The Healing Power of Nature

Simplifying the reconstruction of soft tissue defects following extirpative procedures for cutaneous malignancies using a decellularized fish skin graft*

Matthew Endara, MD
Maury Regional Health, Maury Regional Medical Group, Columbia, TN

INTRODUCTION

Cutaneous malignancies represent the most common and treatable form of cancer in the general population. Practitioners must be aware of their pervasive nature to avoid delay in their necessary excision and cure. If latency ensues, lesions can develop beyond simple excision and closure, necessitating reconstruction. The mainstay of treatment for these cancers is excision and pathologic analysis by a pathologist or the extirpative surgeon trained in Mohs technique. Results of formal pathology are not typically known for days. However, the Mohs technique provides immediate results. Irrespective of margin status, defects can be large and complex, requiring advanced, more invasive reconstructive procedures. Decellularized fish skin graft (FSG) has emerged as a promising reconstructive dermal matrix. The inherent nature of fish skin allows for gentle processing that preserves mechanical and biological properties capable of covering and regenerating exposed deep structures more rapidly than other products on the market. The ability of FSG to integrate rapidly is novel and is proving to be particularly useful in reconstructing post oncologic soft tissue defects when either traditional or mohs technique is used for extirpation

METHODS

Six patients (n=6) underwent surgical excision involving the use of decellularized FSG to aid in the reconstruction. Four Patients (n=4) with skin cancers underwent surgical excision by a reconstructive surgeon followed by pathological analysis with staged closures. The remaining two patients (n=2) underwent definitive reconstruction one-day post excision by a trained Mohs surgeon. In the delayed cases, the FSG was applied during excision to bridge a split-thickness skin graft (STSG). Following the Mohs procedures, patients underwent flap closure using FSG as adjunctive therapy to regenerate soft tissue avoiding donor site morbidity and reducing the need for a skin graft.

CASE 1

BASAL CELL CARCINOMA OF CHEST SKIN

Kerecis Applications: 1
Patient Outcomes: Successful staged reconstruction with definitive closure split thickness skin graft 28 days after primary excision

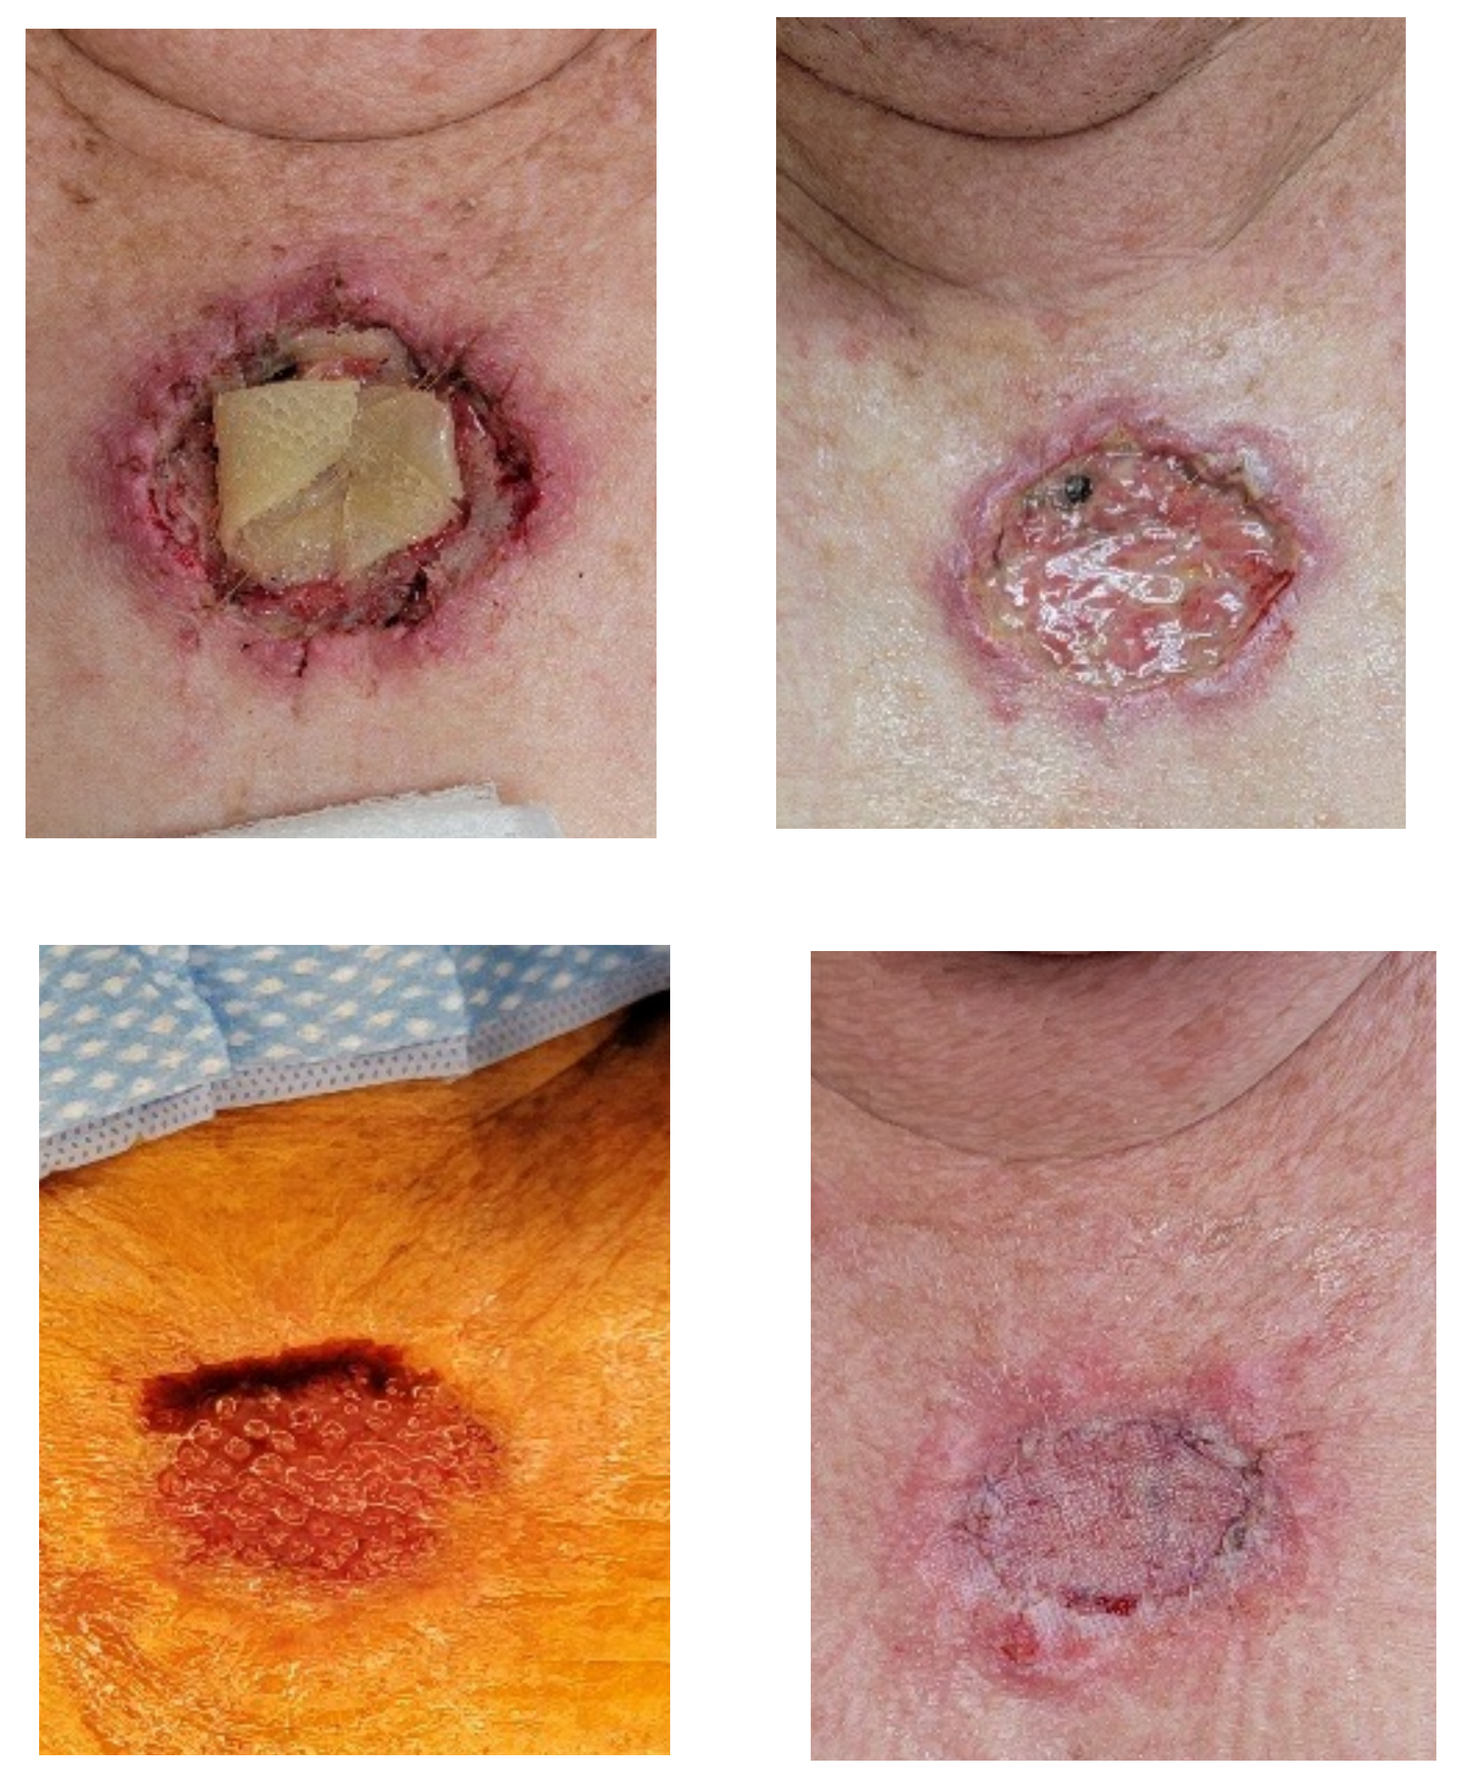

CASE 2

DORSAL FOOT BASAL CELL CANCER

Kerecis Applications: 1
Patient Outcomes: Cancer excised with application of kerecis leading to Successful staged reconstruction with definitive closure via split thickness skin graft 37 days after primary excision

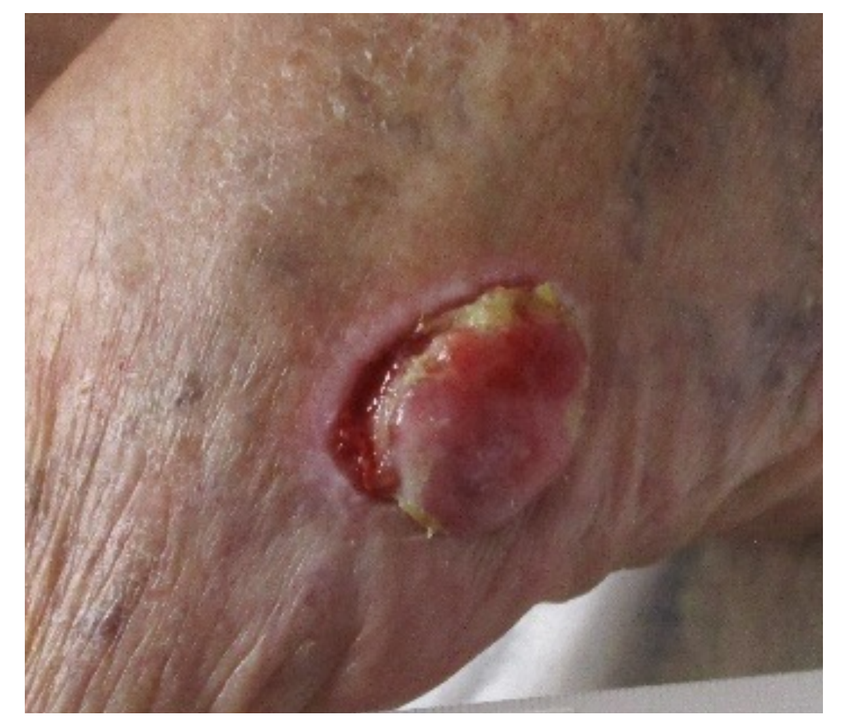

Dorsal foot exophytic mass

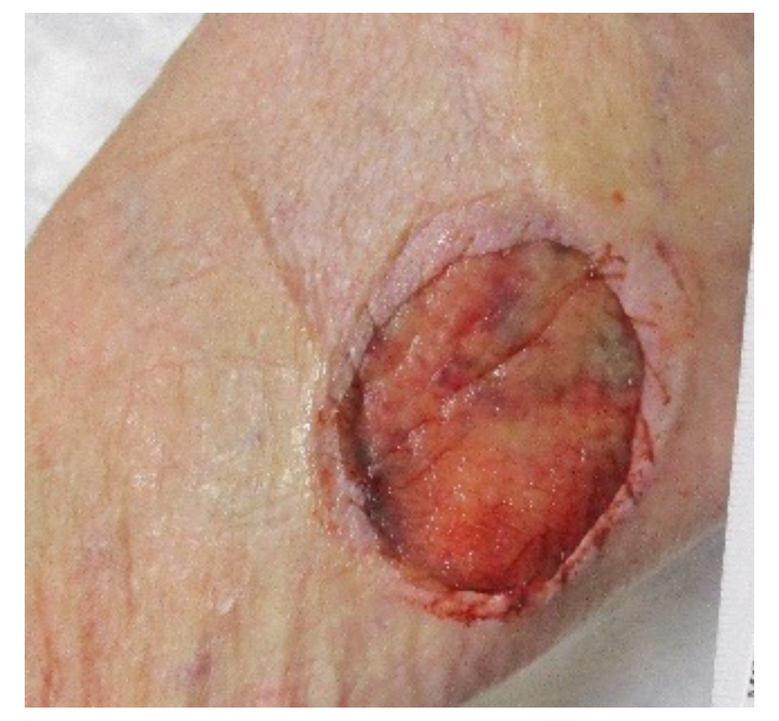

Week 1 status post excision and application of kerecis

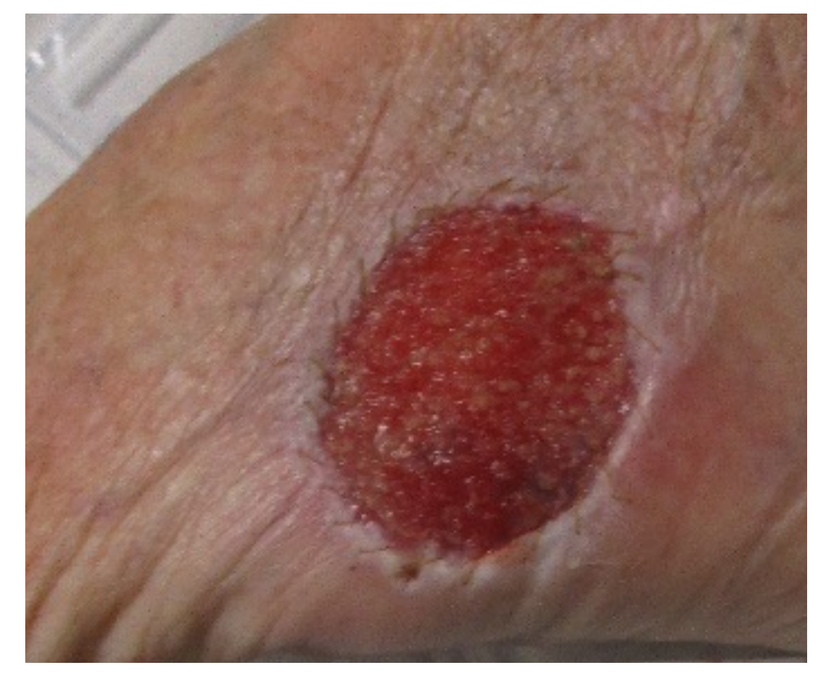

Week 3 status post excision and application of kerecis

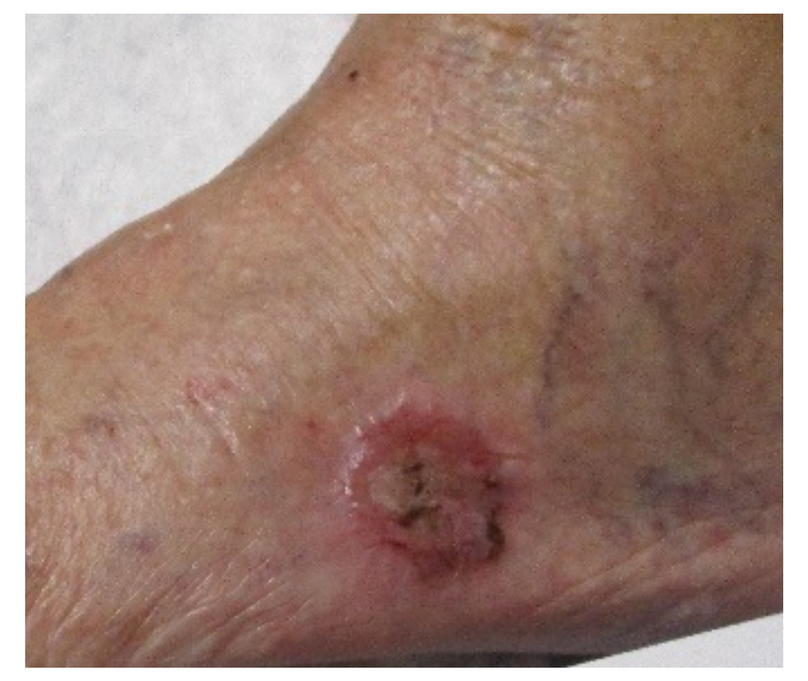

Week 5 Fully healed

CASE 3

LOWER BACK BASAL CELL CARCINOMA

Kerecis Applications: 1
Patient Outcomes: Wound allowed to heal per patient preference and was declared fully closed by post operative day 61.

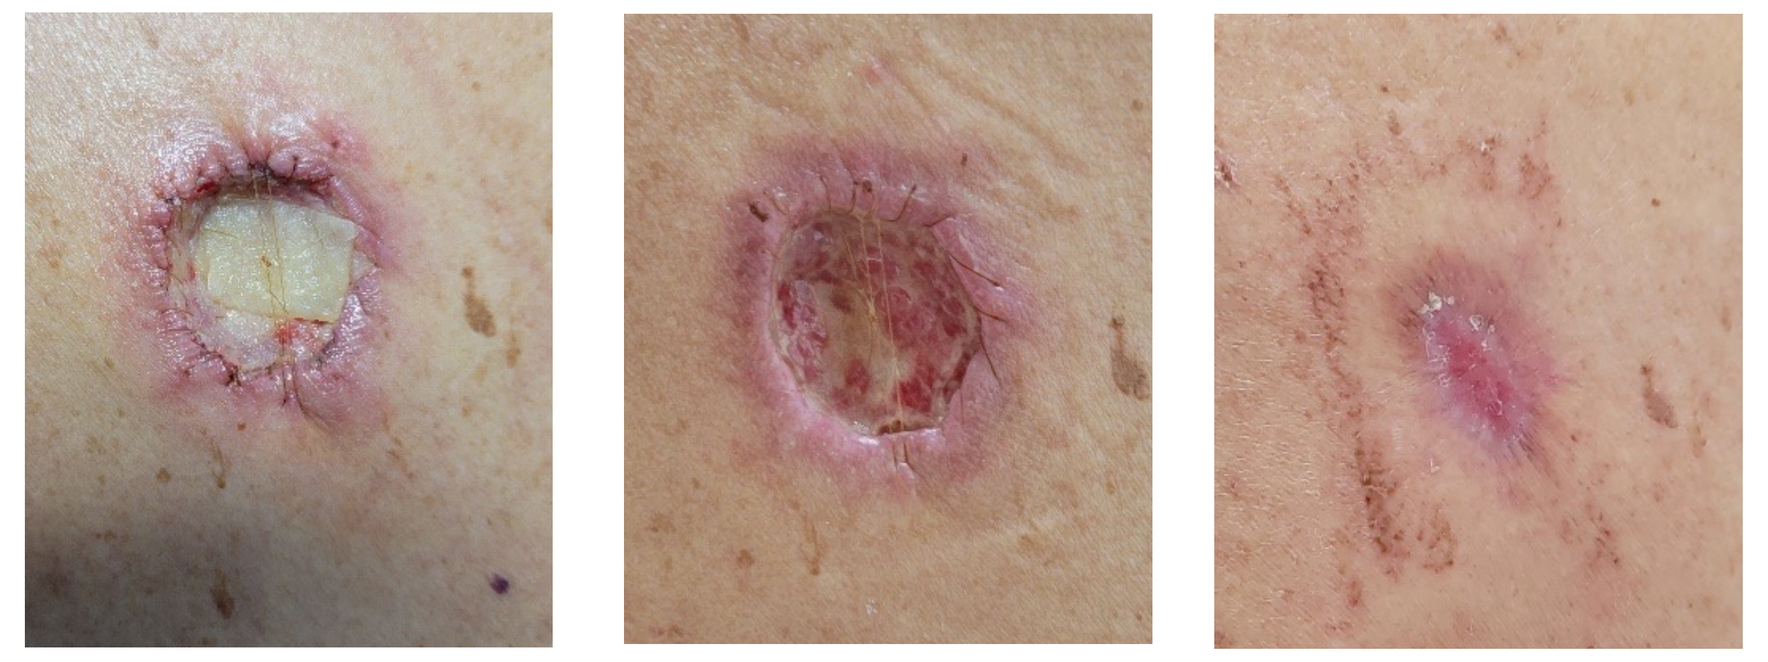

CASE 4

NASAL TIP SKIN CANCER RECONSTRUCTED WITH COMBINATION OF DORSAL NASAL FLAP AND KERECIS

Kerecis Applications: 1
Patient Outcomes: Reconstruction of Moh's defect of nasal tip via dorsal nasal flap and application of kerecis allowed to then heal by secondary intent within 24ays of surgery

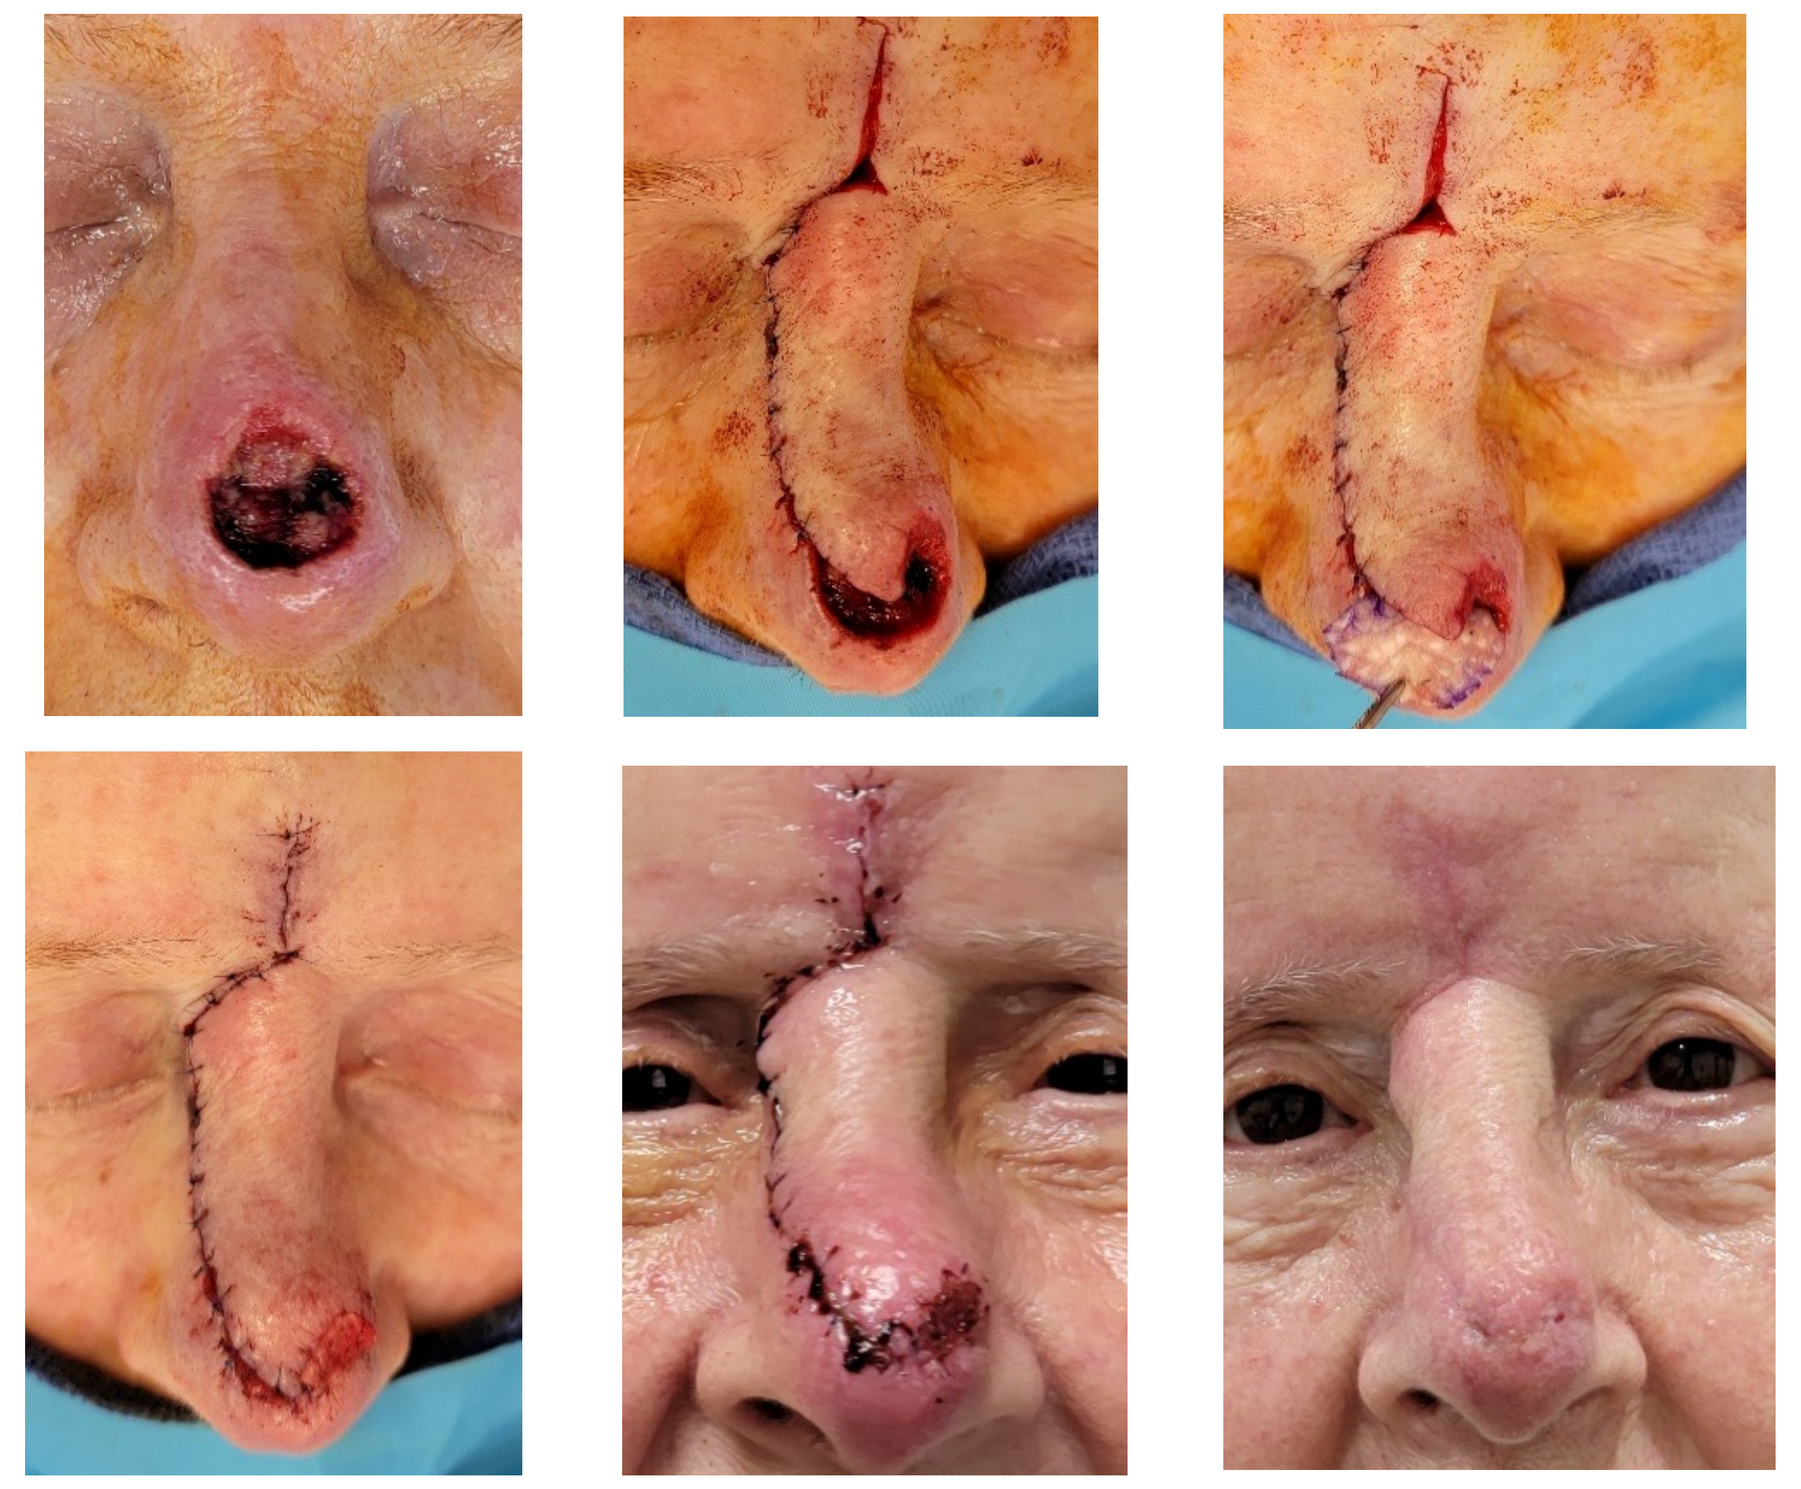

Patient	Patient Age	Primary Diagnosis	Defect Size	Defect Location	Closure method	Time to complete closure
1	77	Basal Cell Carcinoma	5x6cm	Central Chest Wall	STSG	28 days
2	77 66	Basal Cell Carcinoma	7x5cm	Dorsal foot	STSG	37 days
3	81	Basal Cell Carcinoma	3x5x2cm	Lower lateral back	Secondary Intent	61 days
4	82	Basal Cell Carcinoma	2x3cm	Nasal Tip	Flap with Kerecis	24 days
5	66	Squamous Cell Carcinoma	6x7x1.5cm	Left arm	STSG with partial closure	24 days
6	86	Metastatic vulvar ca	9x9x2cm	Right hip	STSG	14 days

RESULTS

All six patients' wounds were successfully reconstructed using STSG, flap or healing by secondary intent. Of the 4 patients who underwent staged closure 3 ultimately progressed to skin graft and 1 patient was allowed to heal by secondary intent per patient preference. In the patients who underwent Mohs excision flap surgeries were necessary for closure with FSG being successfully used to extend the reach of the flap thereby mitigating further donor site morbidity in 1 case and reconstruct the donor site in the other. No FSG, STSG, or flap failure occurred. All patients were satisfied with their results, and no further revisions were needed.

CONCLUSIONS

Cancerous lesions are highly prevalent with closure and cosmesis being equitable aims during excision and reconstruction. Wounds amenable to skin graft may therefore warrant using FSG to avoid graft failure and optimize cosmetic results. FSG is an excellent tool for staging reconstruction and provides coverage while pathology is being analyzed while advancing the reconstructive goals. Rapid integration is a novel strength of FSG over other ADMs, reducing delays and augmenting definitive closure. In this small population of patients, FSG proved to be an excellent tool for limiting additional surgeries and donor site morbidities even with definitive flap closure. More extensive prospective studies are needed to assess and validate these findings.

*Kerecis™, Kerecis, Isafjordur, Iceland