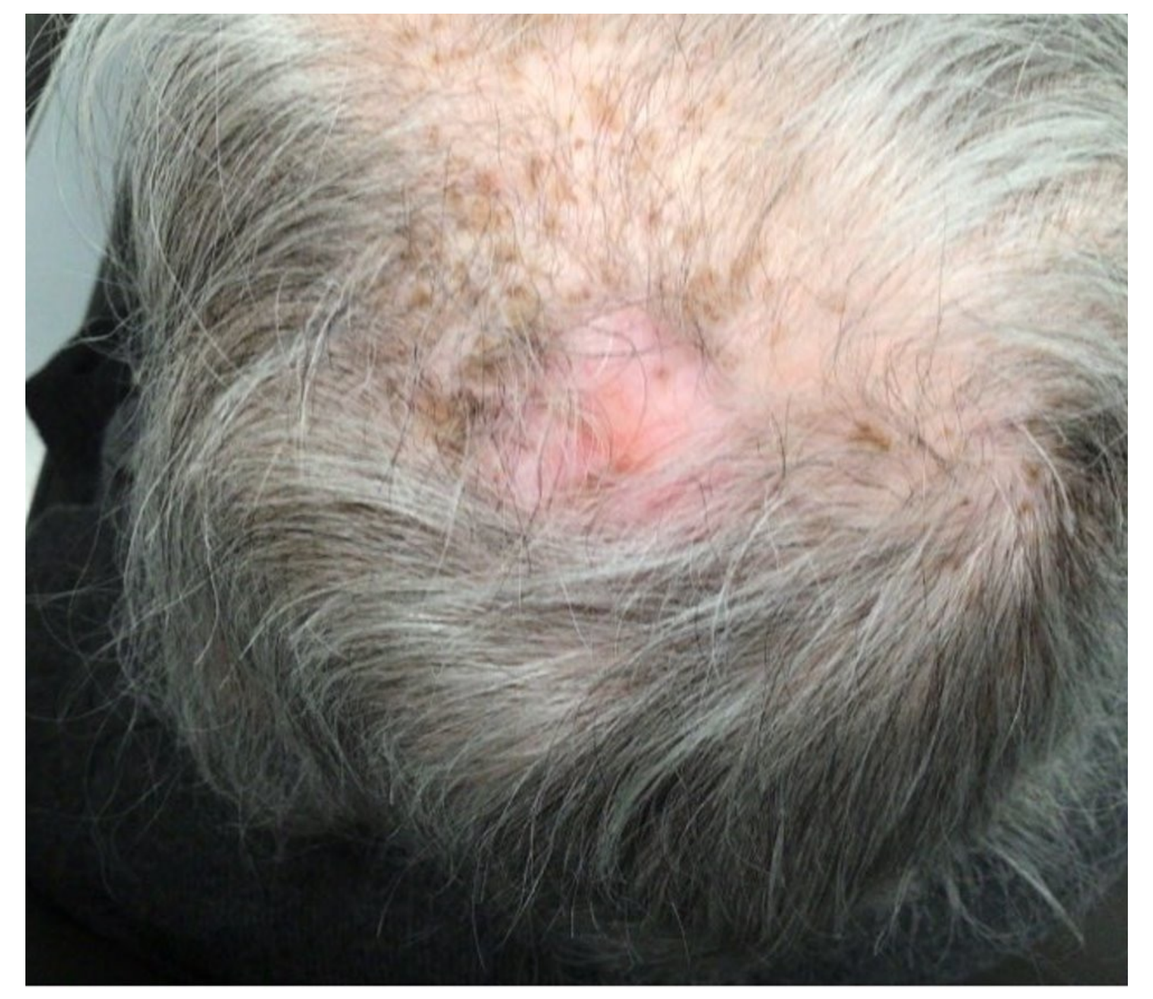

Final appearance of healed wound measuring 1.7 x 1.5 cm following 4 applications of fish skin graft (FSG)

CASE # 1:

Patient History: 85 year old male with poor nutrition, hypertension and coronary artery disease

Wound History: Acute surgical wound following Mohs surgical excision of SCC measuring 9.0 x 7.5 cm wound with exposed periosteum and bone (calvarium)

Kerecis Applications: 9 Applications of fragmented and sheet fish skin graft

Patient Outcomes: Complete coverage of the periosteum and calvarium with healthy granulation tissue with partial closure of the wound to 6.1 x 6.4 cm. The wound was then repaired with a full-thickness skin graft

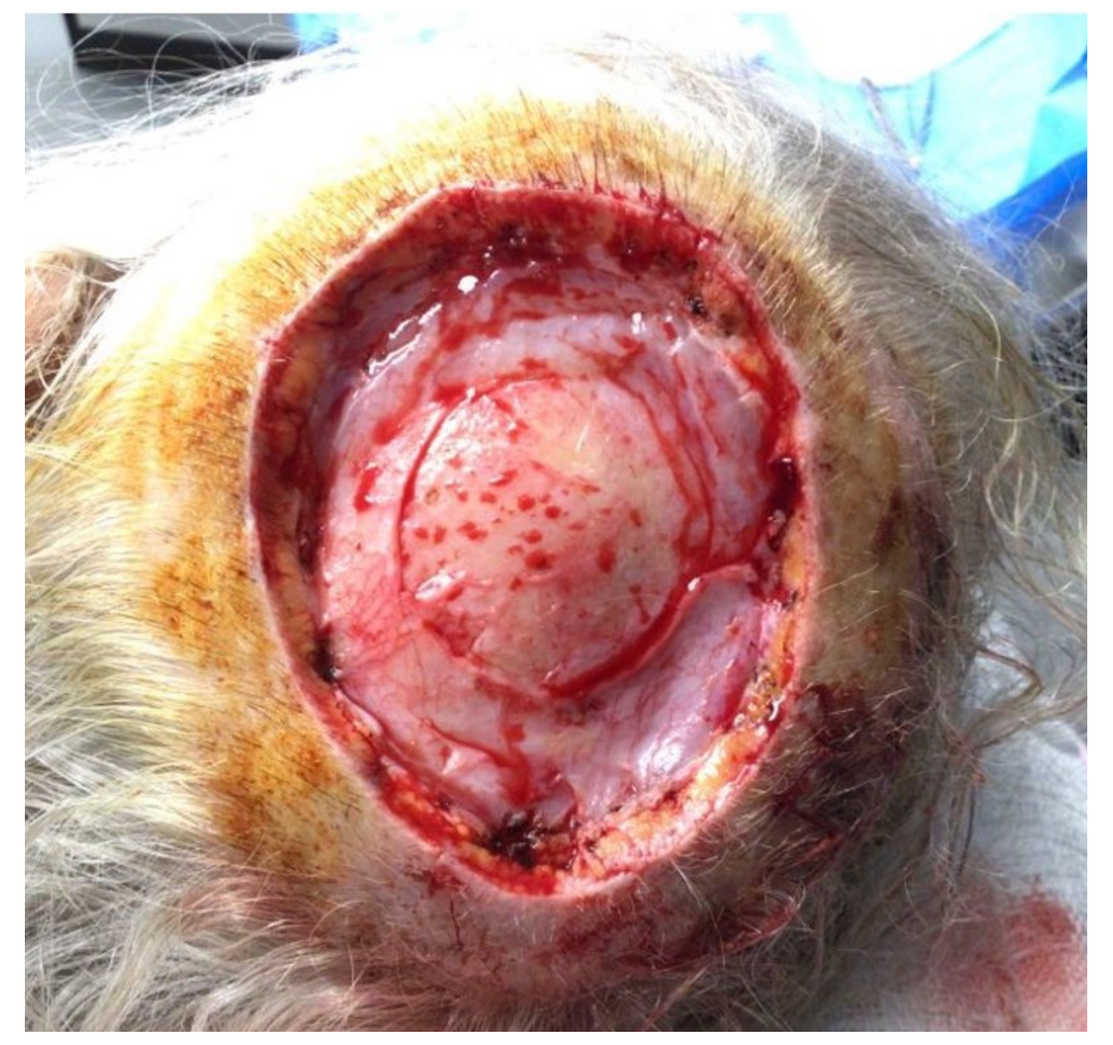

9.0 x 7.5 cm wound to the level of the periosteum (laterally) and bone of the calvarium centrally following Mohs surgical excision of a squamous cell carcinoma (SCC)

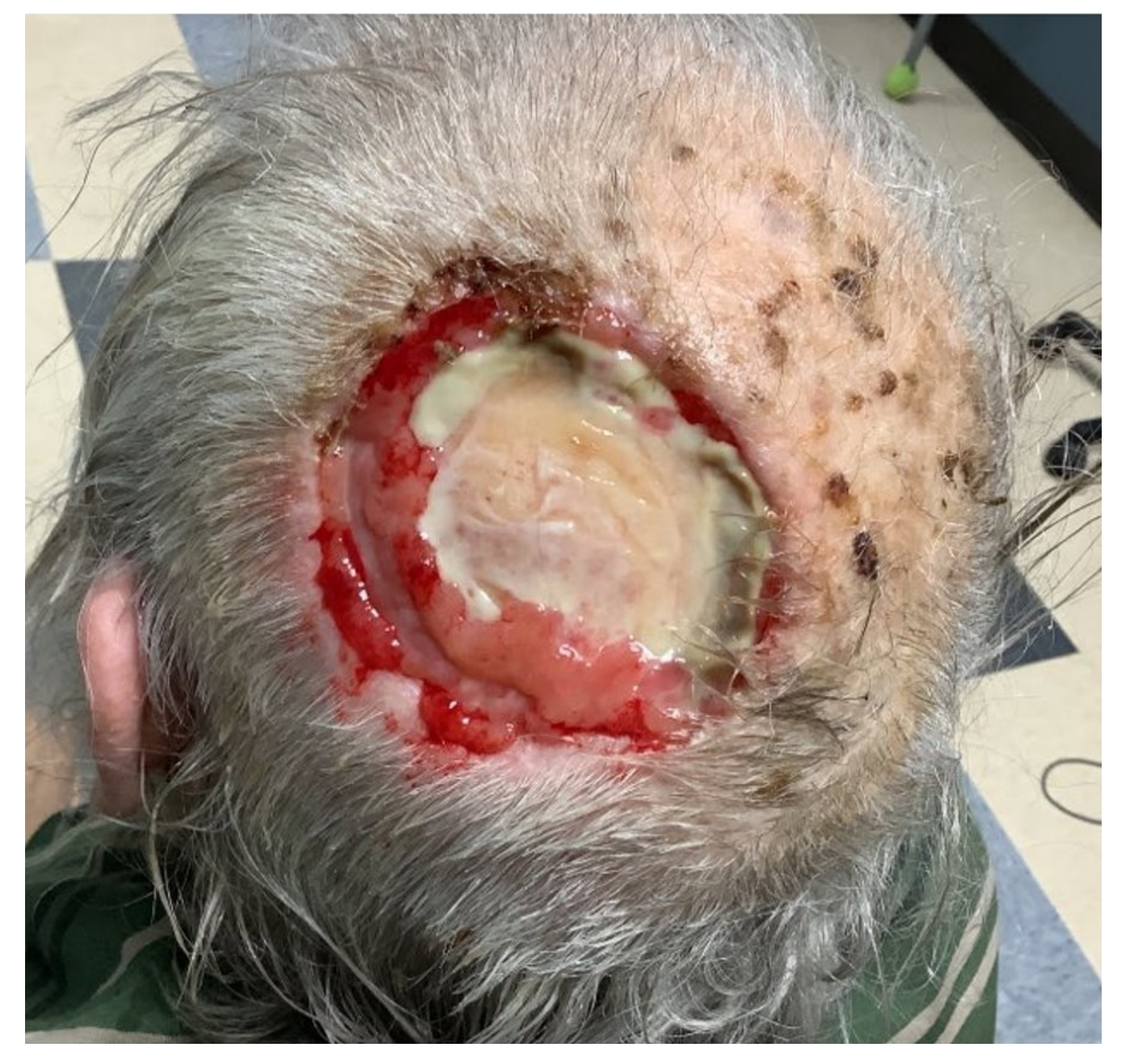

Wound contraction and granulation tissue formation following 4 applications of fragmented fish skin graft (FSG)

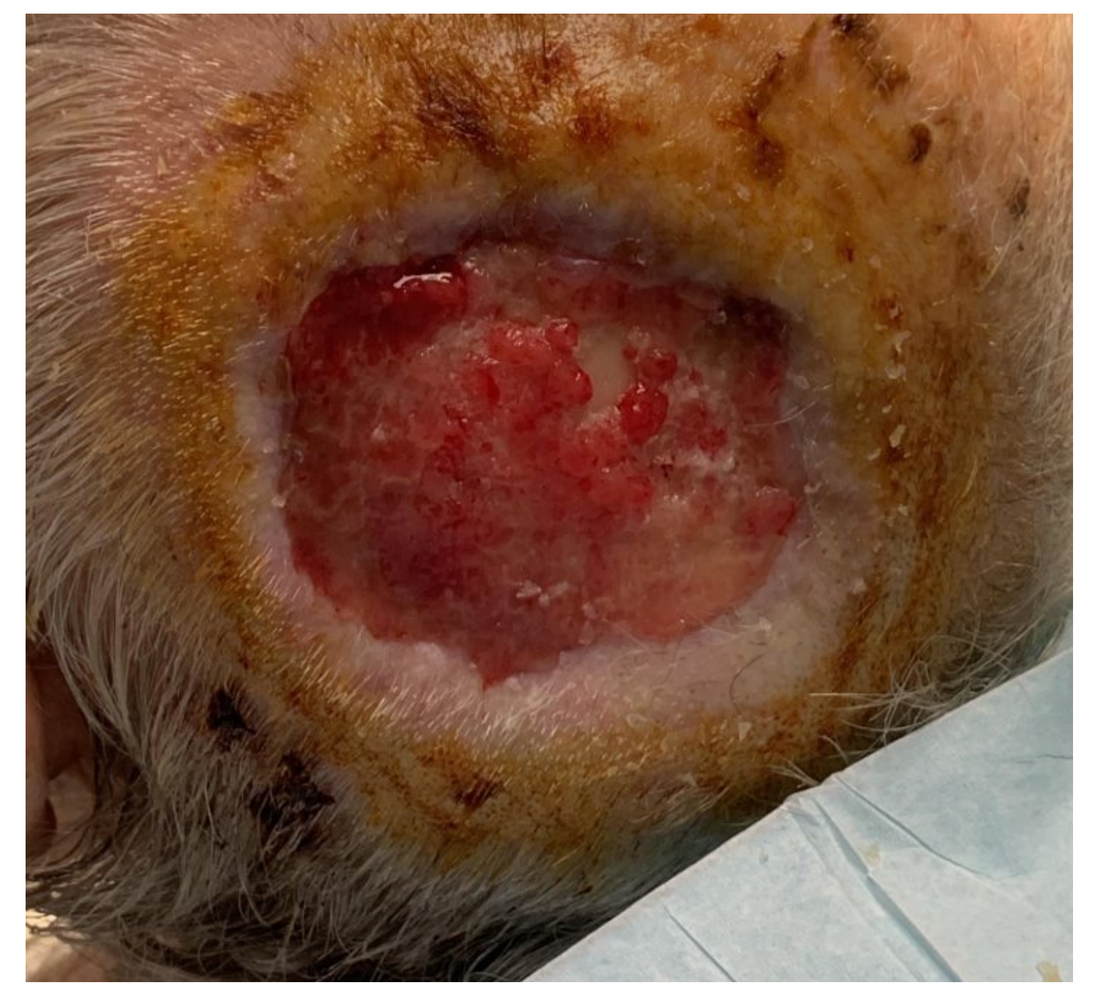

Near complete coverage of the bone of calvarium after 9 applications of fish skin graft (FSG). At this point the wound was covered with a full-thickness skin graft

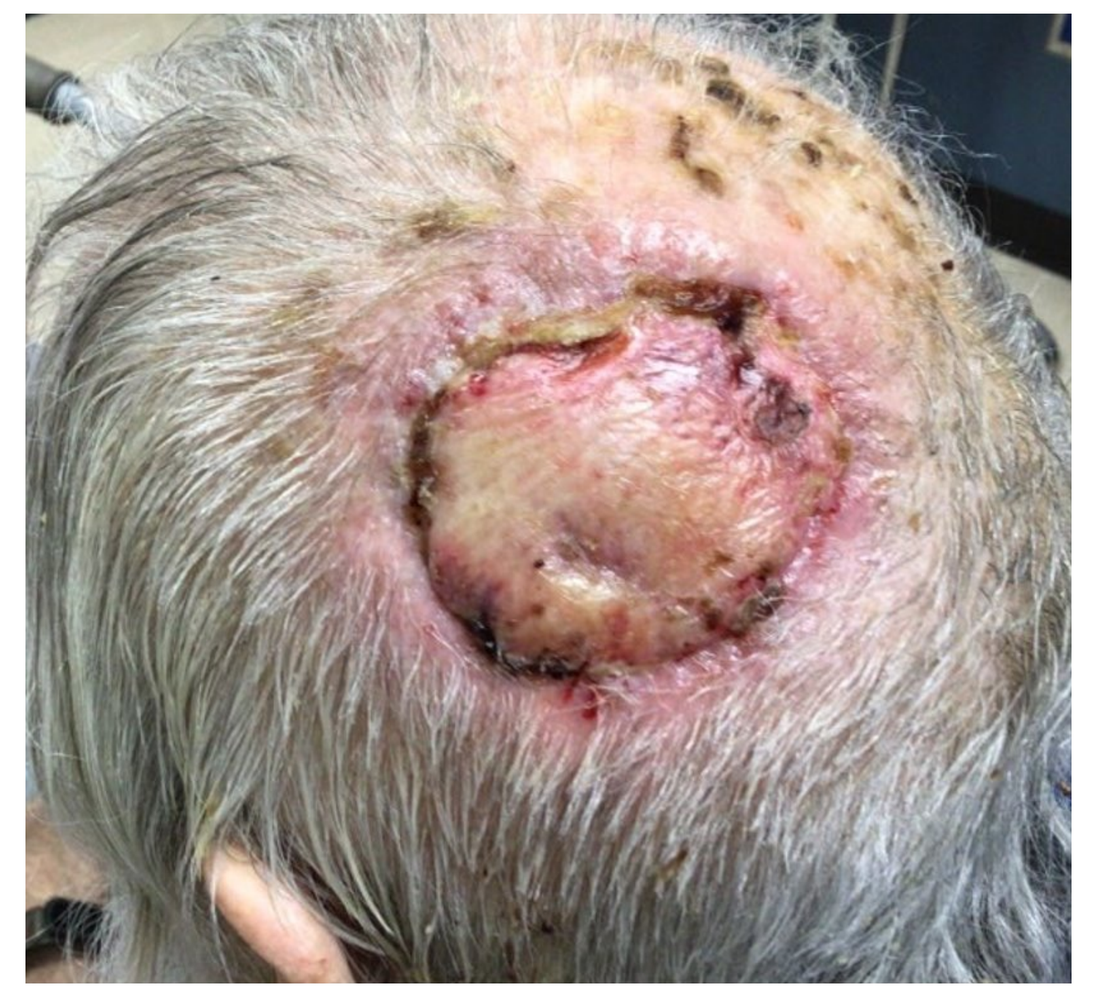

1 week s/p full-thickness skin graft application of the granulated wound

KERECIS APPLICATIONS

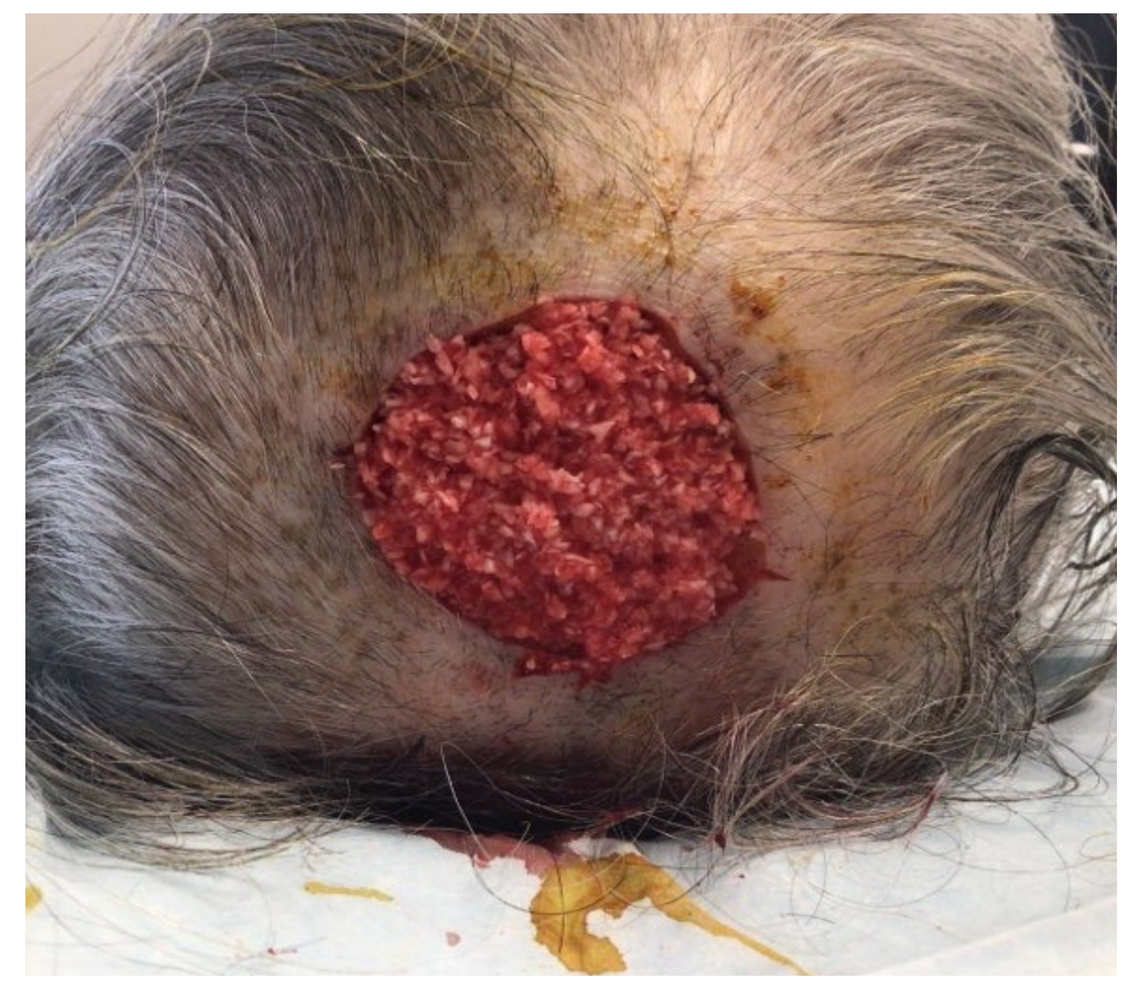

Case 1: Application of fragmented fish skin graft (FSG) saturated with wound blood

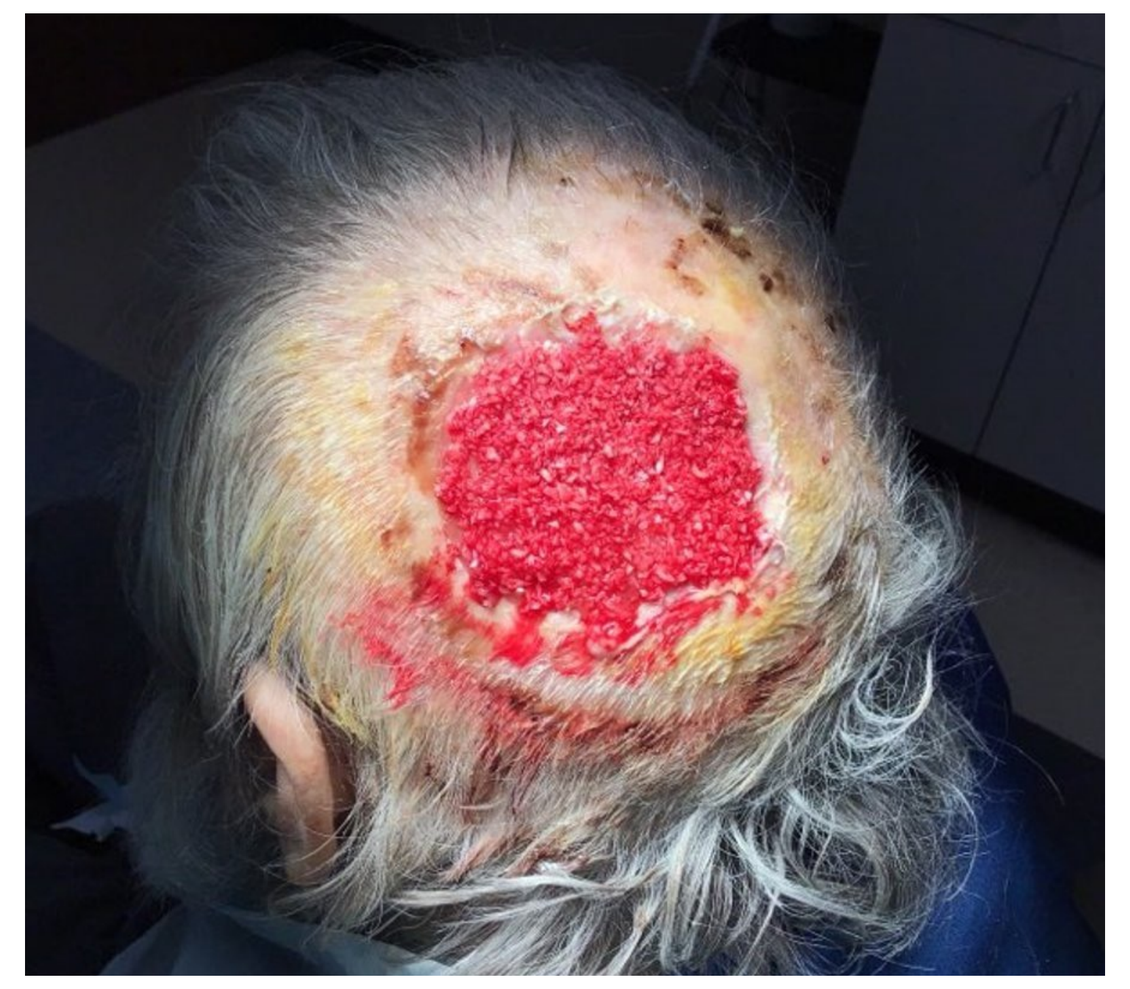

Case 2: Application of fragmented fish skin graft (FSG) saturated with wound blood

RESULTS

Complete coverage of periosteum/bone with granulation tissue was achieved in both patients. One patient was allowed to heal by secondary intention following FSG applications, and one patient was repaired using a full thickness skin graft once periosteum was covered with granulation tissue. Granulation tissue was observed after the first application of FSG, and islands of granulation tissue were noted on both patients growing directly from the calvarium. No infection or pain was noted in either patient. The patient allowed to heal with secondary intent required five applications of FSG and healed in four months. The patient healed using a full thickness skin graft required nine applications of FSG and healed in three months.

DISCUSSION

Scalp wounds with exposed periosteum and bone are challenging to heal. These wounds can be especially difficult to manage in patients that are not surgical candidates due to underlying co-morbidities. The use of fragmented FSGs has shown to promote wound healing in these challenging environments and warrants further investigations as a skin substitute in post-excisional dermatologic wounds.