The Healing Power of Nature

Fish Skin Graft* A promising treatment for managing anal fistula pre and postop complications

Michael S. Romberg[1], MD, FACS,

[1]SAS Surgical, Ltd. Mokena, IL, US

INTRODUCTION

Anal fistulas are highly prevalent and are a significant global burden associated with morbidity and mortality[1]. The exact causality is not entirely understood, but pathological lower digestive tract and anal conditions are predictors[1]. Patients experience pain, fever, malaise, drainage, and incontinence which contribute to a decreased quality of life. Many treatment options are available to the surgeon, including endorectal advancement flap (ERAF), seton, ligation of the intersphincteric fistula
tract (LIFT), fibrin glues, biological fistula plug, adipose-derived stem cells (ASCs), video-assisted anal fistula treatment (VAAFT), and laser technology1. However, the reoccurrence rate and failure to preserve the sphincter with these procedures remains high (30-50%), often resulting in multiple procedures, all of which contribute to decreased quality of life[2]. As such, new methodologies are needed for the treatment of anal fistulas. Fish skin graft (FSG) is a relevantly new biologic that is minimally processed and inherently low immunogenicity with preserved mechanical and biological components[3]. FSG allows rapid cellular ingrowth resulting in faster healing rates and less contracture when compared to mammalian tissues[4,5].

METHODS

Two male patients (n=2) presented with anal fistulas and underwent fistulectomy. Patient one (38-year-old male) and Patient two (50-year-old male) presented with intersphincteric and complicated transphincteric fistulas, respectively, that were treated operatively. Both patients underwent excision and were treated with fragmented FSG packed into the sinus at the time of excision.

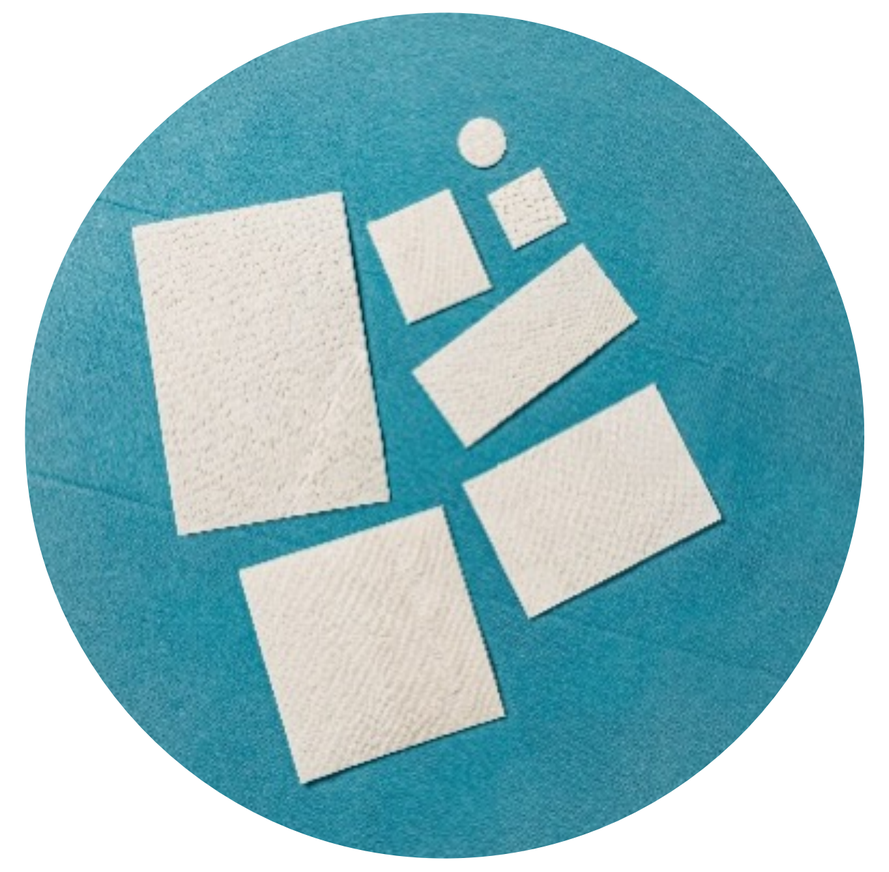

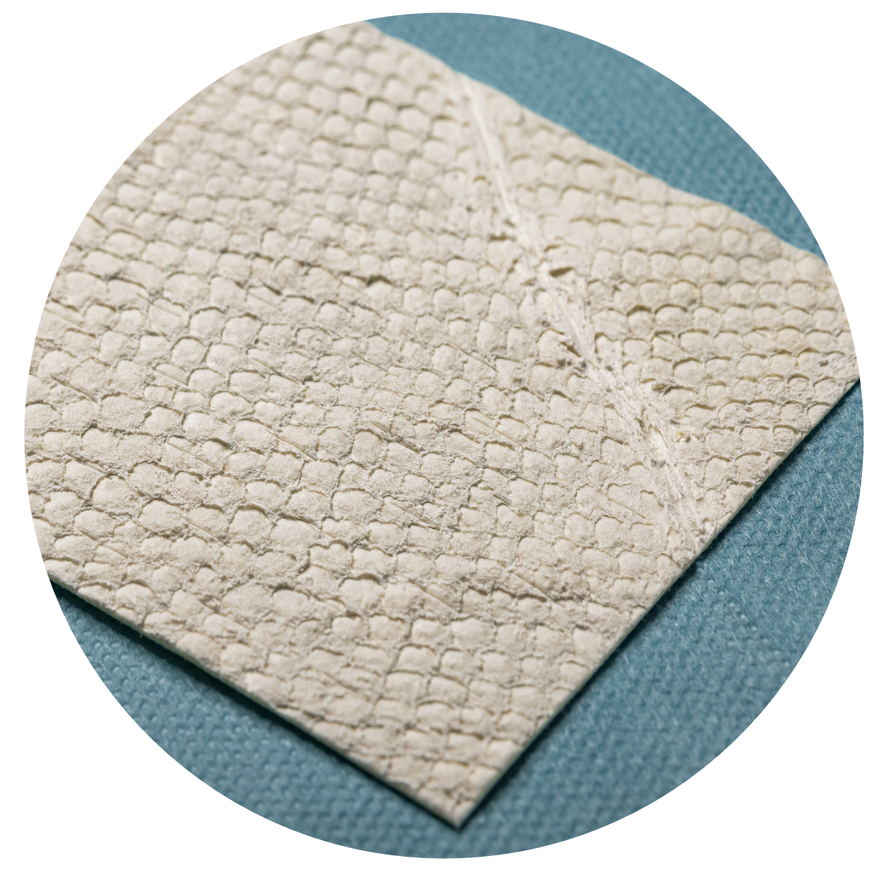

*Kerecis™, Kerecis, Isafjordur, Iceland

CASE 1 38-YEAR-OLD MALE

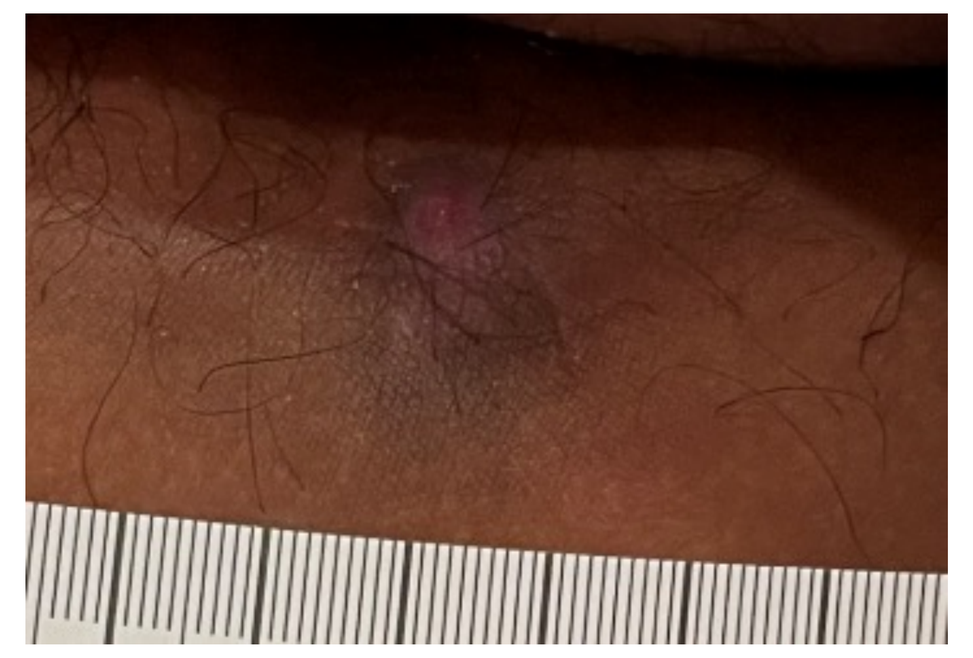

Initial presentation

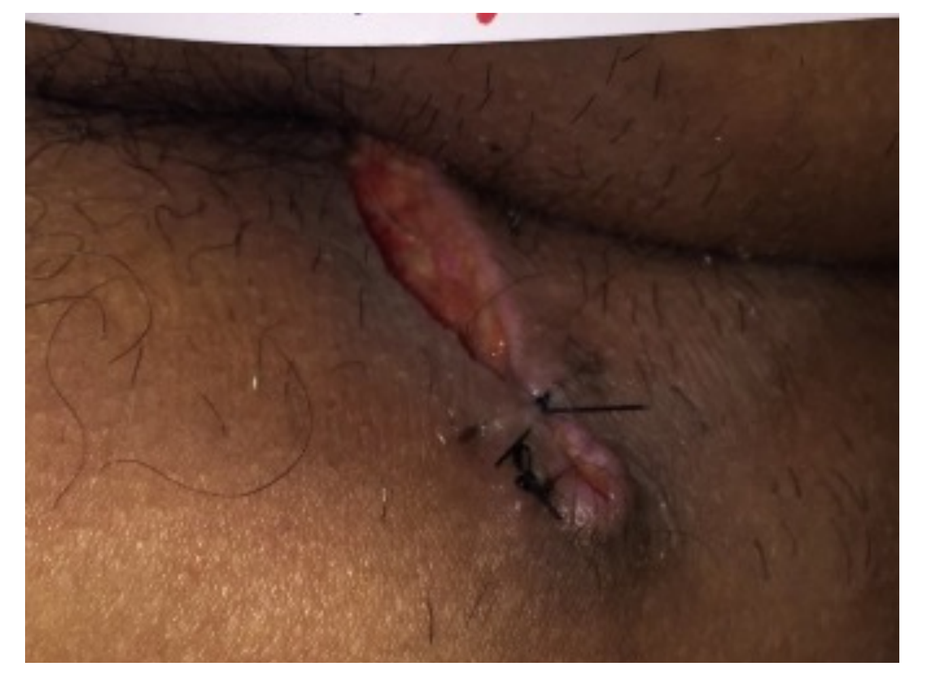

7 days post op
FISTULECTOMY with application of fragmented fish skin graft
Wound is 4 x 1 x 1 cm.

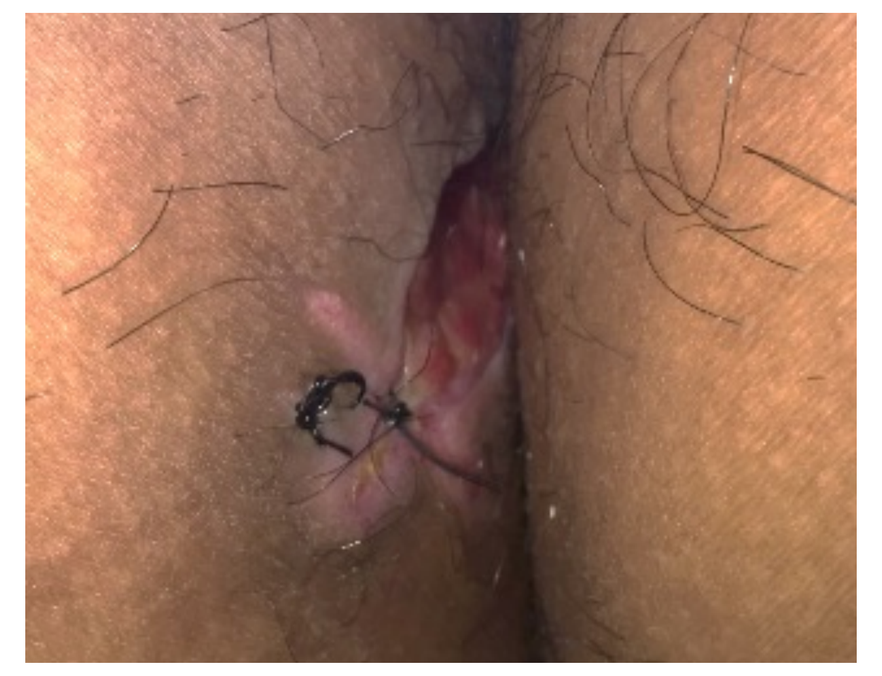

18 days post op

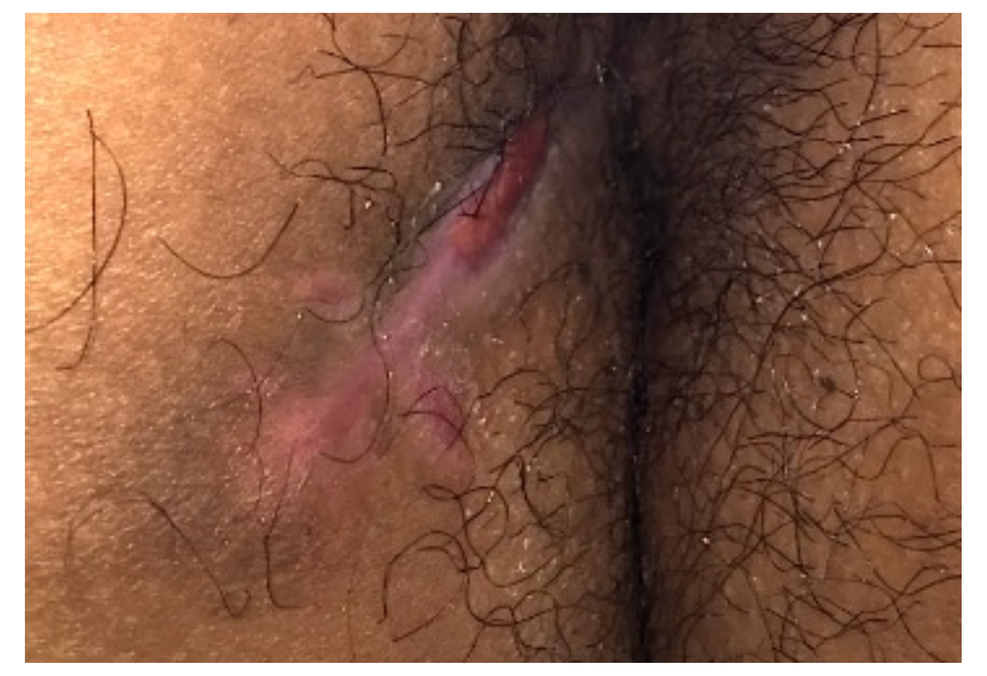

39 days post op
No depth to wound

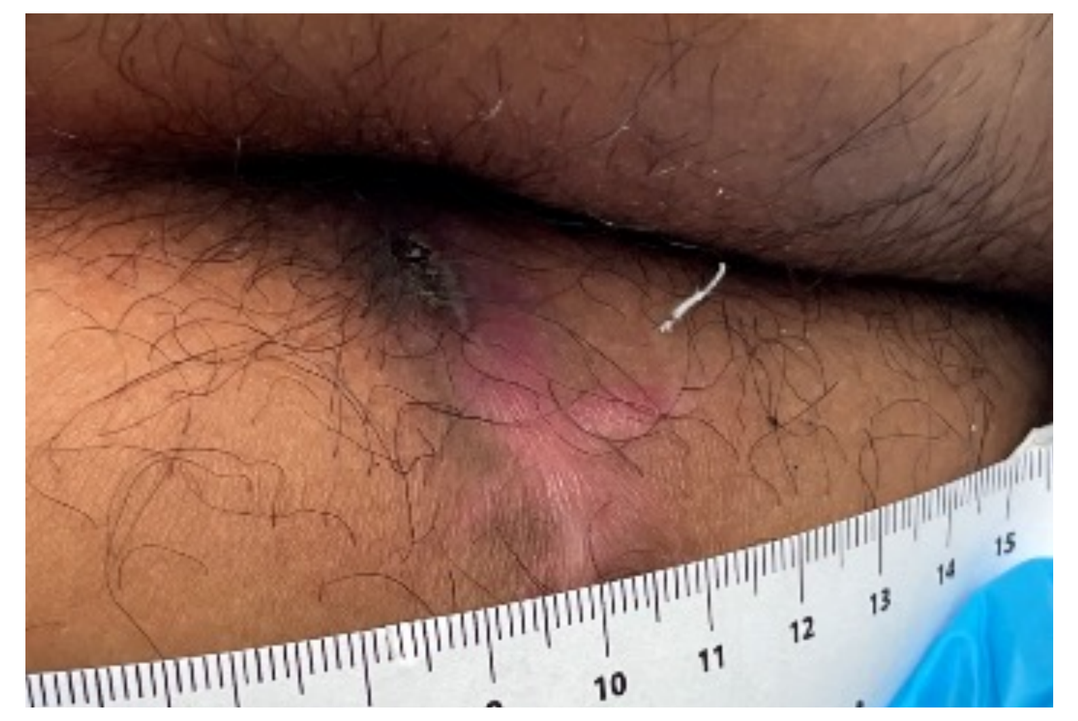

67 days post op
Wound bed almost closed

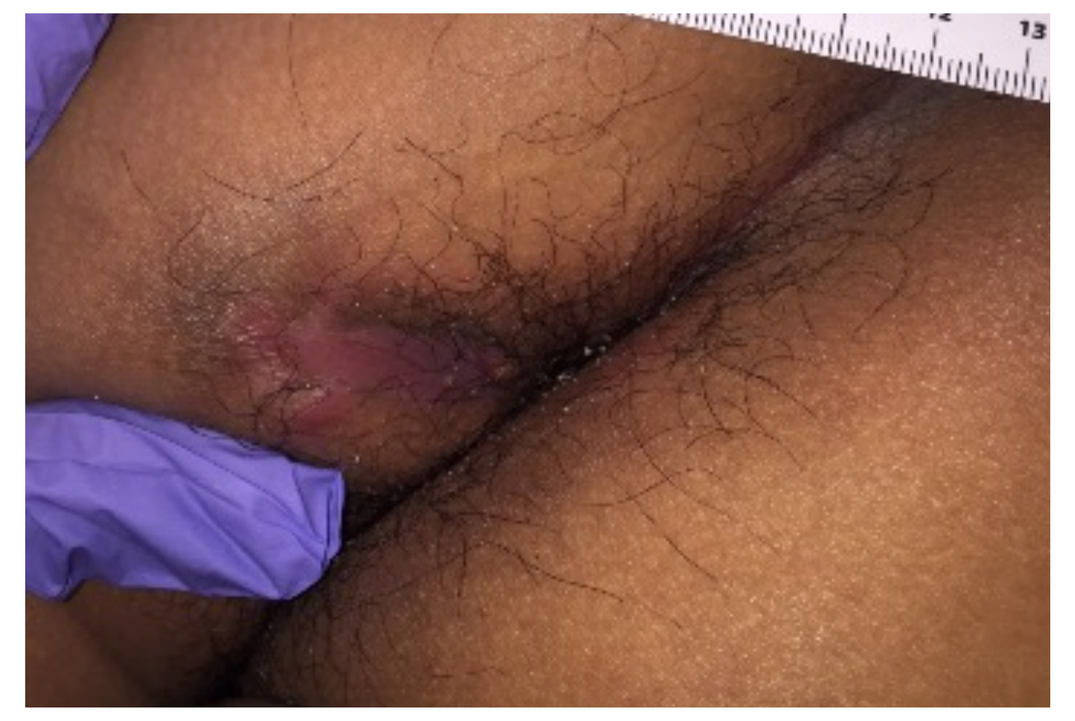

81 days post op
Has a small subcutaneous cavity

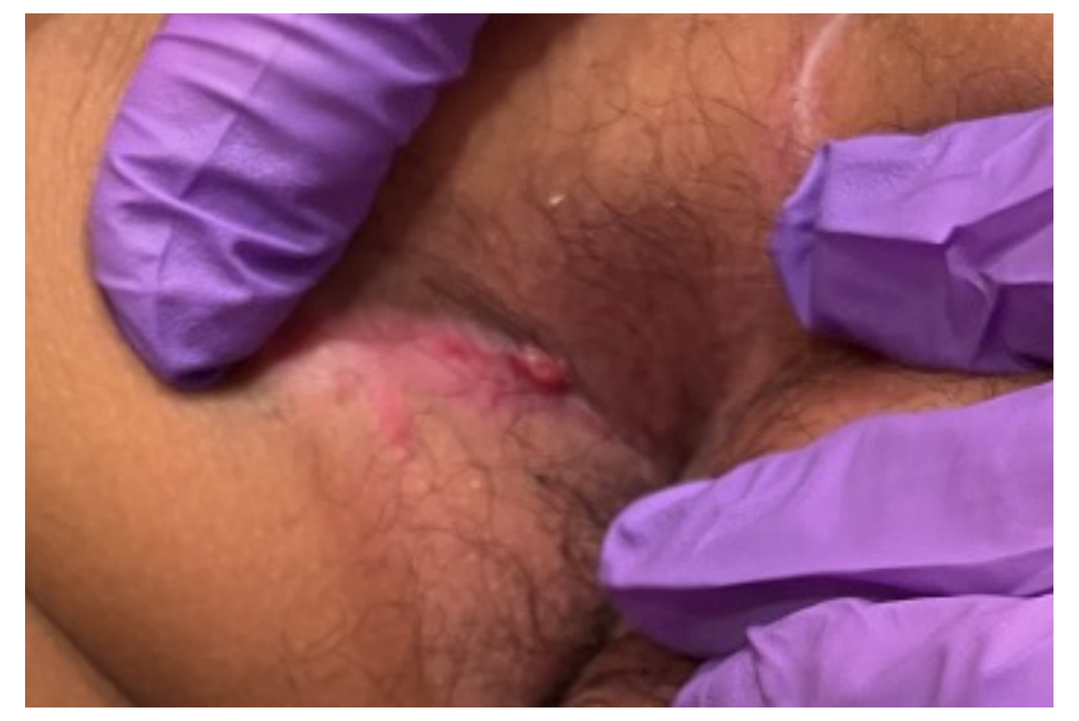

95 days post op

CASE 2 50-YEAR-OLD MALE

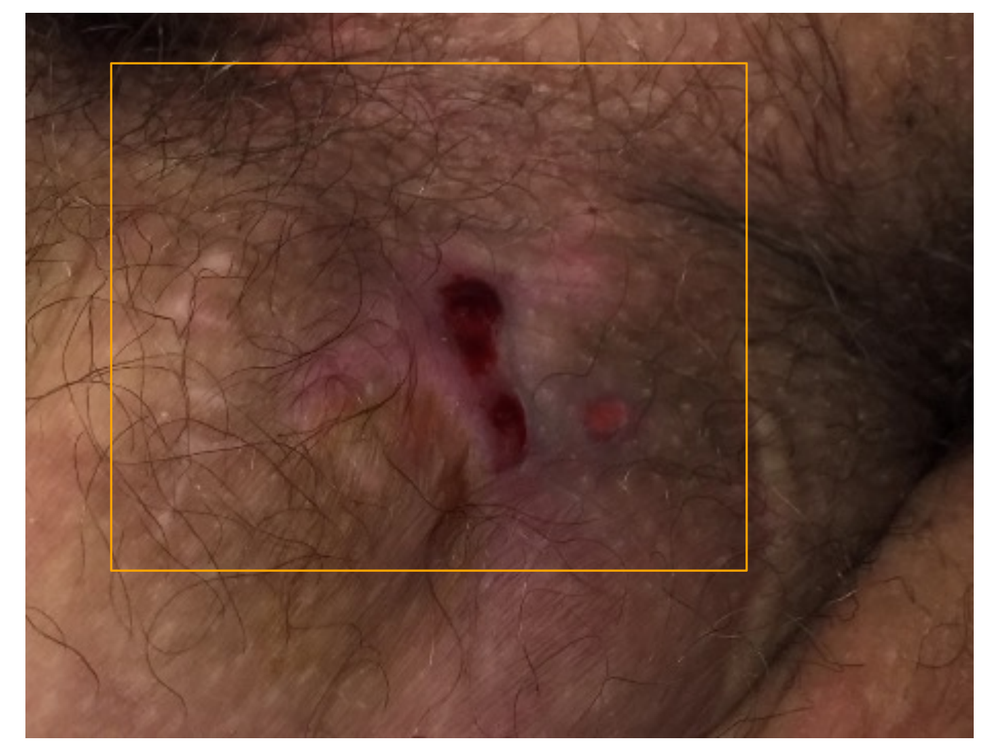

Initial presentation

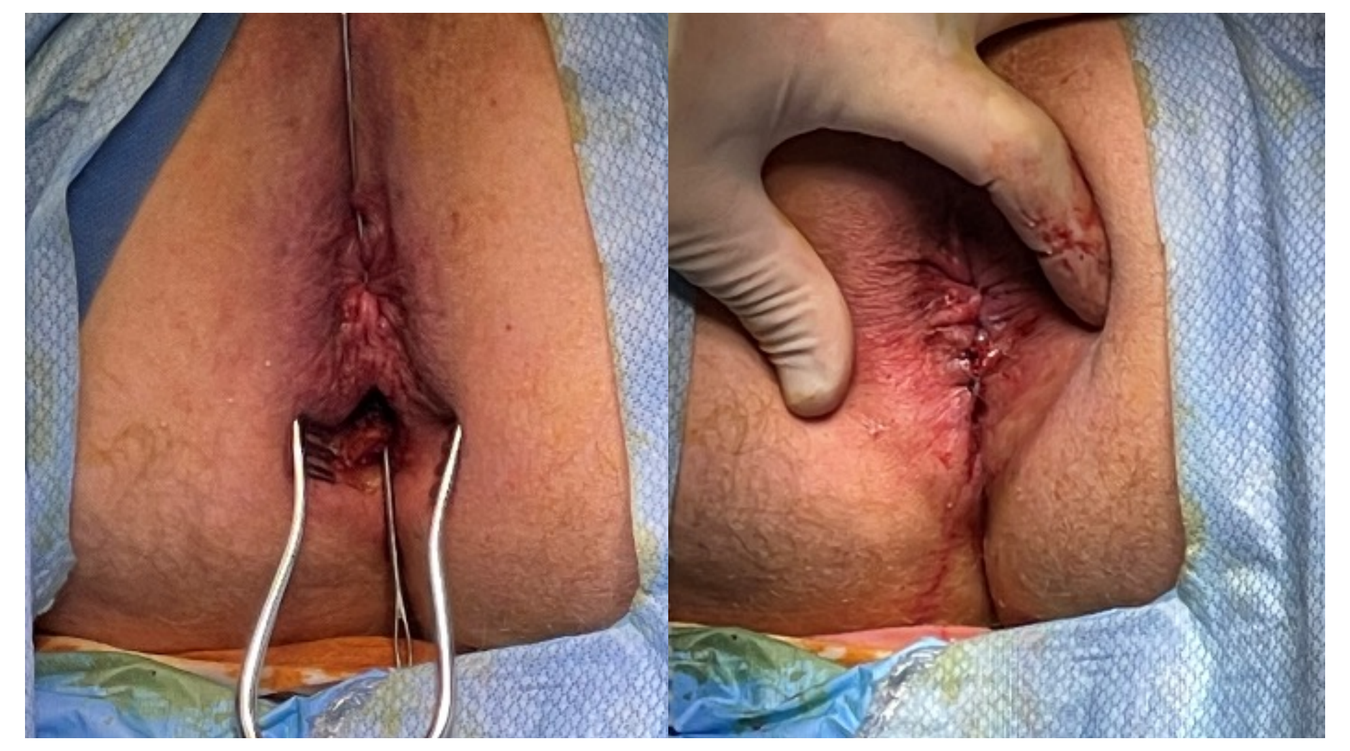

Fistulectomy

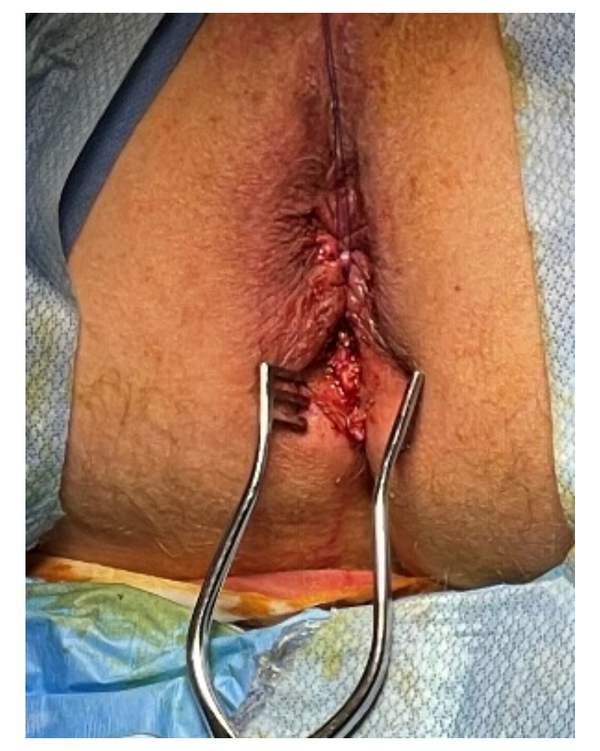

Used Kerecis particulate

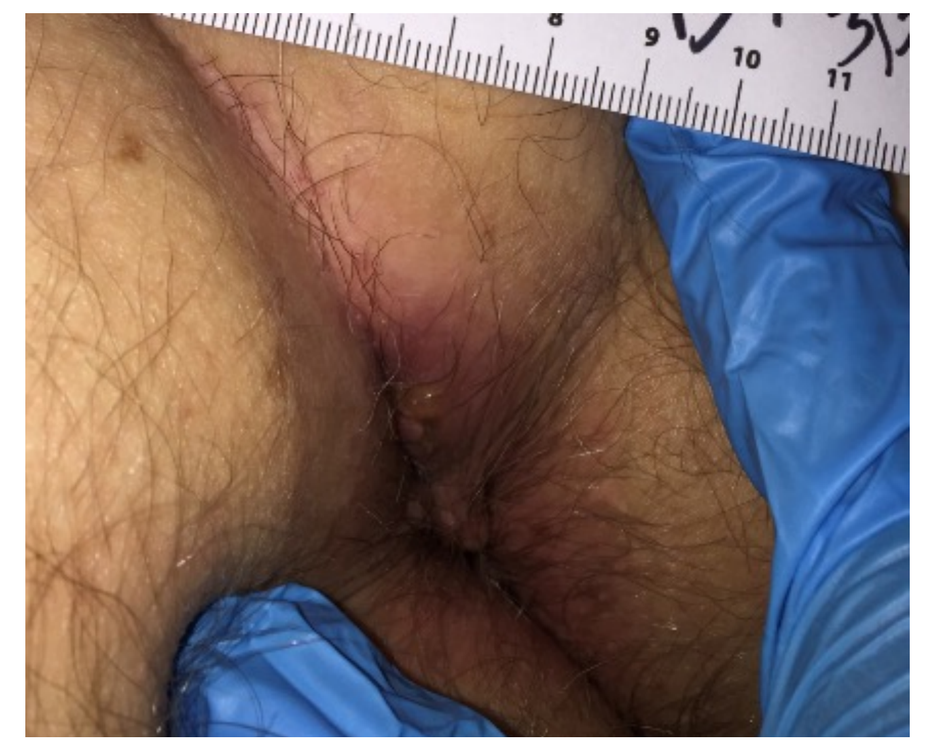

14 days post op
wound 2 x 0.5 cm

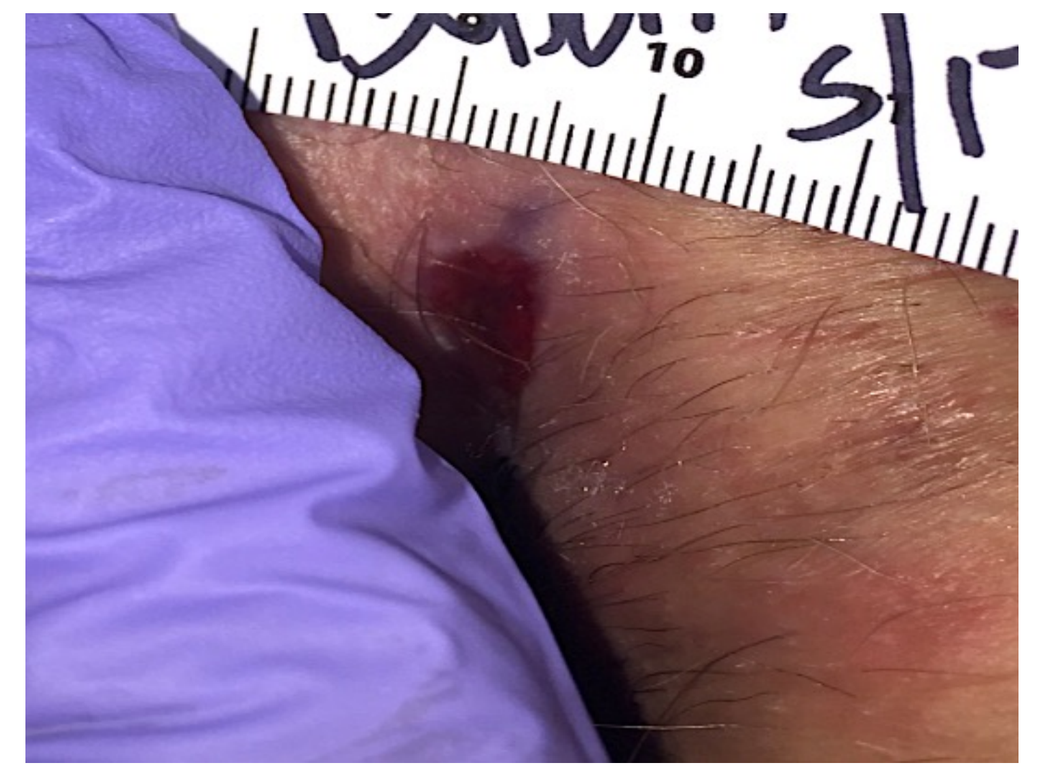

31 days post op
wound 2 x 0.5 cm

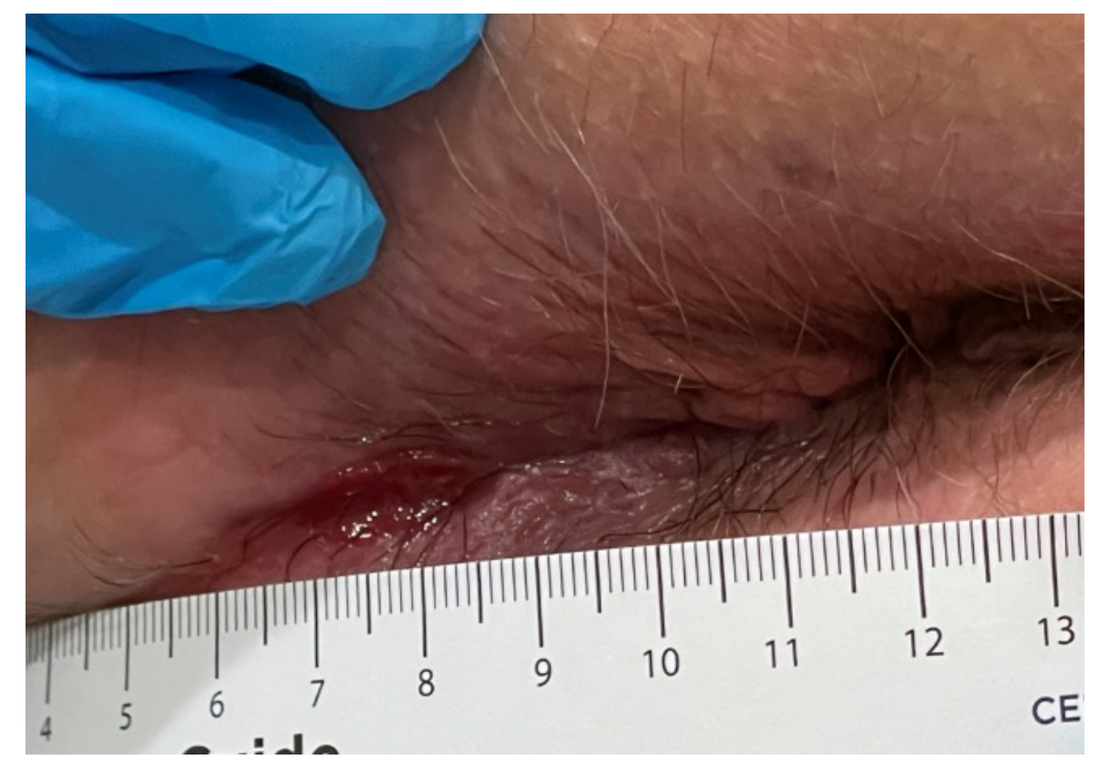

45 days post op
wound 1.5 x 0.5 cm

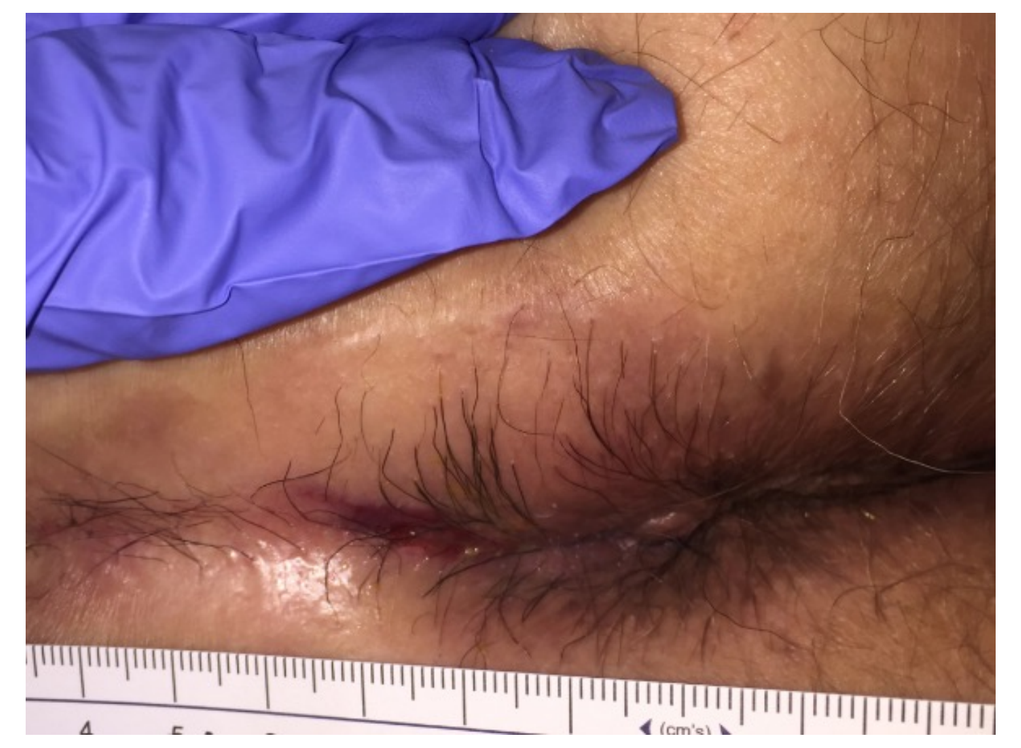

73 days post op

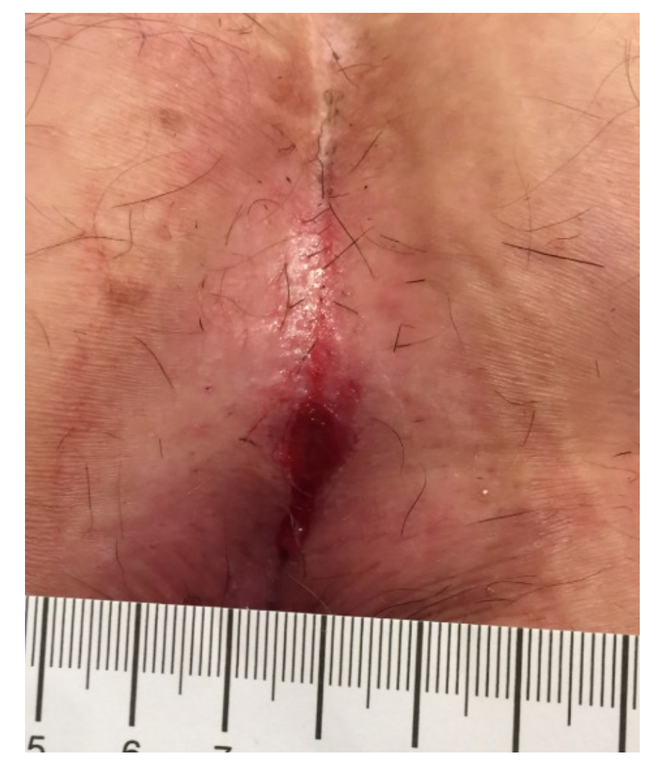

85 days post op

RESULTS

Patient one showed no signs of erythema, and drainage was minimal. The incision displayed advanced healing seven days postop. On day eighteen, continued healing was observed with no complications to report. By day thirty-nine, the incision was fully healed and remained complication-free. The patient was followed prospectively for an additional two months, and the surgical excision and repair were preserved. On day fourteen, Patient two presented with remarkable incisional regeneration with little drainage or pain. By day forty-five, the patient was healed and presented with no complications.

CONCLUSION

Anal Fistulas have a high prevalence and incidence rate, with epidemiological studies estimating an overall prevalence in European countries at 18.37 (95% CI: 18.20-18.55%) per 100,000 individuals[6]. Surgeons have a plethora of treatment options that are guided by severity and etiology. The mainstay surgical treatment goals are to manage pain, secondary infection, and sepsis, promote sinus tract healing and preserve sphincter function and continence mechanism[1]. With postop reoccurrence rates high, there is a need for new treatment options. FSG in this small population appears to attenuate inflammation and secondary infection while rapidly regenerating the sinus tract. FSG has inherent Omega3 fatty acids, low immunogenicity, and is homologous to the human dermis in mechanical and biological content. FSG is an advanced biological scaffold that may reduce inflammation and drainage while rapidly regenerating the anoderm, reducing infection, and preserving sphincter function.

References

1. Limura E, Giordano P. Modern management of anal fistula. World J Gastroenterol. 2015 Jan 7;21(1):12-20. doi: 10.3748/wjg.v21.i1.12. PMID: 25574077; PMCID: PMC4284327.
2. Dudukgian H, Abcarian H. Why do we have so much trouble treating anal fistula? World J Gastroenterol. 2011 Jul 28;17(28):3292-6. doi: 10.3748/wjg.v17.i28.3292. PMID: 21876616; PMCID: PMC3160532.
3. Baldursson BT, Kjartansson H, Konrádsdóttir F, Gudnason P, Sigurjonsson GF, Lund SH. Healing rate and autoimmune safety of full-thickness wounds treated with fish skin acellular dermal matrix versus porcine small-intestine submucosa: a noninferiority study. Int J Low Extrem Wounds. 2015 Mar;14(1):37-43. doi: 10.1177/1534734615573661. Epub 2015 Mar 9. PMID: 25759413.
4. Magnusson S, Baldursson BT, Kjartansson H, Rolfsson O, Sigurjonsson GF. Regenerative and Antibacterial Properties of Acellular Fish Skin Grafts and Human Amnion/Chorion Membrane: Implications for Tissue Preservation in Combat Casualty Care. Mil Med. 2017 Mar;182(S1):383-388. doi: 10.7205/MILMED-D-16-00142. PMID: 28291503.
5. Stone R 2nd, Saathoff EC, Larson DA, Wall JT, Wienandt NA, Magnusson S, Kjartansson H, Natesan S, Christy RJ. Accelerated Wound Closure of Deep Partial Thickness Burns with Acellular Fish Skin Graft. Int J Mol Sci. 2021 Feb 4;22(4):1590. doi: 10.3390/ijms22041590. PMID: 33557424; PMCID: PMC7915828.