The Healing Power of Nature

The Impact of HbA1c Levels on Diabetic Foot Ulcers Using Acellular Fish Skin Grafts

Ryan Huang, D.O, CWSP, Alexis Chan, Shenmei Wu, Shenlone Wu
Professional Wound Specialists, Las Vegas. NV

INTRODUCTION

Diabetic foot ulcers (DFUs) has become a serious public health problem due to its high prevalence and complications. Advanced treatment for DFU is often delayed while blood glucose (HbA1c) is elevated as one of the underlying factors for impaired wound healing. There are currently no studies on the association of HbA1c levels on the efficacy of treatment of DFUs using skin substitutes. Acellular fish skin (AFS) has proven to be effective in treating DFUs compared to SOC. This is a retrospective review exploring the effect of HbA1c levels on DFUs healing for type II diabetes patients treated with Kerecis AFS.

MATERIALS AND METHODS

This study reviewed 20 cases of chronic DFUs (defined as DFUs that did not show improvement after 30 days of standard of care treatment). Patients were broken into two groups (n= 10 patients/group) with HbA1c level between 6-8 and the other is HbA1c >8.

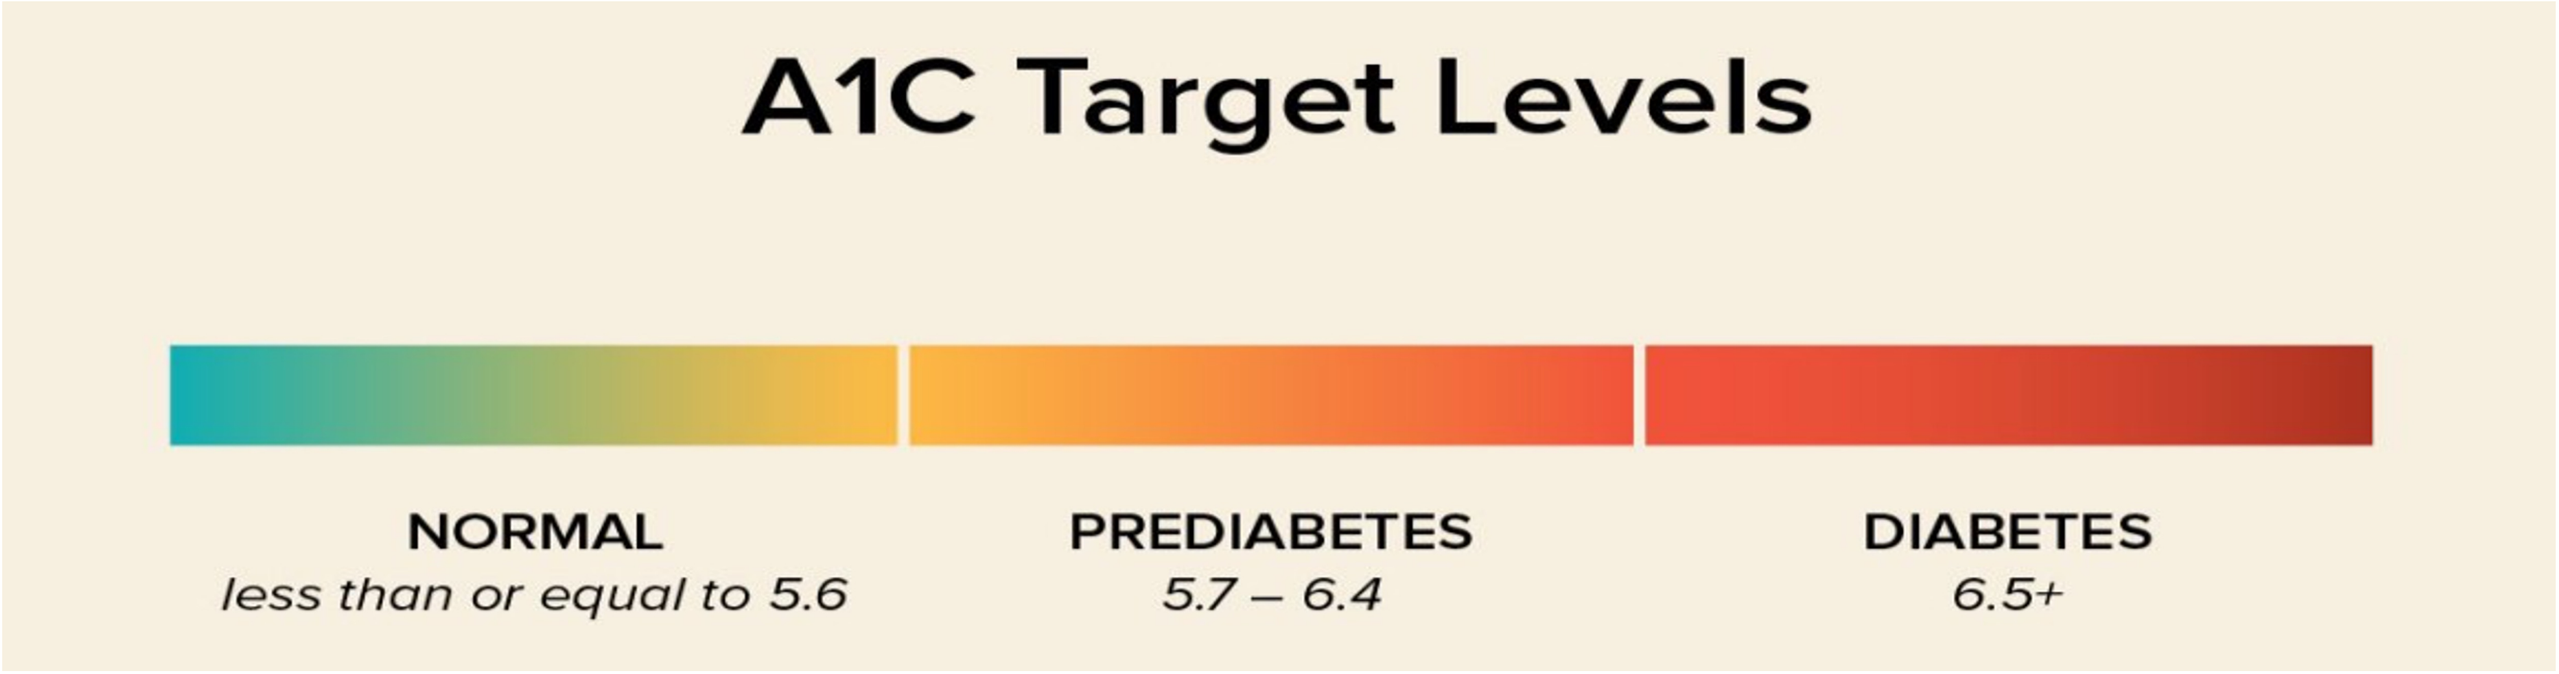
A1C Target Levels
NORMAL
less than or equal to 5.6
PREDIABETES
5.7 – 6.4
DIABETES
6.5+

Figure 1. National Institute of Diabetes and Digestive and Kidney Diseases indication of different A1C levels.

Patients' HbA1c levels were measured within the first 3 months of their first treatment. If the level was not obtained from the primary care physician, then a level was obtained by PWS prior to the study. The patient's chronic DFU was treated once a week using the PWS treatment standard. Wound sizes were measured before the first treatment and after the 4th treatments over a 4-week time period. Demographic information recorded include gender and age

Treatment: The DFU wound bed was first prepared with a wound cleanser. Afterwards, a curette was used to stimulate the wound bed as well as removing any bioburden. AFS was then rehydrated with normal saline and placed on the wound bed with the scale side facing away from the wound bed. A mesh was then placed and secured with steristrips. Finally, an outer dressing was placed over the wound. Treatment was performed on a weekly basis

RESULTS

The group with HbA1c 6-8 showed an average of -41.30% decrease in wound size. The HbA1c >8 group had an average of -30.41% wound size reduction. After performing an independent t-test, t-value: -0.56, p-value: 0.299 larger than 0.05. This indicated that the HbA1c levels do not have a statistically significant effect on wound healing time.

Table 1. Patient age, gender and wound size reduction among two groups

HbA1c 6-8

Patient	Gender	Age	% Size Change
1	Female	91	-7
2	Male	64	-21
3	Female	78	-85
4	Female	73	-77
5	Female	78	-21
6	Female	99	-11
7	Male	72	-39
8	Male	65	-45
9	Female	69	-53
10	Male	79	-54
Average	6F-4M	77	-41.30
SD		11.07	26.72

HbA1c >8

Patient	Gender	Age	% Size Change
11	Male	64	-32
12	Male	44	-53
13	Male	75	-90
14	Female	71	-89
15	Male	86	-57
16	Female	75	66
17	Male	73	-60
18	Male	67	-10
19	Male	66	-40
20	Female	74	61
Average	3F -7M	70	-30.40
SD		10.89	55.01

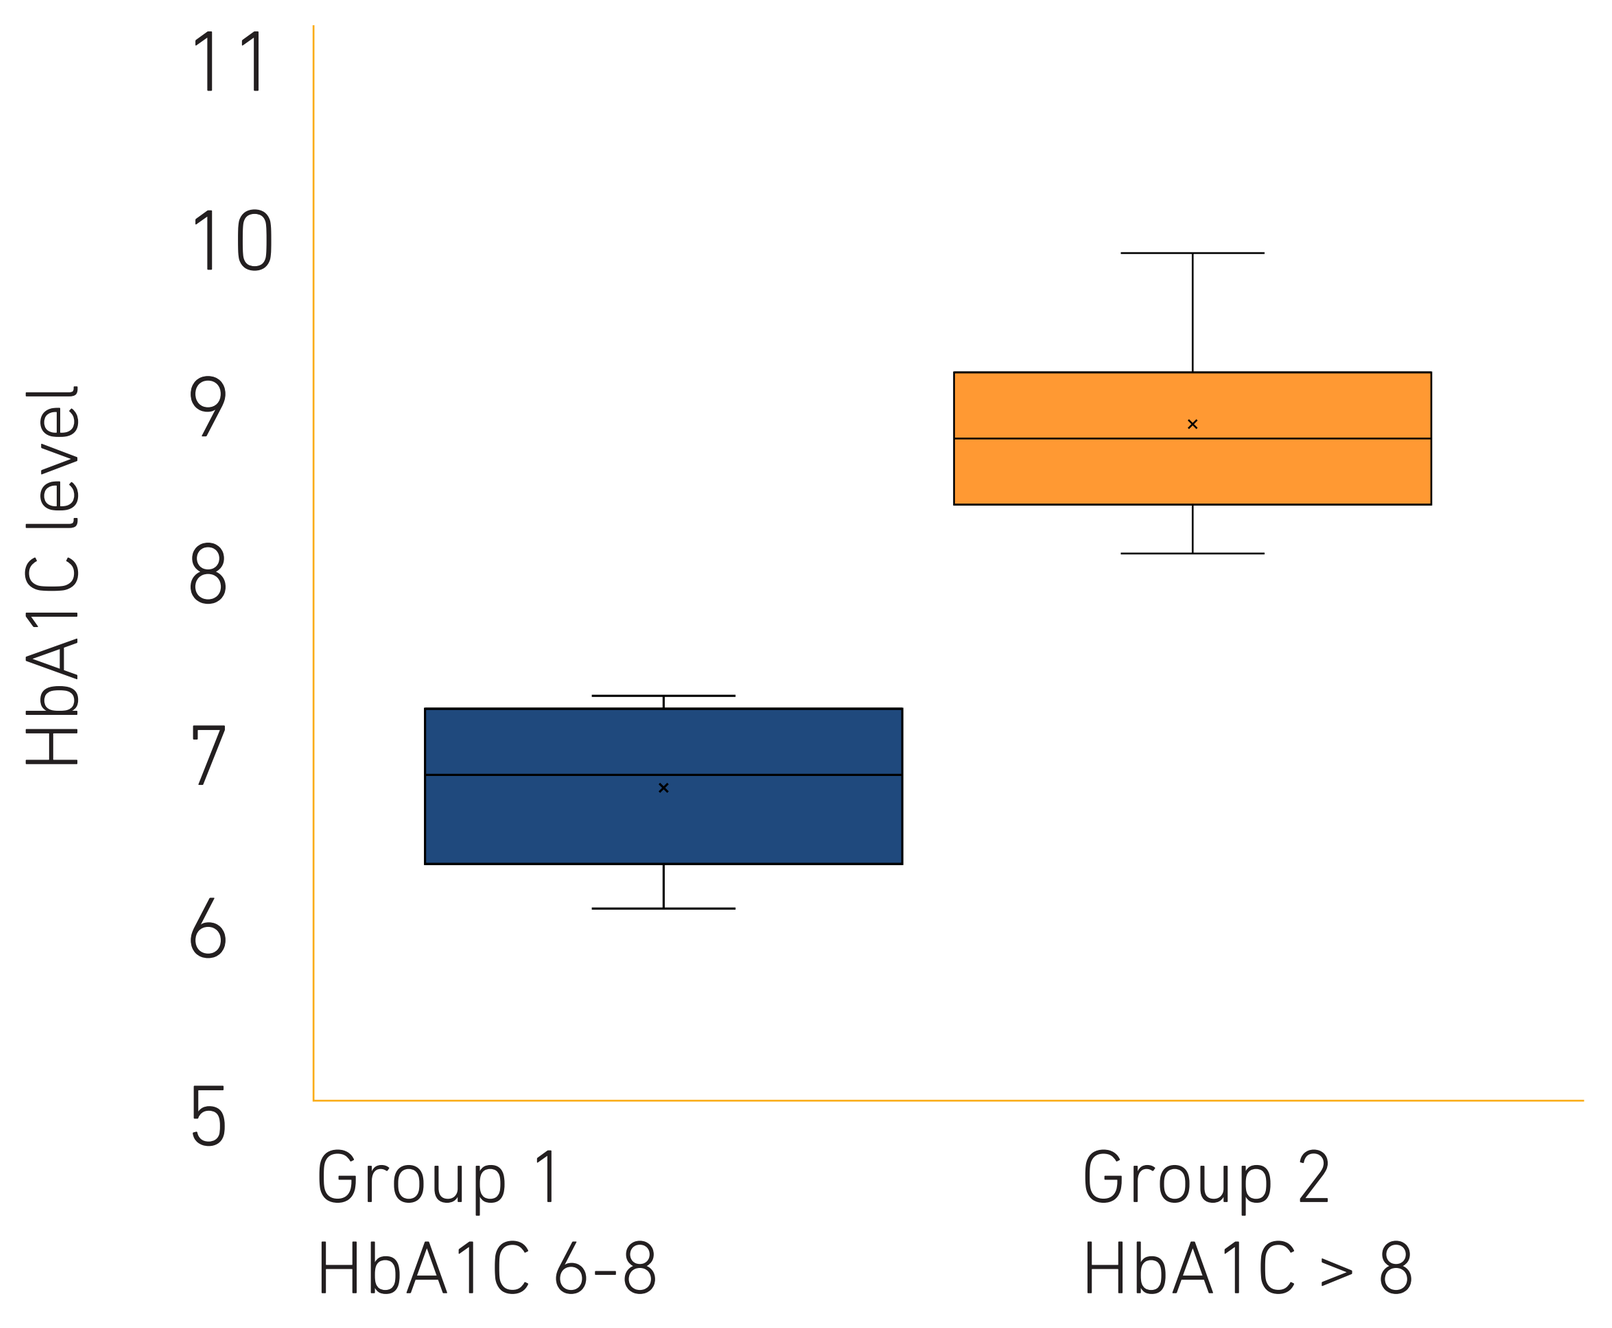
HbA1C level
11
10
9
8
7
6
5
Group 1
HbA1C 6-8
Group 2
HbA1C > 8

Figure 2. Comparison of HbA1c levels between the two groups

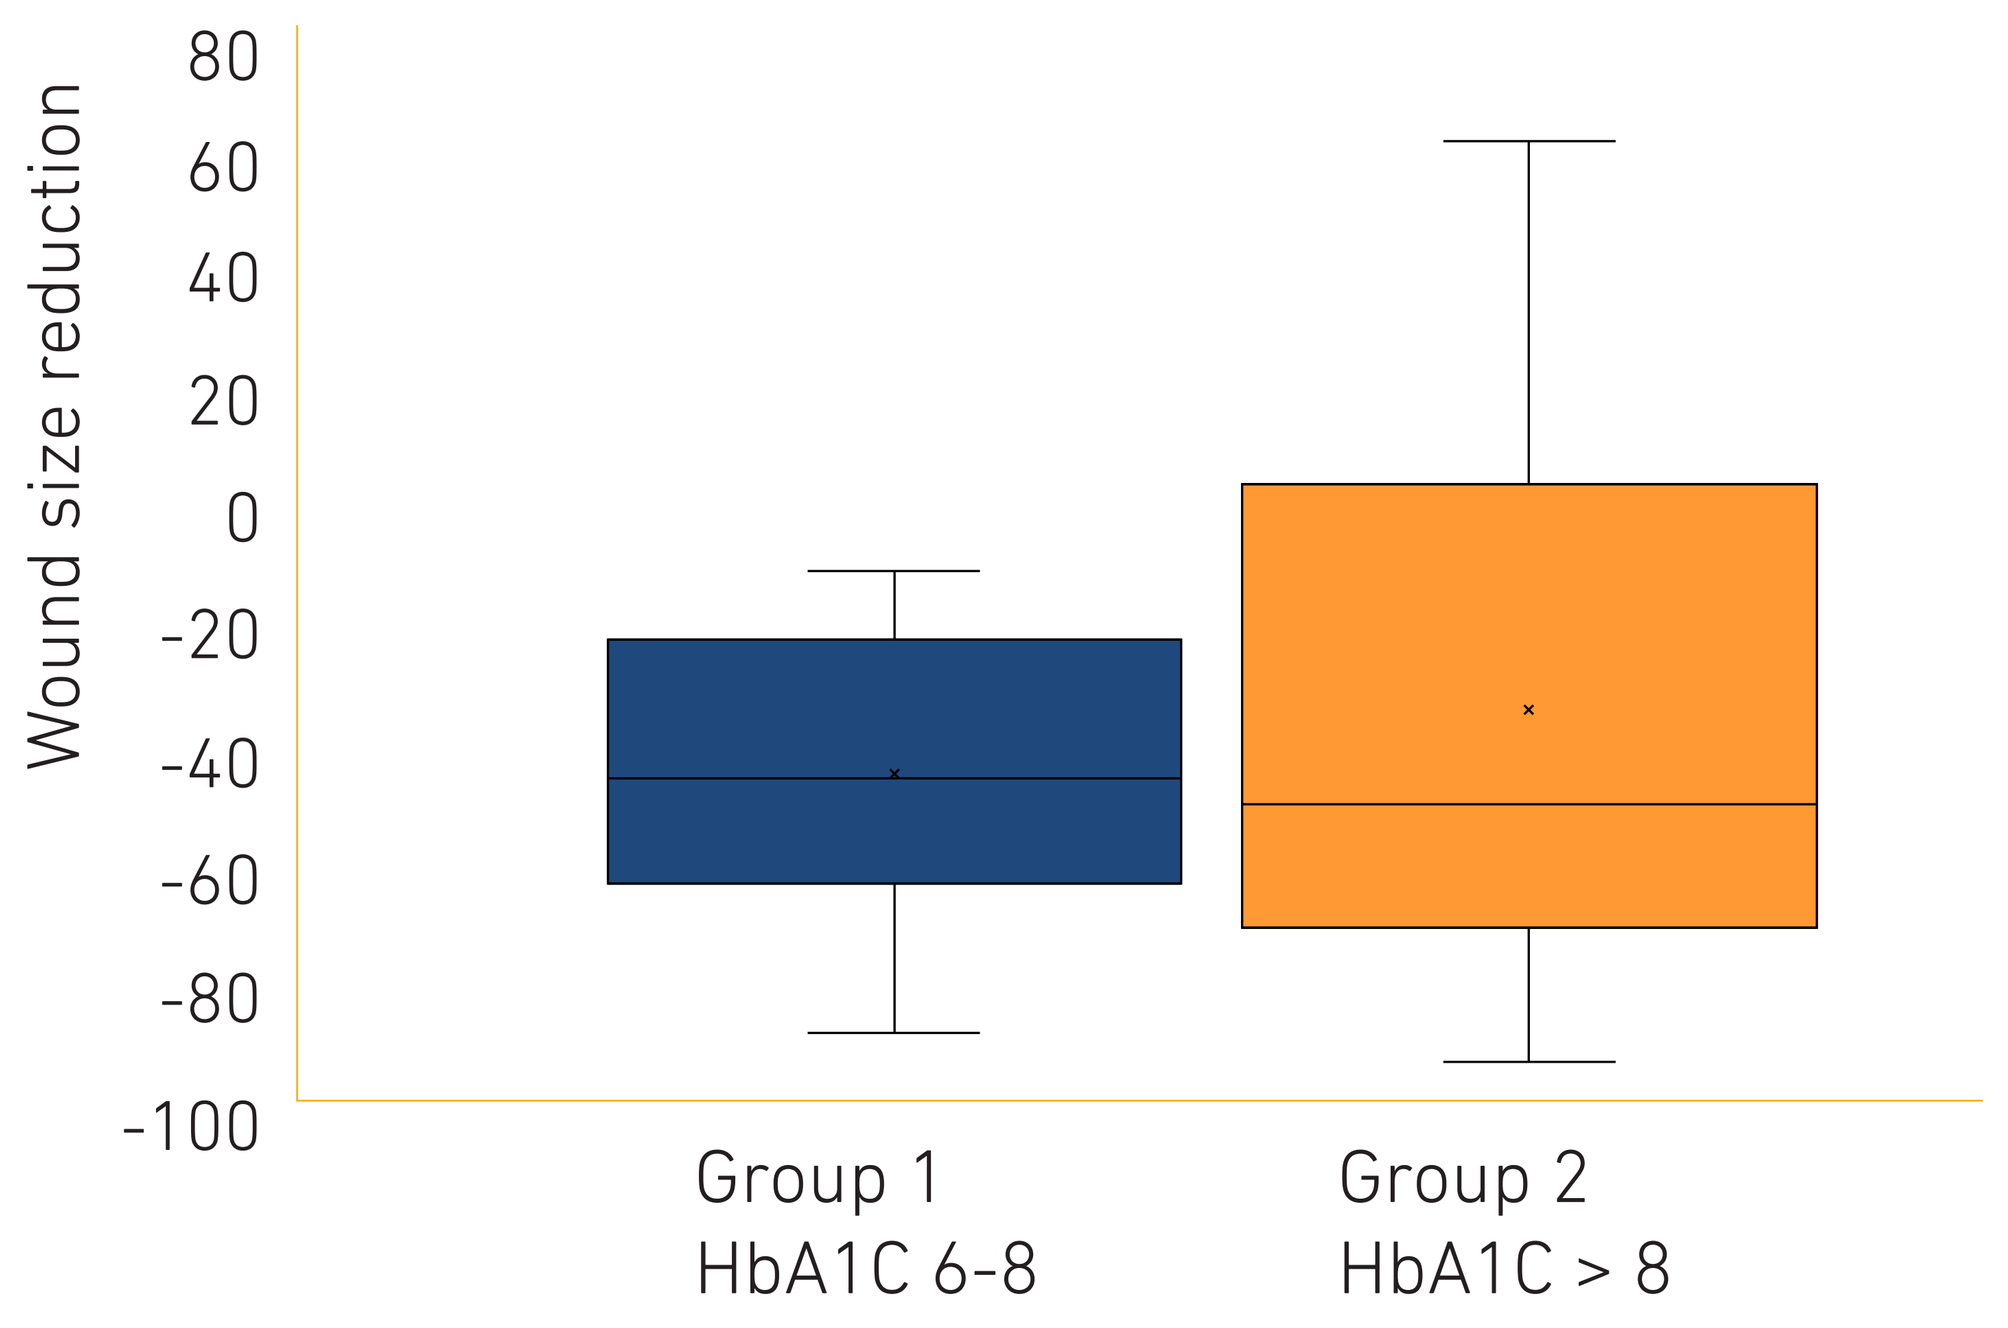
Wound size reduction
80
60
40
20
0
-20
-40
-60
-80
-100
Group 1
HbA1C 6-8
Group 2
HbA1C > 8

Figure 3. Comparison of wound size reduction between the two groups

DISCUSSION

According to the analysis, there is no correlation between HbA1c levels from two groups with their wound size reduction rate. The baseline HbA1c levels were not associated with ulcer healing. The average wound size reduction rate was at 41.3% for lower HbA1c level group and 30.4% for higher HbA1c level group. This result is consistent with previous literature indicating that baseline HbA1c is not associated with lower extremity wound healing in patients with diabetes.

CONCLUSIONS

Advanced therapeutics for DFUs are often delayed while HbA1c levels are corrected because of belief that the wound will not respond to advanced therapy. This delay in treatment can lead to deterioration in the wound and leave the wound at risk of infection. The study findings suggest that AFS is an effective treatment for chronic DFU despite elevated baseline HbA1c levels. The results indicate that advanced therapy could be applied concurrently to HbA1c management as the primary treatment for DFUs. Limitations of the current study is sample size and powered. A larger study is warranted to test the hypothesis.

References
[1] Christman, Andrea L et al. "Hemoglobin A1c predicts healing rate in diabetic wounds." The Journal of investigative dermatology vol. 131,10 (2011): 2121-7. [2]Fesseha, Betiel K et al. "Association of Hemoglobin A1c and Wound Healing in Diabetic Foot Ulcers." Diabetes care vol. 41,7 (2018): 1478-1485. [3]Katie Glover, Alexandros Ch. Stratakos, Aniko Varadi, Dimitrios A. Lamprou, 3D scaffolds in the treatment of diabetic foot ulcers: New trends vs conventional approaches, International Journal of Pharmaceutics, Volume 599, 2021, 120423, ISSN 0378-5173, [4] Lullove EJ, Liden B, Winters C, McEneaney P, Raphael A, Lantis II JC. A multicenter, blinded, randomized controlled clinical trial evaluating the effect of Omega-3–rich fish skin in the treatment of chronic, nonresponsive diabetic foot ulcers. Wounds. 2021;33(7):169-177. [5] Oliver TI, Mutluoglu M. Diabetic Foot Ulcer. [Updated 2021 Aug 19]. In: StatPearls [Internet]. Treasure Island (FL): StatPearls Publishing; 2022 Jan-. [6] Singh N, Armstrong DG, Lipsky BA. Preventing foot ulcers in patients with diabetes. JAMA. 2005 Jan 12;293(2):217-28. doi: 10.1001/jama.293.2.217. PMID: 15644549. [7] Younis, Bilal Bin et al. "Frequency of foot ulcers in people with type 2 diabetes, presenting to specialist diabetes clinic at a Tertiary Care Hospital, Lahore, Pakistan." BMC endocrine disorders vol. 18,1 53. 6 Aug. 2018.

Professional Wound Specialists