EVALUATION OF THE EFFICACY AND SAFETY OF A TWO LAYER DUAL COMPRESSION SYSTEM (DCS)* BANDAGE SYSTEM IN THE TREATMENT OF LOWER LIMB LYMPHEDEMA

DEBASHISH CHAKRAVARTHY[1], PH.D, **SERGE BOHBOT**[2], MD

[1]URGO MEDICAL, NORTH AMERICA, [2]URGO MEDICAL, FRANCE.

INTRODUCTION

Primary lymphedema is a chronic illness due to a lymphatic system dysfunction. Lymph fluids collects abnormally typically in lower limb subcutaneous tissues. There is no permanent cure. A combination of cutaneous care, lymphatic drainage, exercises in association with compression bandaging allows an appropriate management of this pathology. Lymphedema is found in 0.13-2% of population, and negatively impacts quality of life. A novel two-layer Dual Compression System, which combines both long and short stretch elements, and is engineered to provide therapeutic compression (20/40mm), is well known in treatment of venous insufficiency. Recently, DCS indications have been extended to lymphedema management.

METHODS

A clinical evaluation was conducted in an outpatient hospital setting, recognized for its expertise in lymphedema, to evaluate efficacy of this two-layer Dual Compression System Bandage in this pathology ; The primary outcome criterion was the volume reduction of the lower limb after treatment and an assessment of the safety of the system in lymphedematous conditions in the lower limb.

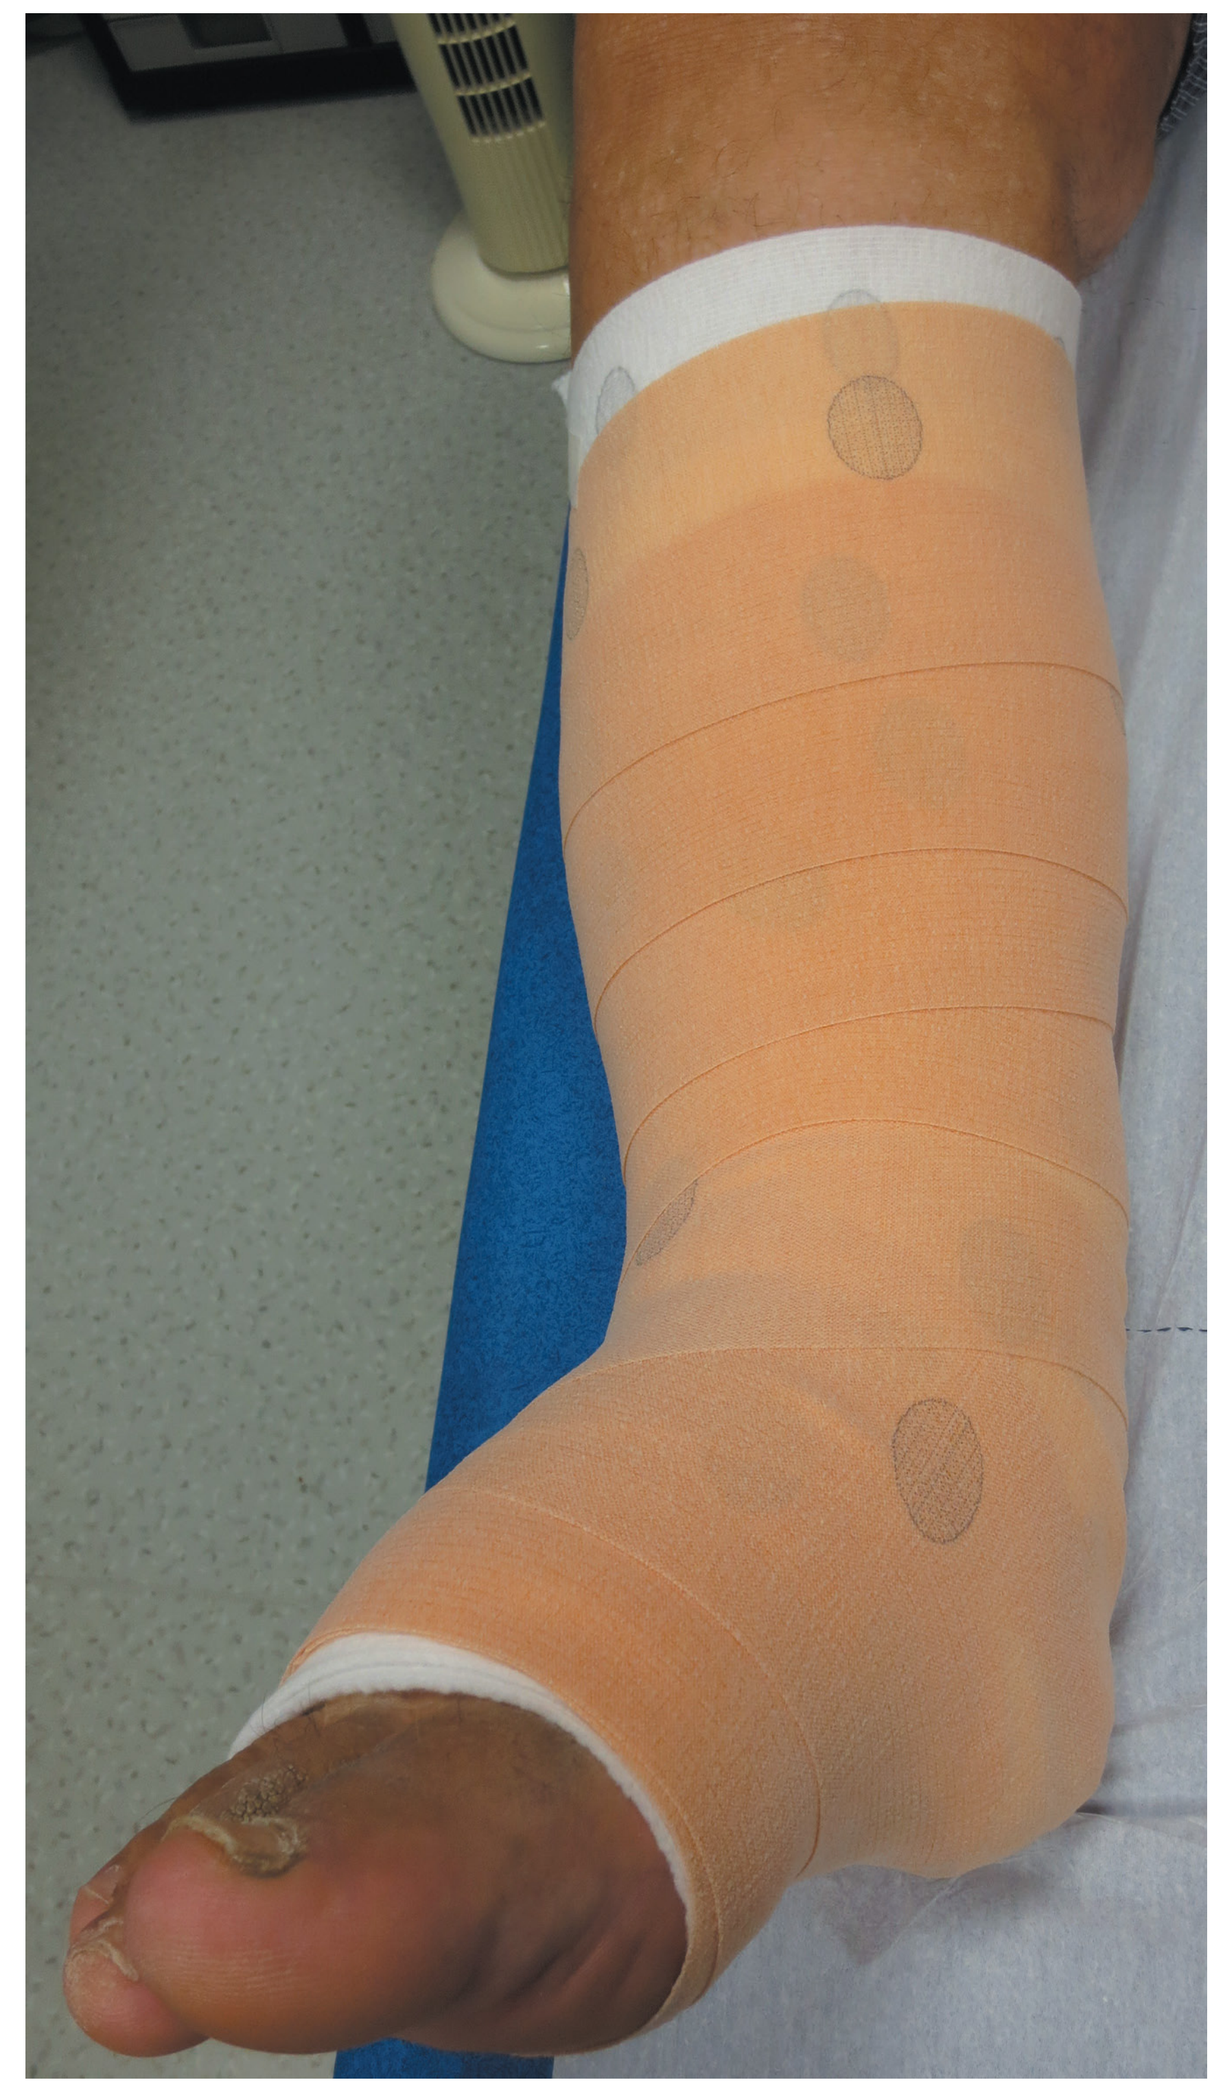

OBSERVATIONS

PATIENT 1

88-year-old female patient with an history of high blood pressure, moderate obesity (BMI=31 kg/m^2), stroke, thyroidectomy and skin basal cell carcinoma. This patient presents an advanced stage II primary lymphoedema of the left lower leg without venous insufficiency. At inclusion, the affected limb presents an erythema, a lymphangiectasy, a positive Stemmer's sign (inability to pick up a fold of skin at the base of the second toe) and the objective measurement of the amount of fluid in the limb is 13 827 ml. Therapy of this lymphoedema is a combination of manual lymphatic drainage and compression bandaging. After 31 days of treatment with the DCS, the volume of the affected limb is 11 705 ml, thus a pourcentage reduction of 15 % with a clear improvement of the skin (mild erythema compared with inclusion).

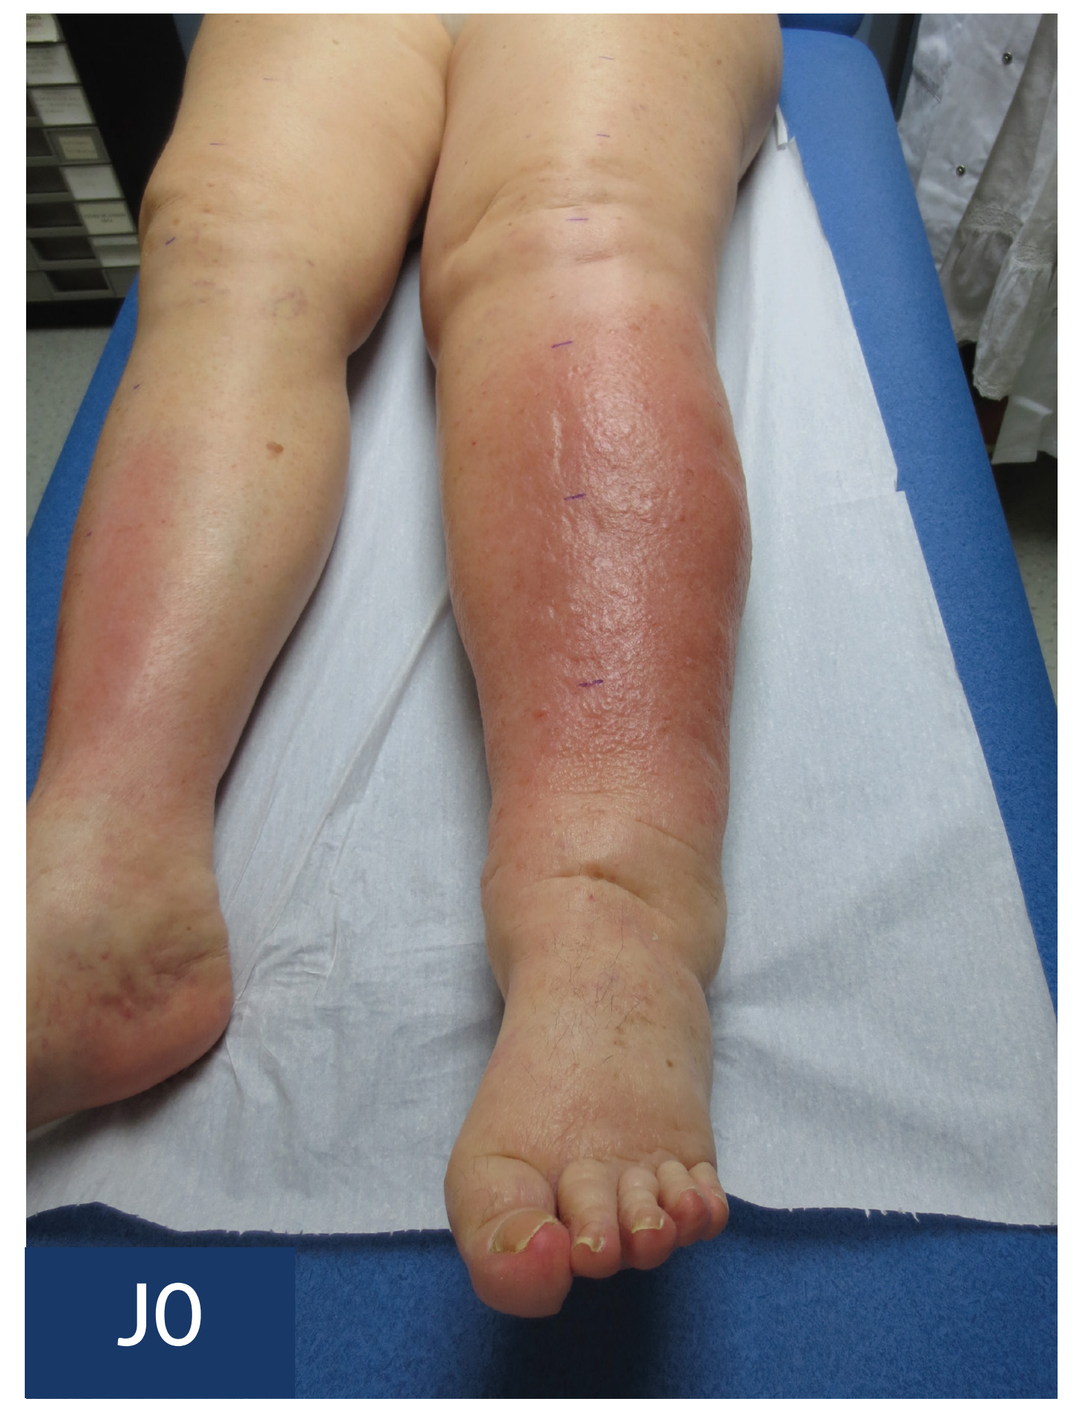
J0

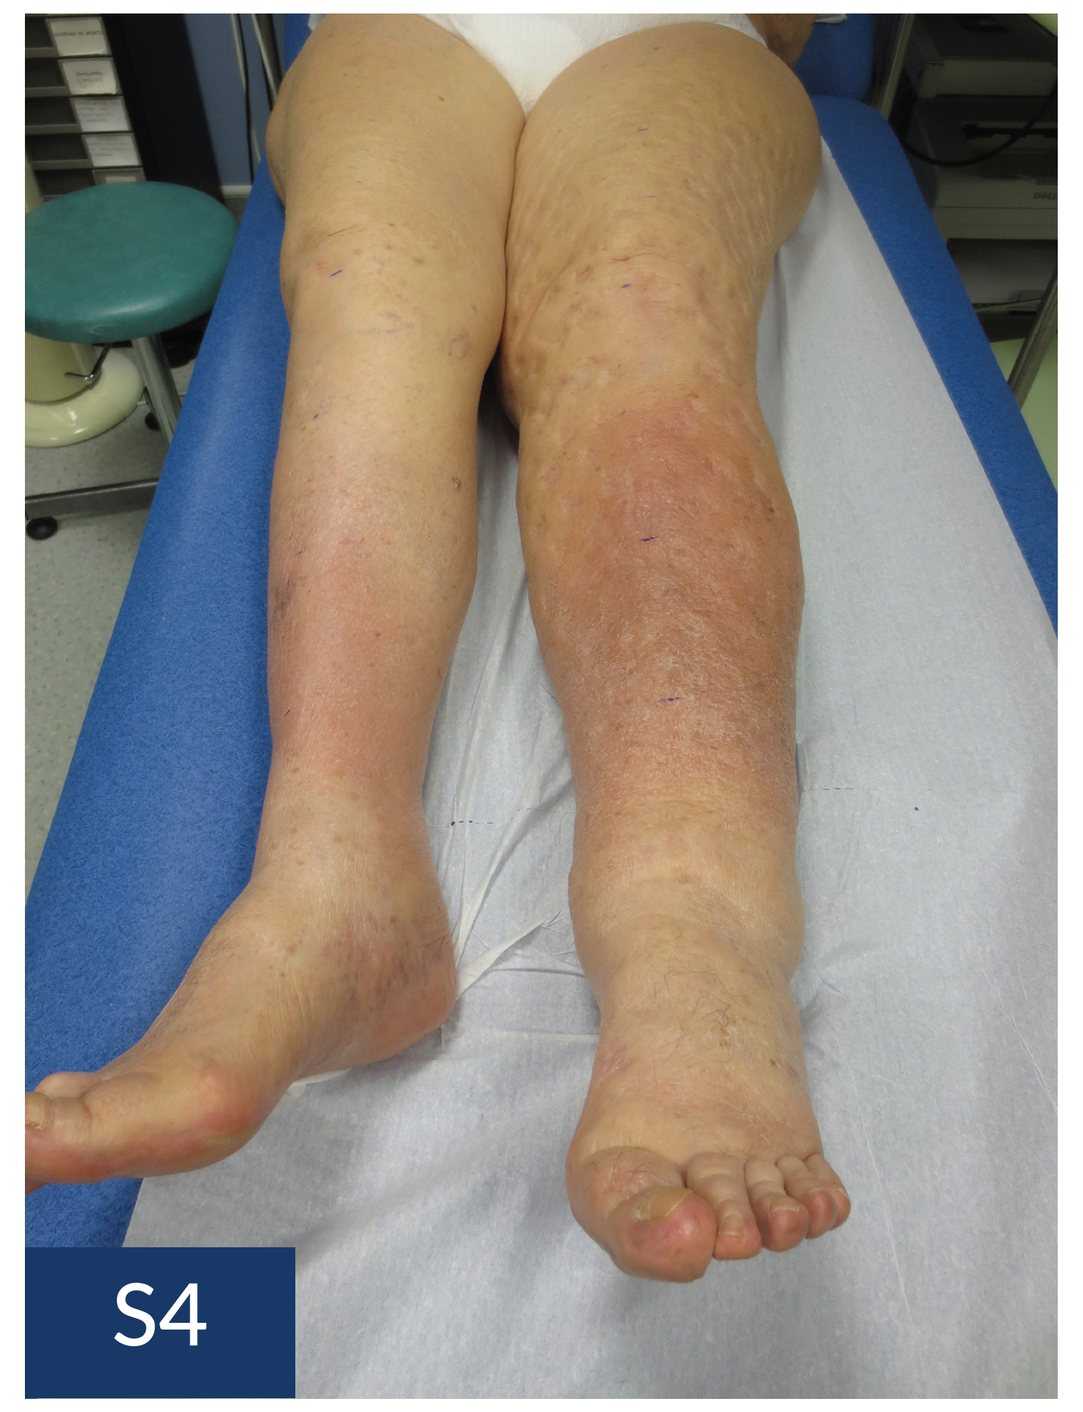
S4

PATIENT 2

69-year-old patient with an history of high blood pressure, insulin-dependent diabetes with a severe obesity (BMI > 37 kg/m^2), cardiac arrythmia (AF), erysipelas and gastric ulcer. This patient presents an advanced stage II primary lymphoedema of the right lower limb with a venous insufficiency (ABPI = 1,3). At inclusion, the leg affected by swelling presents an erythema and an eczema due to viscous lymphatic suffusions. A therapy with skin care has been associated to the compression system and the volume of the limb measured at the beginning of the treatment is 13 494 ml. After 31 days of treatment with the two-layer compression system UrgoK2, the volume of the limb has decreased to 10 990 ml, thus a reduction of 19 %. In addition, the inspection of the leg shows a significant decrease of the skin erythema and of eczematous lesions.

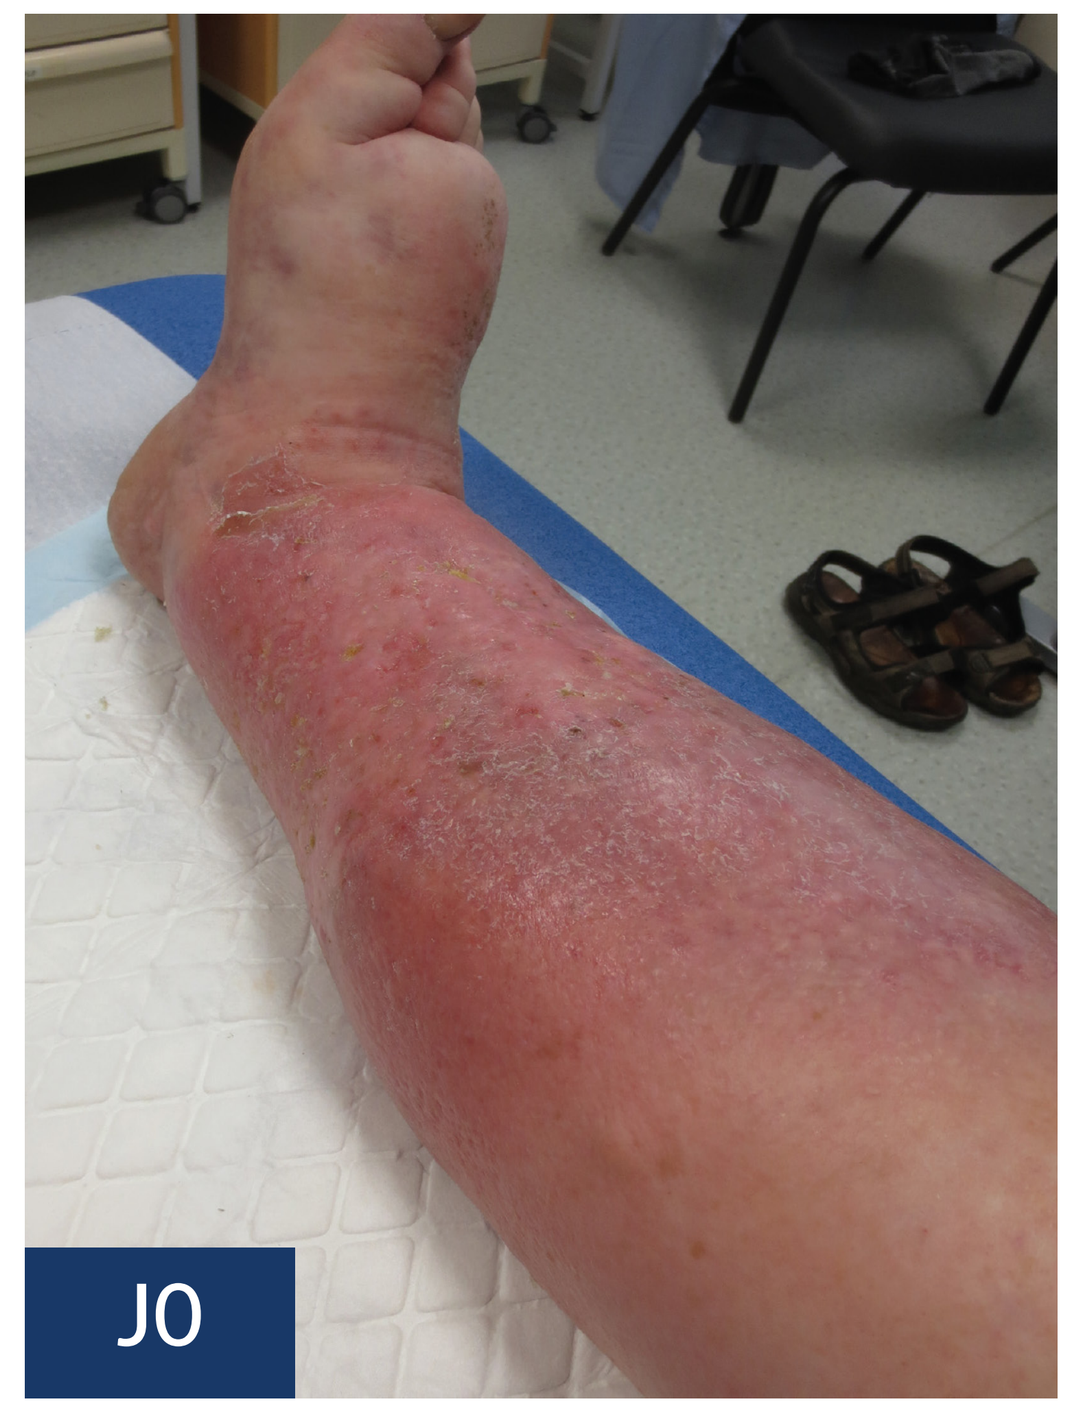
J0

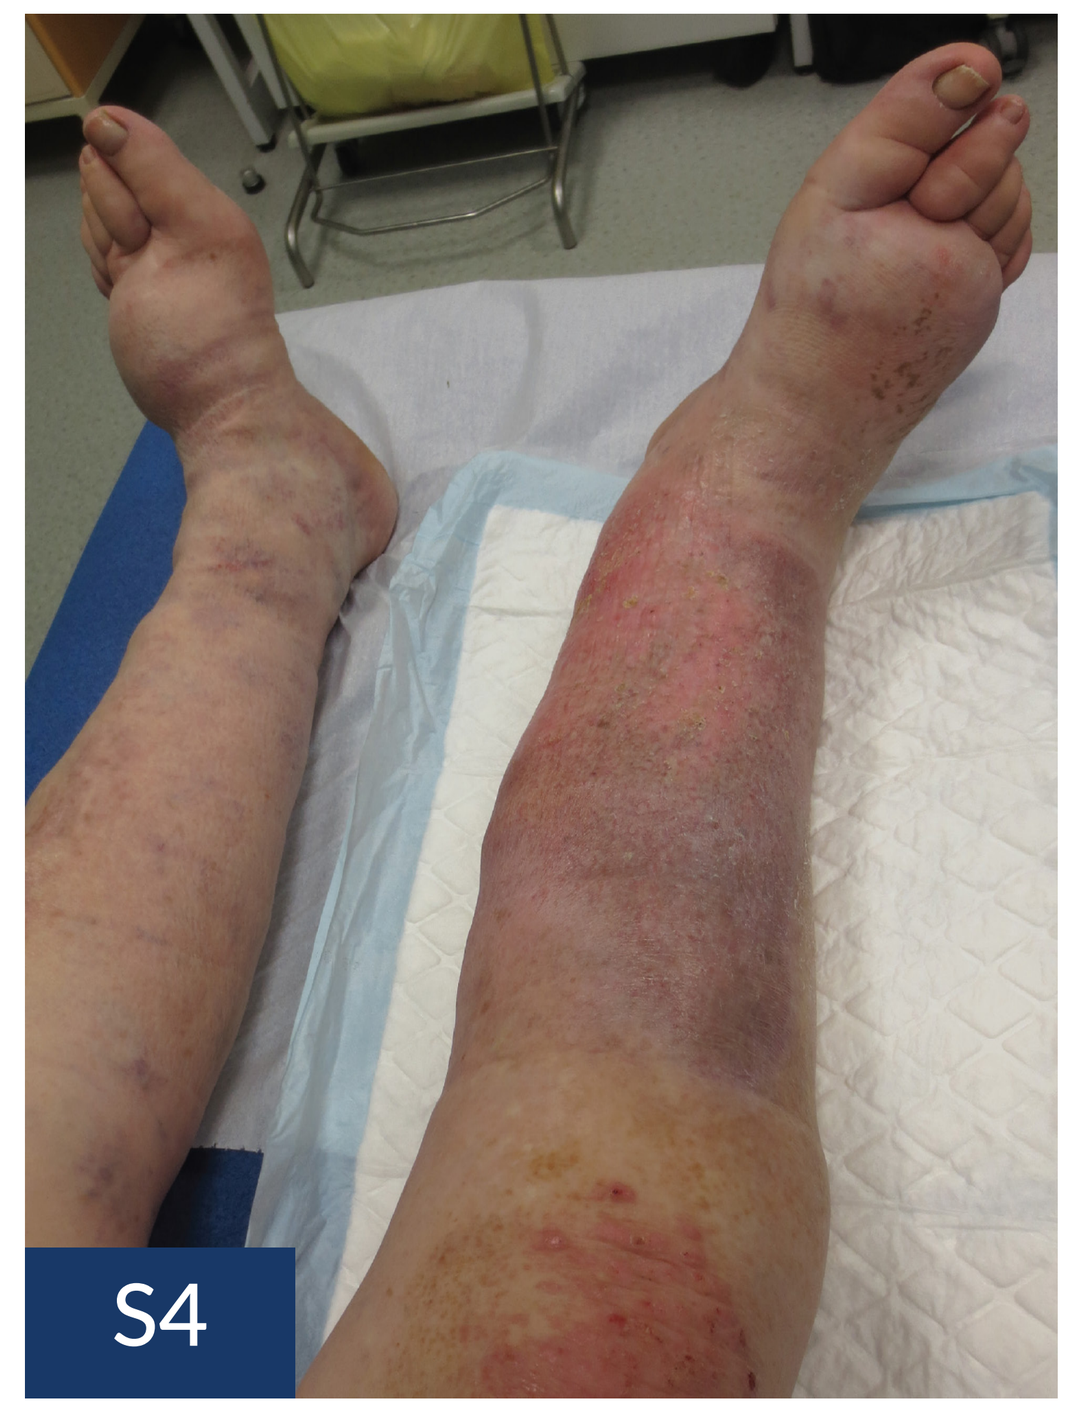
S4

PATIENT 3

73-year-old patient with an history of high blood pressure, non-insulin-dependent diabetes, morbid obesity (BMI over 48 kg/m^2) and cardiac arrythmia (AF). This patient presents an advanced stage II primary lymphoedema of the right leg and avenous insufficiency (ABPI = 0,9). At inclusion, the visual inspection shows skin lesions of eczema due to transcutaneous lymphatic suffusions and an erythema. A positive Stemmer's sign is present. Skin care, manual lymphatic drainage, exercises and movements have been associated with a two-layer compression bandage system. After 32 days of treatment with UrgoK2, we observe a reduction of volume of the limb and a clear improvement of the skin with a significant decrease of erythema.

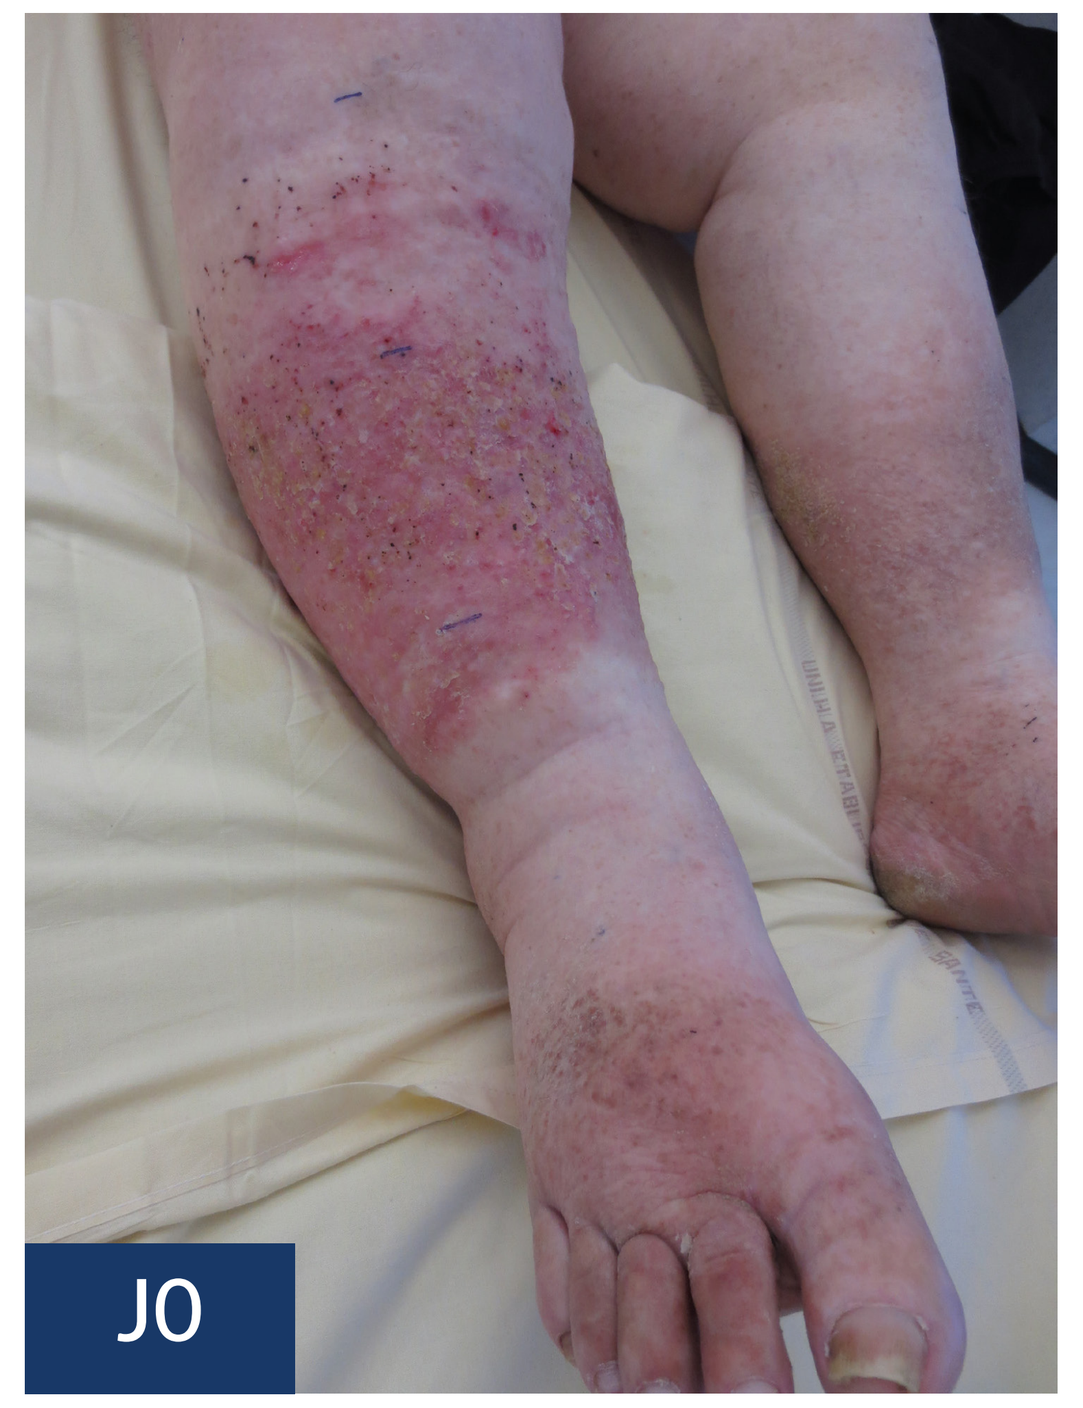
J0

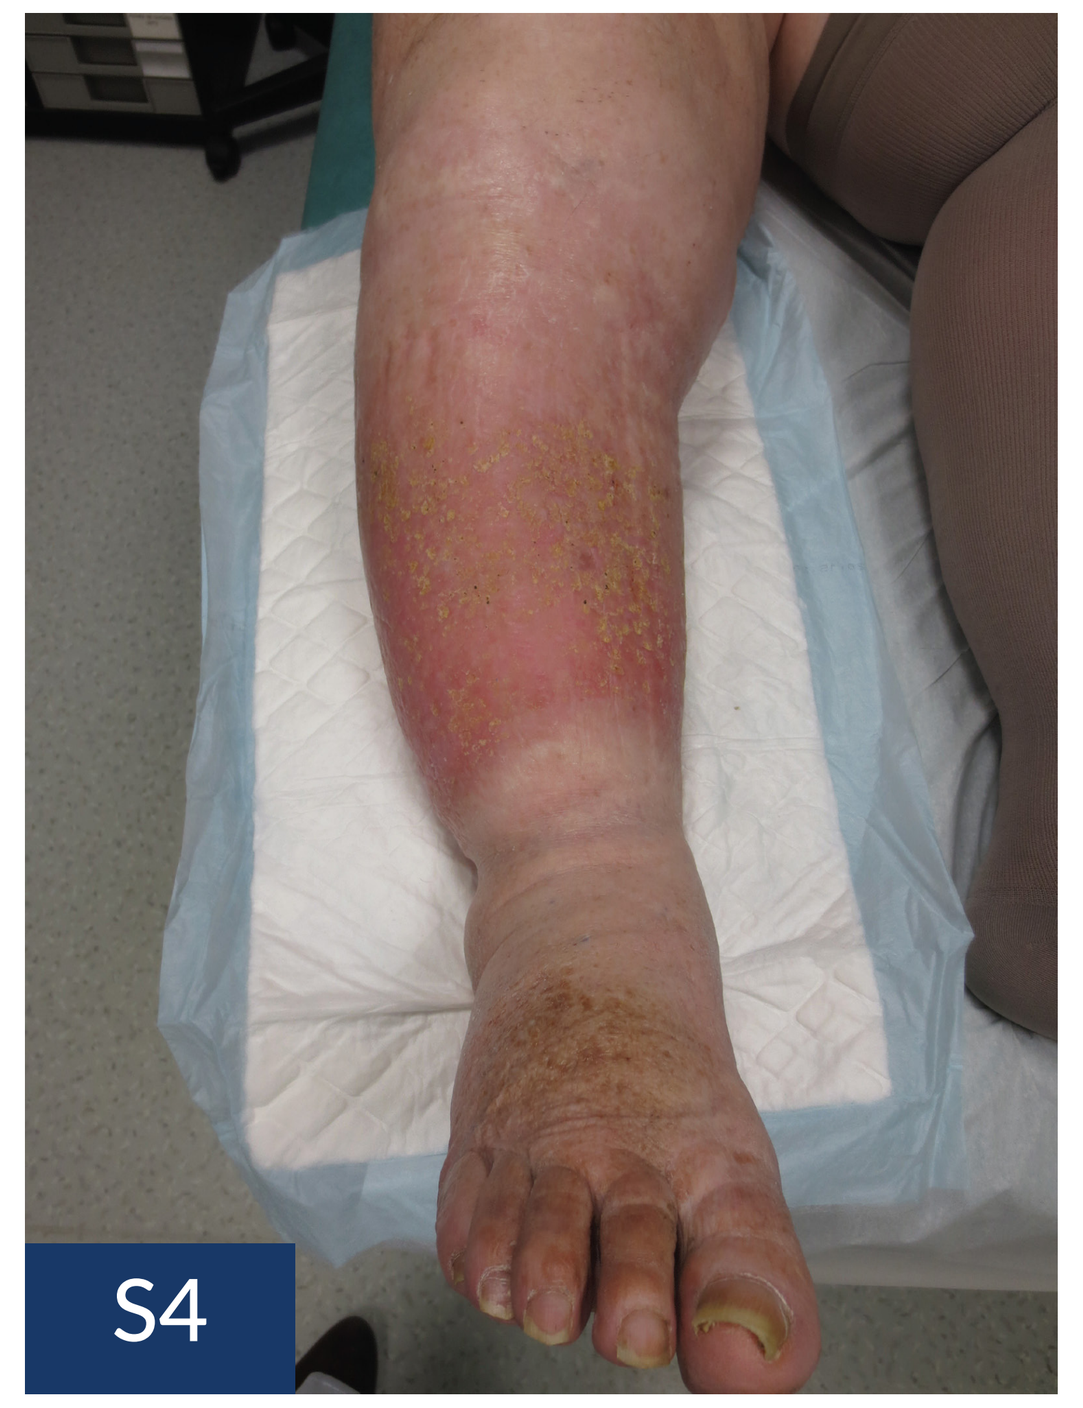
S4

CONCLUSION

The DCS compression bandage system is judged as effective and well-tolerated in this chronic pathology and may play an important role in the treatment of lower limbs lymphedema. Further studies are needed in this area. It is possible that the respective dynamics of the short stretch and long stretch bandages come together in synergistic ways to provide lymphedema relief. Biomechanical studies are warranted in this area.

*UrgoK2 Bandage System, Urgo Medical North America

REFERENCES

1. Ayman A. Grada, Tania J. Phillips, Lymphedema: Pathophysiology and clinical manifestations, Journal of the American Academy of Dermatology, Volume 77, Issue 6, 2017, Pages 1009-1020,

2. Benigni JP, Lazareth I, Parpex P, Gerard JL, Alves M, Vin F, Meaume S, Senet P, Allaert FA. Efficacy, safety and acceptability of a new two-layer bandage system for venous leg ulcers. J Wound Care. 2007 Oct;16(9):385-90. doi: 10.12968/jowc.2007.16.9.27866. https://pubmed.ncbi.nlm.nih.gov/17987751/

3. Hanna R, Bohbot S, Connolly N. A comparison of inferface pressures of three compression bandage systems. Br J Nurs. 2008 Nov 13-26;17(20):S16-24. doi: 10.12968/bjon.2008.17.Sup9.31661. https://pubmed.ncbi.nlm.nih.gov/19043323/

4. Jünger M, Ladwig A, Bohbot S, Haase H. Comparison of interface pressures of three compression bandaging systems used on healthy volunteers. J Wound Care. 2009 Nov;18(11):474, 476-80. doi: 10.12968/jowc.2009.18.11.45000. https://pubmed.ncbi.nlm.nih.gov/19901877/

5. Lantis JC 2nd, Barrett C, Couch KS, Ehmann S, Greenstein E, Ostler M, Tickner A. A dual compression system: preliminary clinical insights from the US. J Wound Care. 2020 Sep 1;29(Sup9):S29-S37. doi: 10.12968/jowc.2020.29.Sup9.S29. https://pubmed.ncbi.nlm.nih.gov/32924806/

6. Lazareth I, Moffatt C, Dissemond J, Lesne Padieu AS, Truchetet F, Beissert S, Wicks G, Tilbe H, Sauvadet A, Bohbot S, Meaume S. Efficacy of two compression systems in the management of VLUs: results of a European RCT. J Wound Care. 2012 Nov;21(11):553-4, 556, 558 passim. doi: 10.12968/jowc.2012.21.11.553. https://pubmed.ncbi.nlm.nih.gov/23413494/