WOUND CLEANSING: IRRIGATION VS BIOFILM-BASED WOUND CARE (BBWC); HOW GUIDELINES MEET PRACTICE WITH USE OF THE PURE HYPOCHLOROUS ACID (PHA) SOLUTION.

ELIZABETH FAUST, MSN, CRNP, CSWS, CWOCN-AP, MAPWCA
TOWER HEALTH SYSTEM, WEST READING, PA

INTRODUCTION

The latest International Wound Infection Institute (IWII) Clinical Practice guidelines states that wound hygiene involves cleansing, debridement, and prevention of biofilm reformation. Wound cleansing is recommended to be done with irrigation with 4-15 PSI. IWII guidelines also recommend Biofilm Based Wound Care (BBWC) which should include physically removing microorganisms from the wound bed and creating an environment that prevents or delays biofilm formation. This can be achieved via the newer generation of wound cleansers.

Another International Consensus Document, published in the Journal of Wound Care, titled "Defying hard-to-heal wounds with an early antibiofilm intervention strategy: Wound hygiene" contains some very relevant points pertinent to wound cleansing which are in alignment with the IWII guidelines.

"Saline or water rinses/flushes will not remove biofilm. ***Cleansing with intent and appropriate tools/ solutions prepares the wound bed for debridement.*** *It is essential that the periwound skin is cleansed to remove further sources of contamination."*

"Highly cytotoxic solutions, such as those containing povidone-iodine and hydrogen peroxide, are not recommended. Ideally, a skin cleanser designed for daily used should be chosen, to balance the need to ***disrupt the microbial load while maintaining skin integrity****."*

The guidelines moreover urge the wound pH to be deliberately moved towards acidic, something that is not possible with saline, or the more alkaline cleansers such as Dakin's, which are cytotoxic. This is possible with some cleansers, for example the pure Hypochlorous Acid (pHA)-based cleansers.

Clinically, patients who has simple saline cleansing did not have true wound hygiene performed. This poster will show case studies that highlight the clinical relevance of BBWC and soaking vs spraying to cleanse and wound or perform wound hygiene.

METHOD

A retrospective chart review was performed on cases that the wound, ostomy, continence nurse (CWOCN) was consulted for wounds on admission. In each case, the wound was cleansed first by the bedside staff with normal saline solution (NSS). In each of these cases, subsequently, the CWOCN soaked the wounds with pHA cleanser on a gauze for 5-10 minutes followed by firm wiping with gauze/other mechanical action to remove the loosened debris and germs/biofilm loosened.

RESULTS

In all four cases presented, the patient's wound had debris remain within the wound base despite cleansing with NSS. After soaking with pHA cleanser, all the cases show that while debris remained on the wound base when cleansed with NSS, with soaking of a pHA cleanser of 5-10 minutes resulted in clearance of the debris.

Initial Presentation

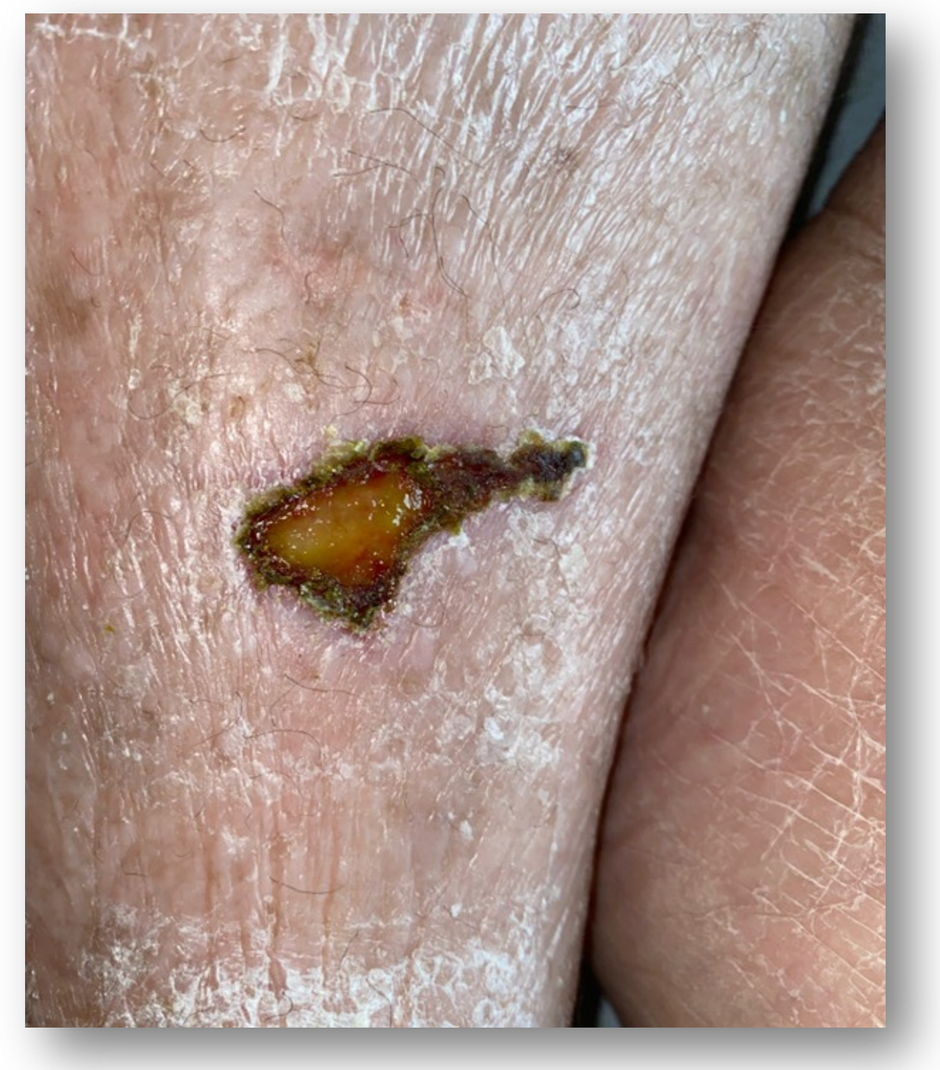

CASE 1

Traumatic leg wound. NO malodor, no surrounding erythema or signs of infection. Obvious need to properly cleanse and treat the wound.

After 5-minute soak with Vashe

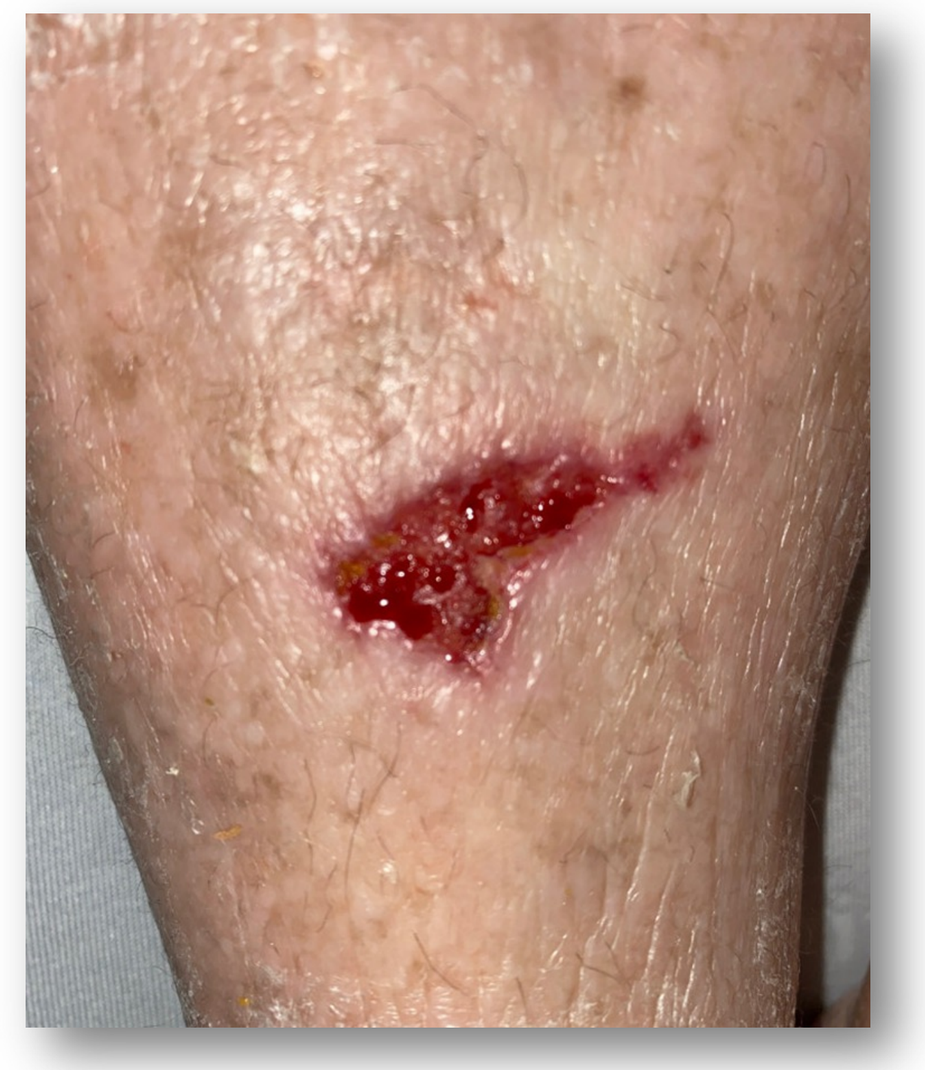

Initial Presentation

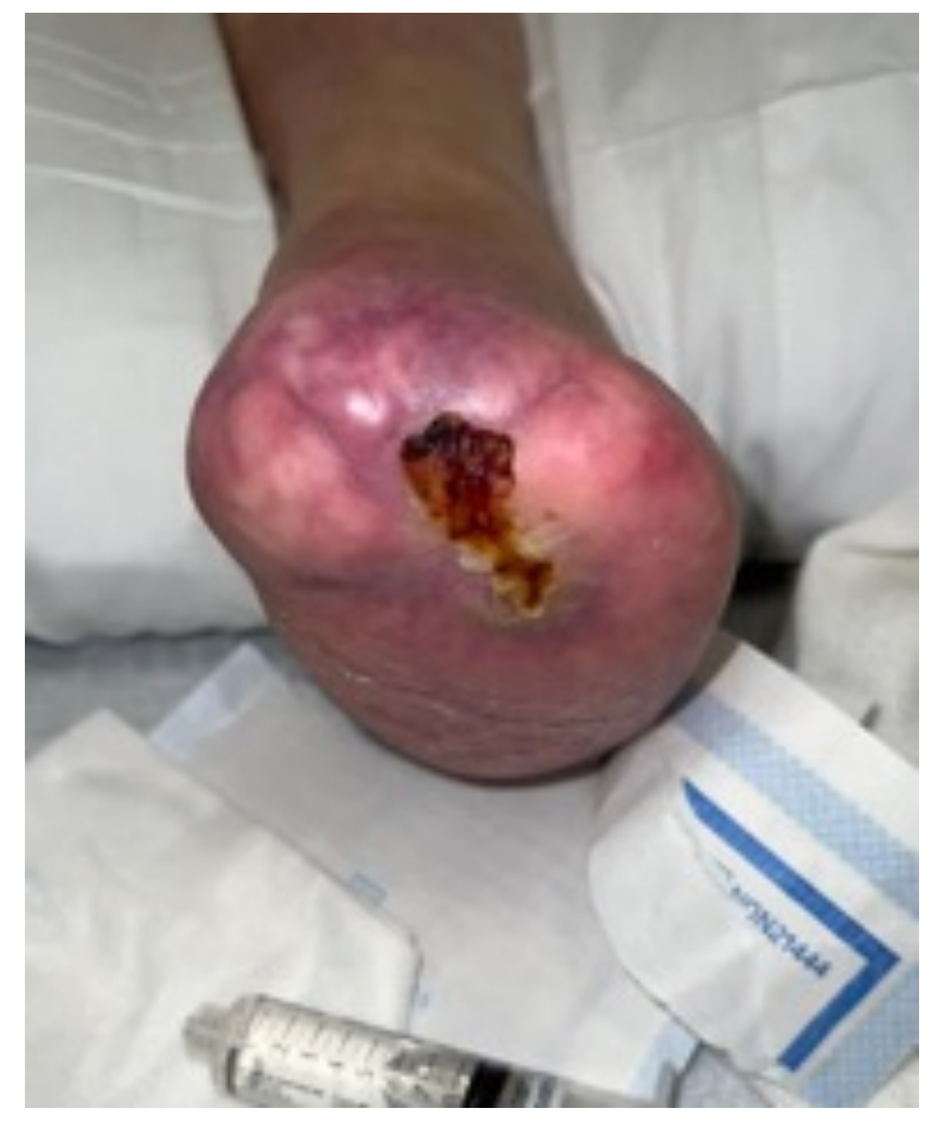

CASE 2

Diabetic foot ulcer on a left TMA. No signs of infection.

After 5-minute soak with Vashe

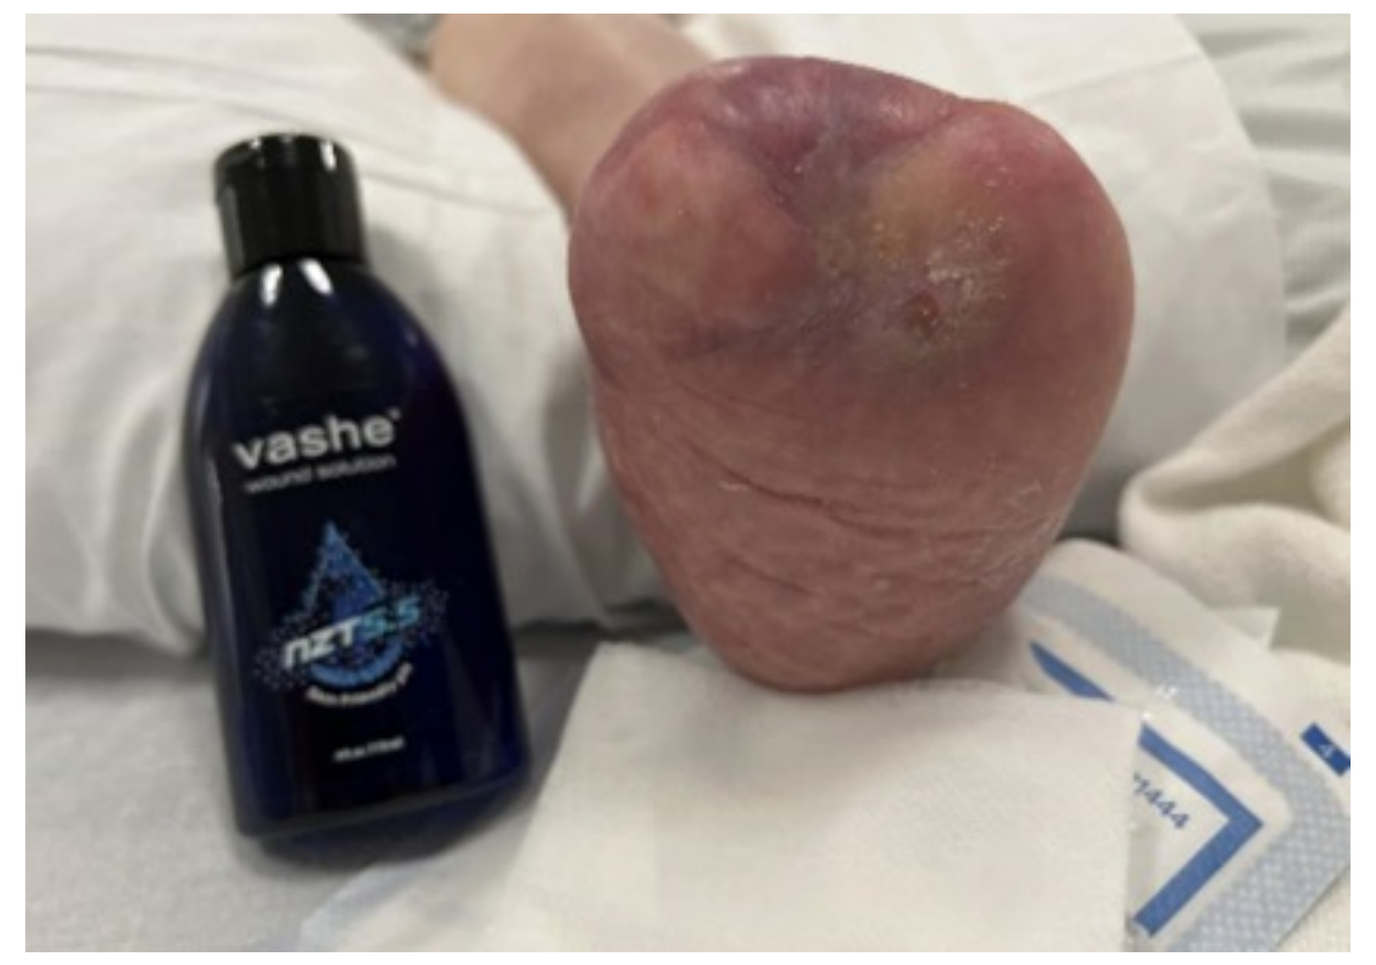
vashe

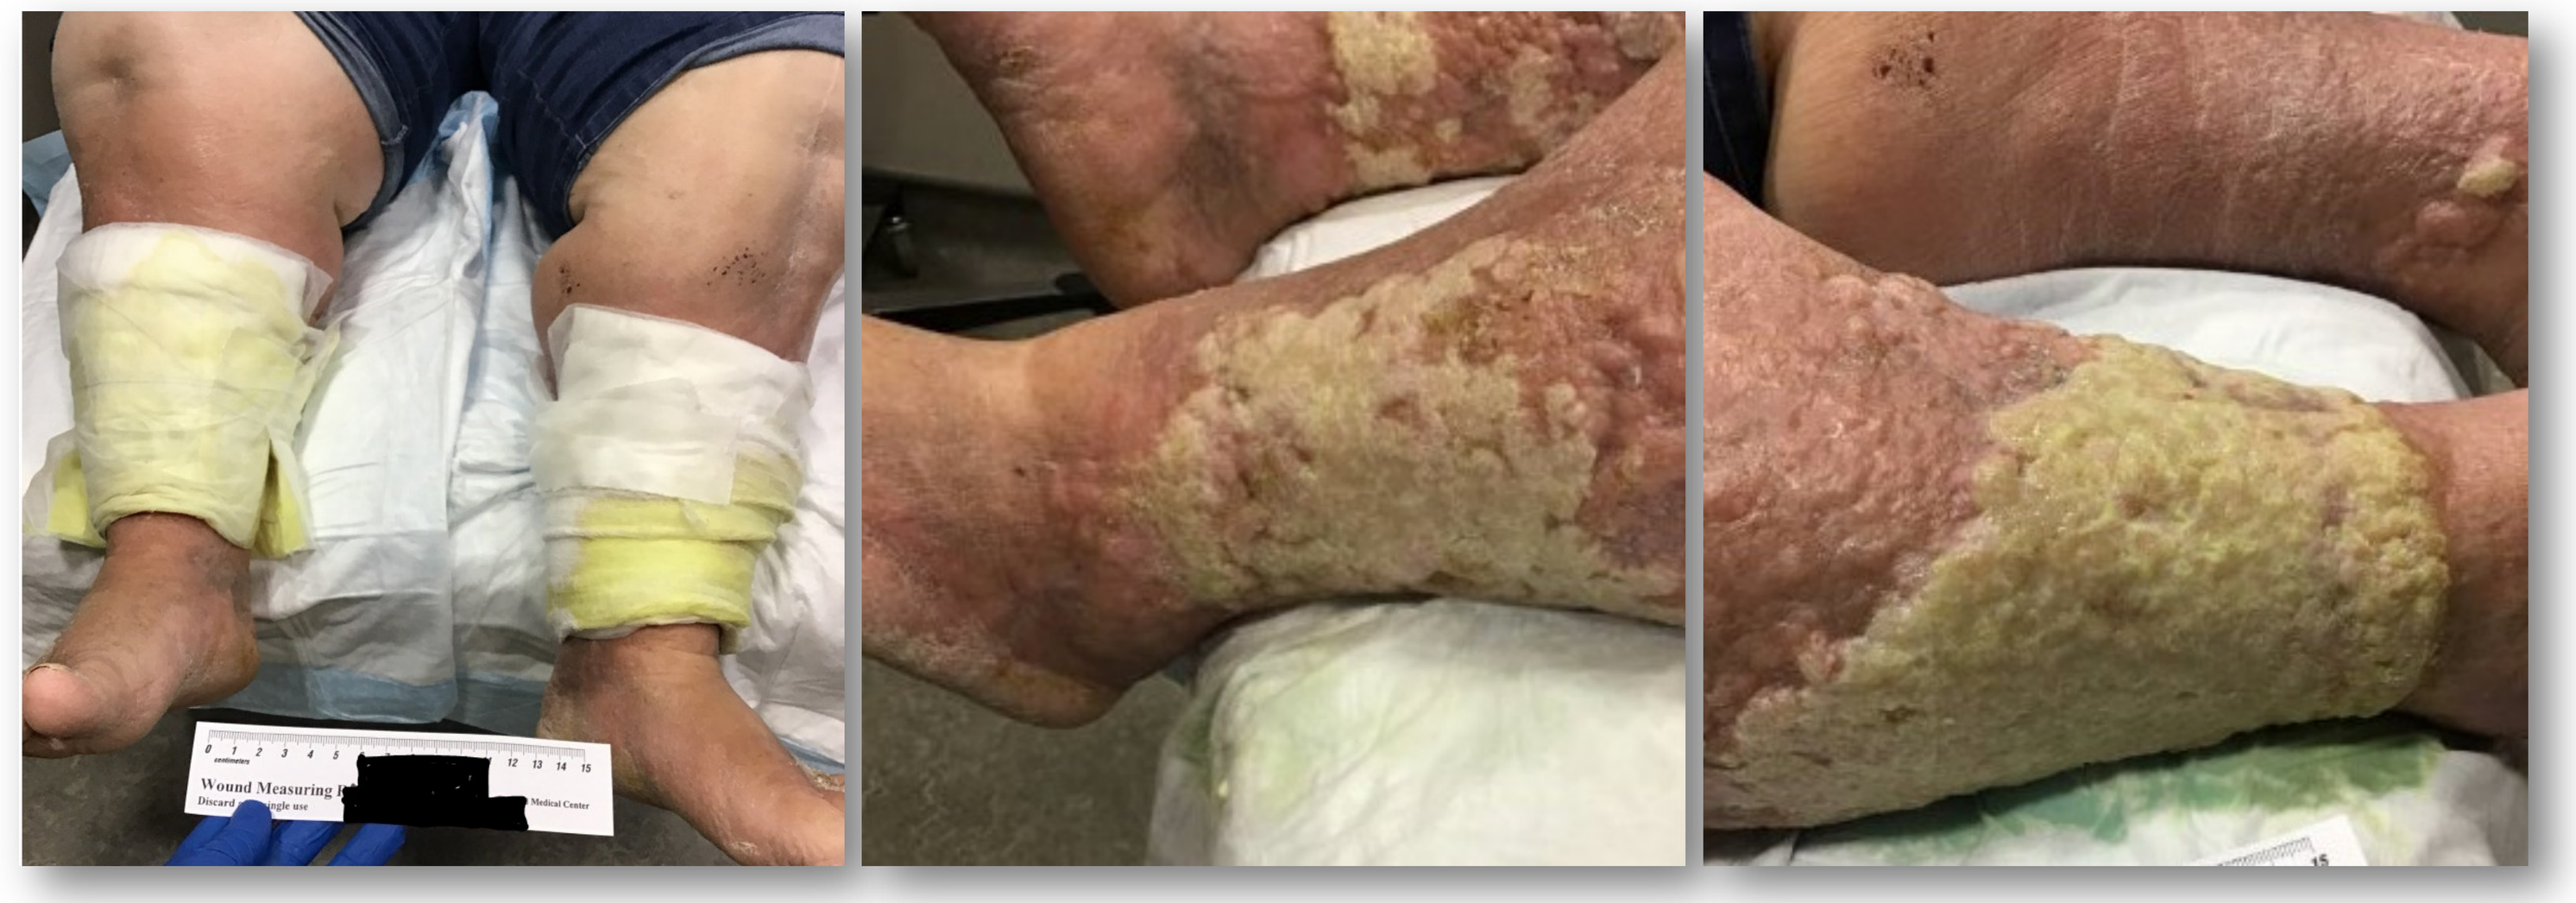

CASE 3

Bilateral Venous Leg Ulcers (VLU) with bacterial burden upon presentation to wound care clinic.

After Vashe soak and cleansing

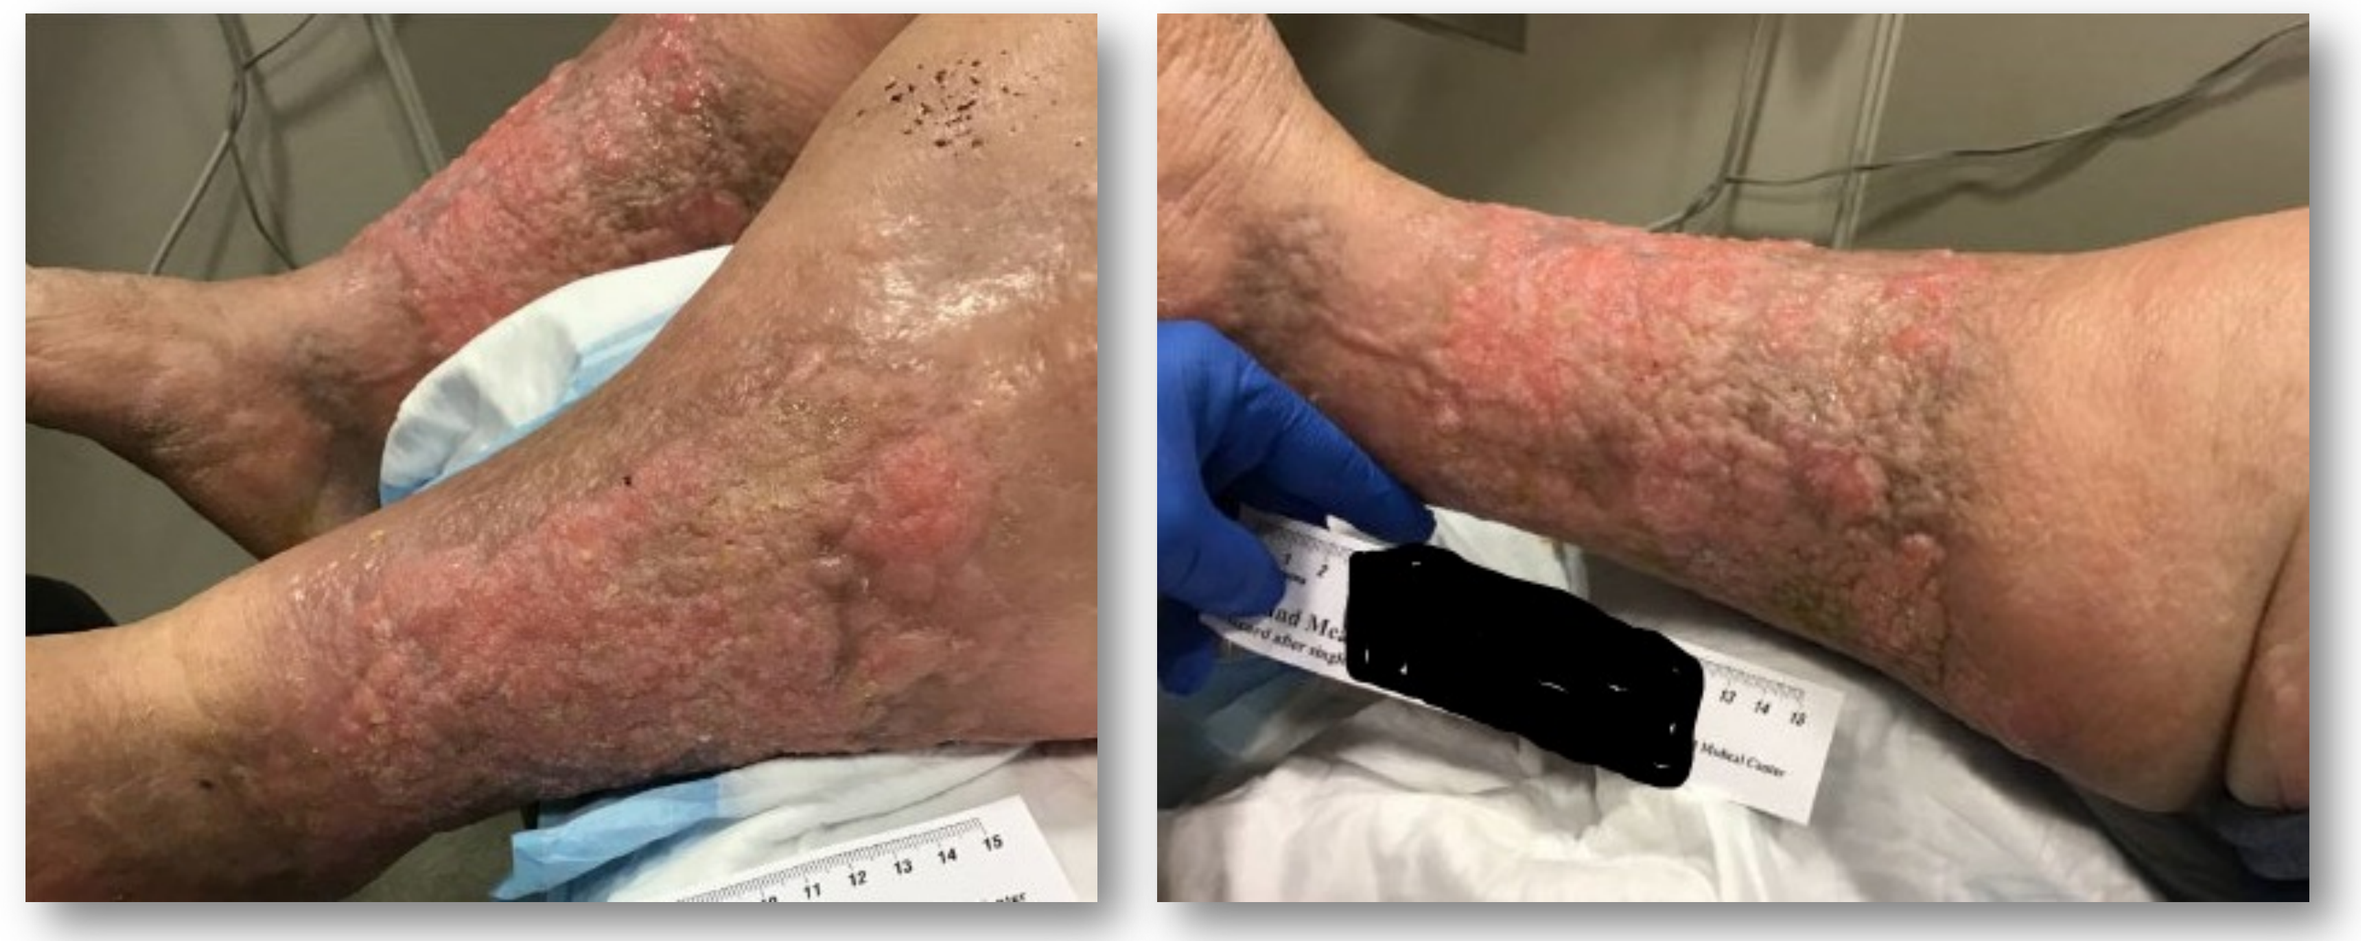

CASE 4

Debrided traumatic flap (POD3) who was going to return to the OR for debridement. After a 10-minute soak and mechanical debridement, the wound showed rapid improvement. NPWTi-d was applied with pHA cleanser and the patient was discharge with NPWT 2 days after without trip required to go to the OR.

Pre-Vashe Soak

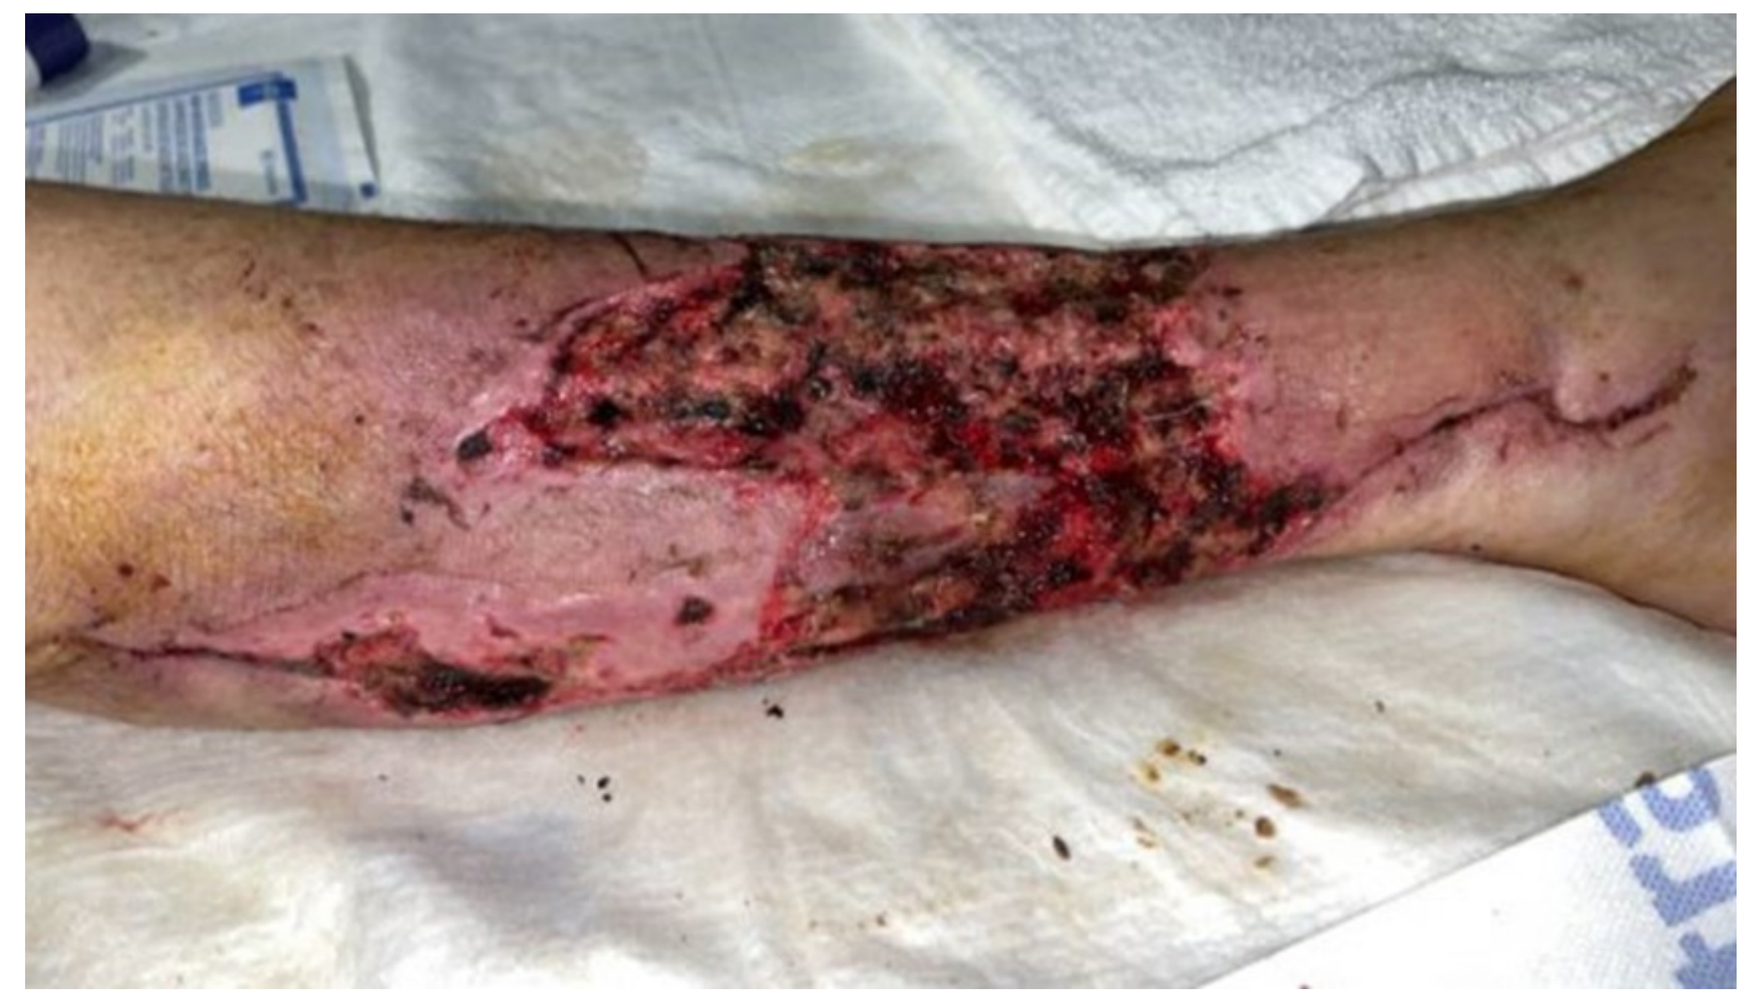

Post-Vashe Soak

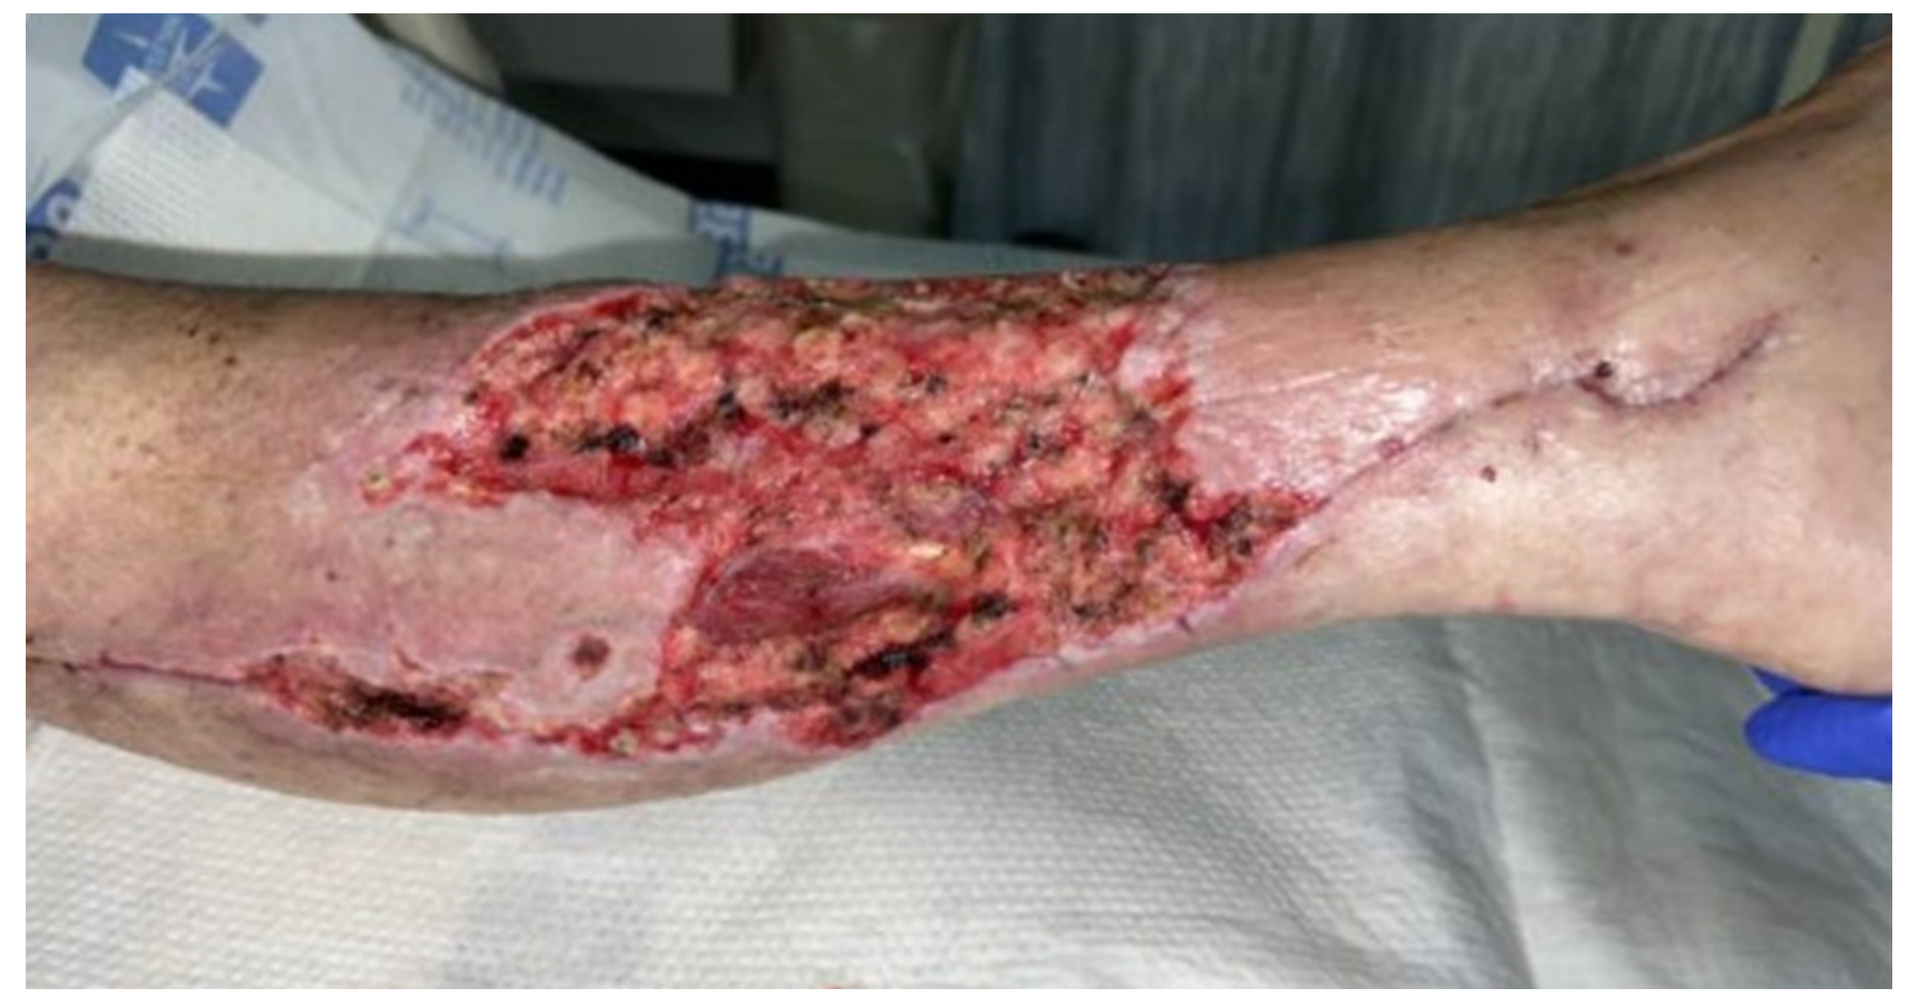

DISCUSSION

The use of a pHA has extensive evidence base in chronic and acute wounds. This data is not based on irrigation, or spraying, or dripping, but rather by soaking for 5-10 minutes The value of soaking, rather than irrigating or spraying a wound should be directly compared in wound cleansing with standard methods. While pHA cleanser is an integral part of BBWC in her own practice, this author would argue that it should become the standard of care for wound cleansing. Additionally, the pHA is relatively not cytotoxic compared to other antimicrobial cleansers, and particularly to those that contain hypochlorite, and can provide wound cleansing OR BBWC without compromising on wound progression based on the cases presented as well as the peer reviewed evidence.

REFERENCES

1. International Wound Infection Institute (IWII) Wound Infection in Clinical Practice. Wounds International, 2022.
2. Murphy C, Atkin L, Swanson T, Tachi M, Tan YK, de Ceniga MV, Weir D, Wolcott R, Ĉernohorská J, Ciprandi G, Dissemond J, James GA, Hurlow J, Lázaro MartÍnez JL, Mrozikiewicz-Rakowska B, Wilson P. Defying hard-to-heal wounds with an early antibiofilm intervention strategy: wound hygiene. J Wound Care. 2020 Mar 1;29(Sup3b):S1-S26.
3. Sibbald, GR MD, Elliott, JA. Et. Al. (2021) Wound Bed Preparation 2021, Advances in Skin & Wound Care: April 2021 - Volume 34 - Issue 4 - p 183-195
4. Day A, Alkhalil A, Carney BC, Hoffman HN, Moffatt LT, Shupp JW. Disruption of Biofilms and Neutralization of Bacteria Using Hypochlorous Acid Solution: An In Vivo and In Vitro Evaluation. Adv Skin Wound Care 2017 Dec;30(12):543-551.
5. Robson MC. Treating chronic wounds with hypochlorous acid disrupts biofilm. Wound Prevention and Management 2020;66 (5):9-10.
6. Harriott MM, Bhindi N, Kassis S, Summitt B, Perdikis G, Wormer BA, Rankin TM, Kaoutzanis C, Samaha M, Stratton C, Schmitz JE. Comparative Antimicrobial Activity of Commercial Wound Care Solutions on Bacterial and Fungal Biofilms. Ann Plast Surg. 2019 Oct;83(4):404-410.
7. Eriksson, E, Liu, PY, Schultz, GS, et al. Chronic wounds: Treatment consensus. Wound Rep Reg. 2022; 30(2): 156- 171.
8. Rodeheaver, GT, Ratliff, CR. Wound Cleansing, wound irrigation, wound disinfection. In: Krasner DL, ed. Chronic Wound Care: The Essentials. Malvern, PA: HMP Communications, 2014; 47-62