B0386

Real-World Clinical Effectiveness and Safety of Vedolizumab and Adalimumab in Biologic-Naïve Patients With Crohn's Disease: Results From the EVOLVE Study

Andres Yarur,[1] Brian Bressler,[2] Neil R. Brett,[3] Marielle Bassel,[3] Shashi Adsul,[4] Pravin Kamble,[5] Gerassimos J. Mantzaris[6]

[1]Cedars Sinai Medical Center, Los Angeles, CA, USA; [2]St. Paul's Hospital, Vancouver, BC, Canada; [3]PPD, part of Thermo Fisher Scientific, Montreal, QC, Canada; [4]Takeda Pharmaceuticals International AG, Zurich, Switzerland; [5]Takeda Pharmaceuticals International, Cambridge, MA, USA; [6]Evangelismos-Polykliniki' General Hospital, Athens, Greece

Background

- Approved advanced therapies for Crohn's disease (CD) include vedolizumab and adalimumab, among others[1]
 - Vedolizumab is a gut-selective humanized monoclonal anti-$\alpha_4\beta_7$ integrin antibody that reduces lymphocyte trafficking into intestinal tissue[2]
 - Adalimumab is an anti-tumor necrosis factor agent
- Data comparing the effectiveness and safety of vedolizumab versus adalimumab as first-line therapy may help to better position biologics in the therapeutic algorithm

Aim

- To evaluate the real-world clinical effectiveness and safety of vedolizumab versus adalimumab in biologic-naïve patients diagnosed with CD

Methods

- The EVOLVE retrospective cohort study examined medical records of patients diagnosed with CD receiving first-line biologic treatment with vedolizumab or adalimumab (date of initiation defined as the index date) between May 2014 and March 2018
- The study population included biologic-naïve adults diagnosed with CD who had ≥6 months of follow-up data after treatment initiation
- Pre-index baseline data collected included patient demographics, clinical and treatment history, and disease severity
- The following clinical outcomes were defined based on the following hierarchical algorithm during the postindex period[3]:
 - ***Clinical response***, defined as a positive change in CD activity index (CDAI) category from baseline (CDAI categories: score of <150, score of 151-219, score of 220-450, score of >450); or if CDAI score unknown, decrease of ≥3 points from baseline in Harvey-Bradshaw index (HBI) score; or if HBI unknown, decrease of ≥3 points from baseline in modified HBI (mHBI) score; or if mHBI unknown, treatment response recorded in medical chart as "complete response" or "partial response"
 - ***Clinical remission,*** defined as a CDAI score of <150 points; or if CDAI unknown, HBI score of ≤4 points; or if HBI unknown, mHBI score of ≤4 points; or if unknown, remission status recorded in medical chart as "in remission"
 - ***Mucosal healing,*** defined as an endoscopic assessment score of 0 or 1 (normal or inactive); or if unknown, a Simple Endoscopic Score for CD of <3, or lack of ulcerations, or other medical chart entries indicating an absence of active disease, inflammatory activity, or pathologic findings following endoscopy
 - ***Treatment persistence,*** defined as patients who did not discontinue their index treatment for any reason during the study follow-up period
- Additional outcomes evaluated were CD-related exacerbations and surgeries, serious adverse events (SAEs), and serious infections (SIs)
- Adjusted analyses were performed using inverse probability weighting (IPW). The baseline covariates included in the IPW model were age, sex, disease location, disease duration, CD-related hospitalizations, disease severity, steroid dependency status, fistula status, and a composite biochemical marker (fecal calprotectin, C-reactive protein, and albumin)

Results

- The demographic and baseline characteristics of patients treated with vedolizumab (n=218) and adalimumab (n=144) are presented in **Table 1**
- After IPW adjustment, patient demographic and baseline characteristics were similar between the treatment groups (standardized difference <0.10; data not shown)

Table 1. Unadjusted baseline characteristics of biologic-naïve patients diagnosed with CD and treated with vedolizumab or adalimumab

Baseline Characteristics	Vedolizumab (n=218)	Adalimumab (n=144)	p Value
Mean (SD) age, y	51.7 (16.8)	40.0 (14.9)	<0.001
Male, n (%)	114 (52.3)	75 (52.1)	0.969
Disease duration, n with available data	176	111	0.016
<2 years, n (%)	50 (28.4)	50 (45.0)	
2 to <5 years, n (%)	32 (18.2)	16 (14.4)	
≥5 years, n (%)	94 (53.4)	45 (40.5)	
Missing, n (%)	42 (19.3)	33 (22.9)	
Median (min-max) observation period, months[a]	15.7 (4.2-45.9)	19.3 (6.1-49.3)	<0.001
CD location at index, n with available data	196	121	0.047
Colonic with/without upper GI disease, n (%)	42 (21.4)	35 (28.9)	
Illeal with/without upper GI disease, n (%)	85 (43.4)	36 (29.8)	
Illeocolonic with/without upper GI disease, n (%)	69 (35.2)	50 (41.3)	
Unknown, n (%)	22 (10.1)	23 (16.0)	
Disease severity at index, n with available data	180	116	0.161
Moderate, n (%)	84 (46.7)	58 (50.0)	
Severe, n (%)	17 (9.4)	17 (14.7)	
Pre-index CD-related hospitalizations, n (%)			
Yes	22 (10.1)	14 (9.7)	0.909
No	196 (89.9)	130 (90.3)	
Composite biochemical marker,[b] n (%)			0.382
Within normal range	41 (18.8)	21 (14.6)	
Outside of normal range	50 (22.9)	50 (34.7)	
Unknown	32 (14.7)	16 (11.1)	
Steroid dependency status, n (%)			0.004
Dependent	32 (14.7)	22 (15.3)	
Intolerant	6 (2.8)	0	
Not dependent or intolerant	86 (39.4)	37 (25.7)	
Not dependent, intolerant, or refractory	47 (21.6)	54 (37.5)	
Refractory	5 (2.3)	2 (1.4)	
Unknown	42 (19.3)	29 (20.1)	
Active fistulae prior to index event, n (%)			0.052
Yes	8 (3.7)	14 (9.7)	
No	181 (83.0)	109 (75.7)	
Unknown	29 (13.3)	21 (14.6)	
Disease behavior, n (%)			0.371
Nonstricturing, nonpenetrating, with or without perianal disease	92 (42.2)	53 (36.8)	
Penetrating, with or without perianal disease	17 (7.8)	9 (6.3)	
Stricturing, with or without perianal disease	45 (20.6)	27 (18.8)	
Unknown	64 (29.4)	55 (38.2)	

CD, Crohn's disease; GI, gastrointestinal; max, maximum; min, minimum.
[a]Although all patients were required to have 6 months of follow-up from time of treatment initiation to data abstraction, some patients were lost to follow-up; therefore, the minimum duration during the observation periods was <6 months.
[b]The composite biochemical marker used a hierarchical algorithm for the 3 baseline biochemical disease indicators, based on data availability, with fecal calprotectin at the top of the hierarchy (fecal calprotectin [≥250 mg/kg] → C-reactive protein [≥5 mg/L] → albumin [<35 g/L]).

- Cumulative treatment persistence, clinical remission, clinical response, and mucosal healing in biologic-naïve patients diagnosed with CD and treated with vedolizumab or adalimumab are shown in **Figure 1**
- Vedolizumab-treated patients were significantly less likely to experience SAEs over 12 months versus adalimumab-treated patients (**Figure 2**)
- There were no statistical differences in CD-related exacerbations, CD-related surgeries, or SIs between the treatment cohorts (**Figure 2**)

Figure 1. Cumulative A) treatment persistence, B) clinical remission, C) clinical response, and D) mucosal healing in biologic-naïve patients diagnosed with CD and treated with vedolizumab or adalimumab

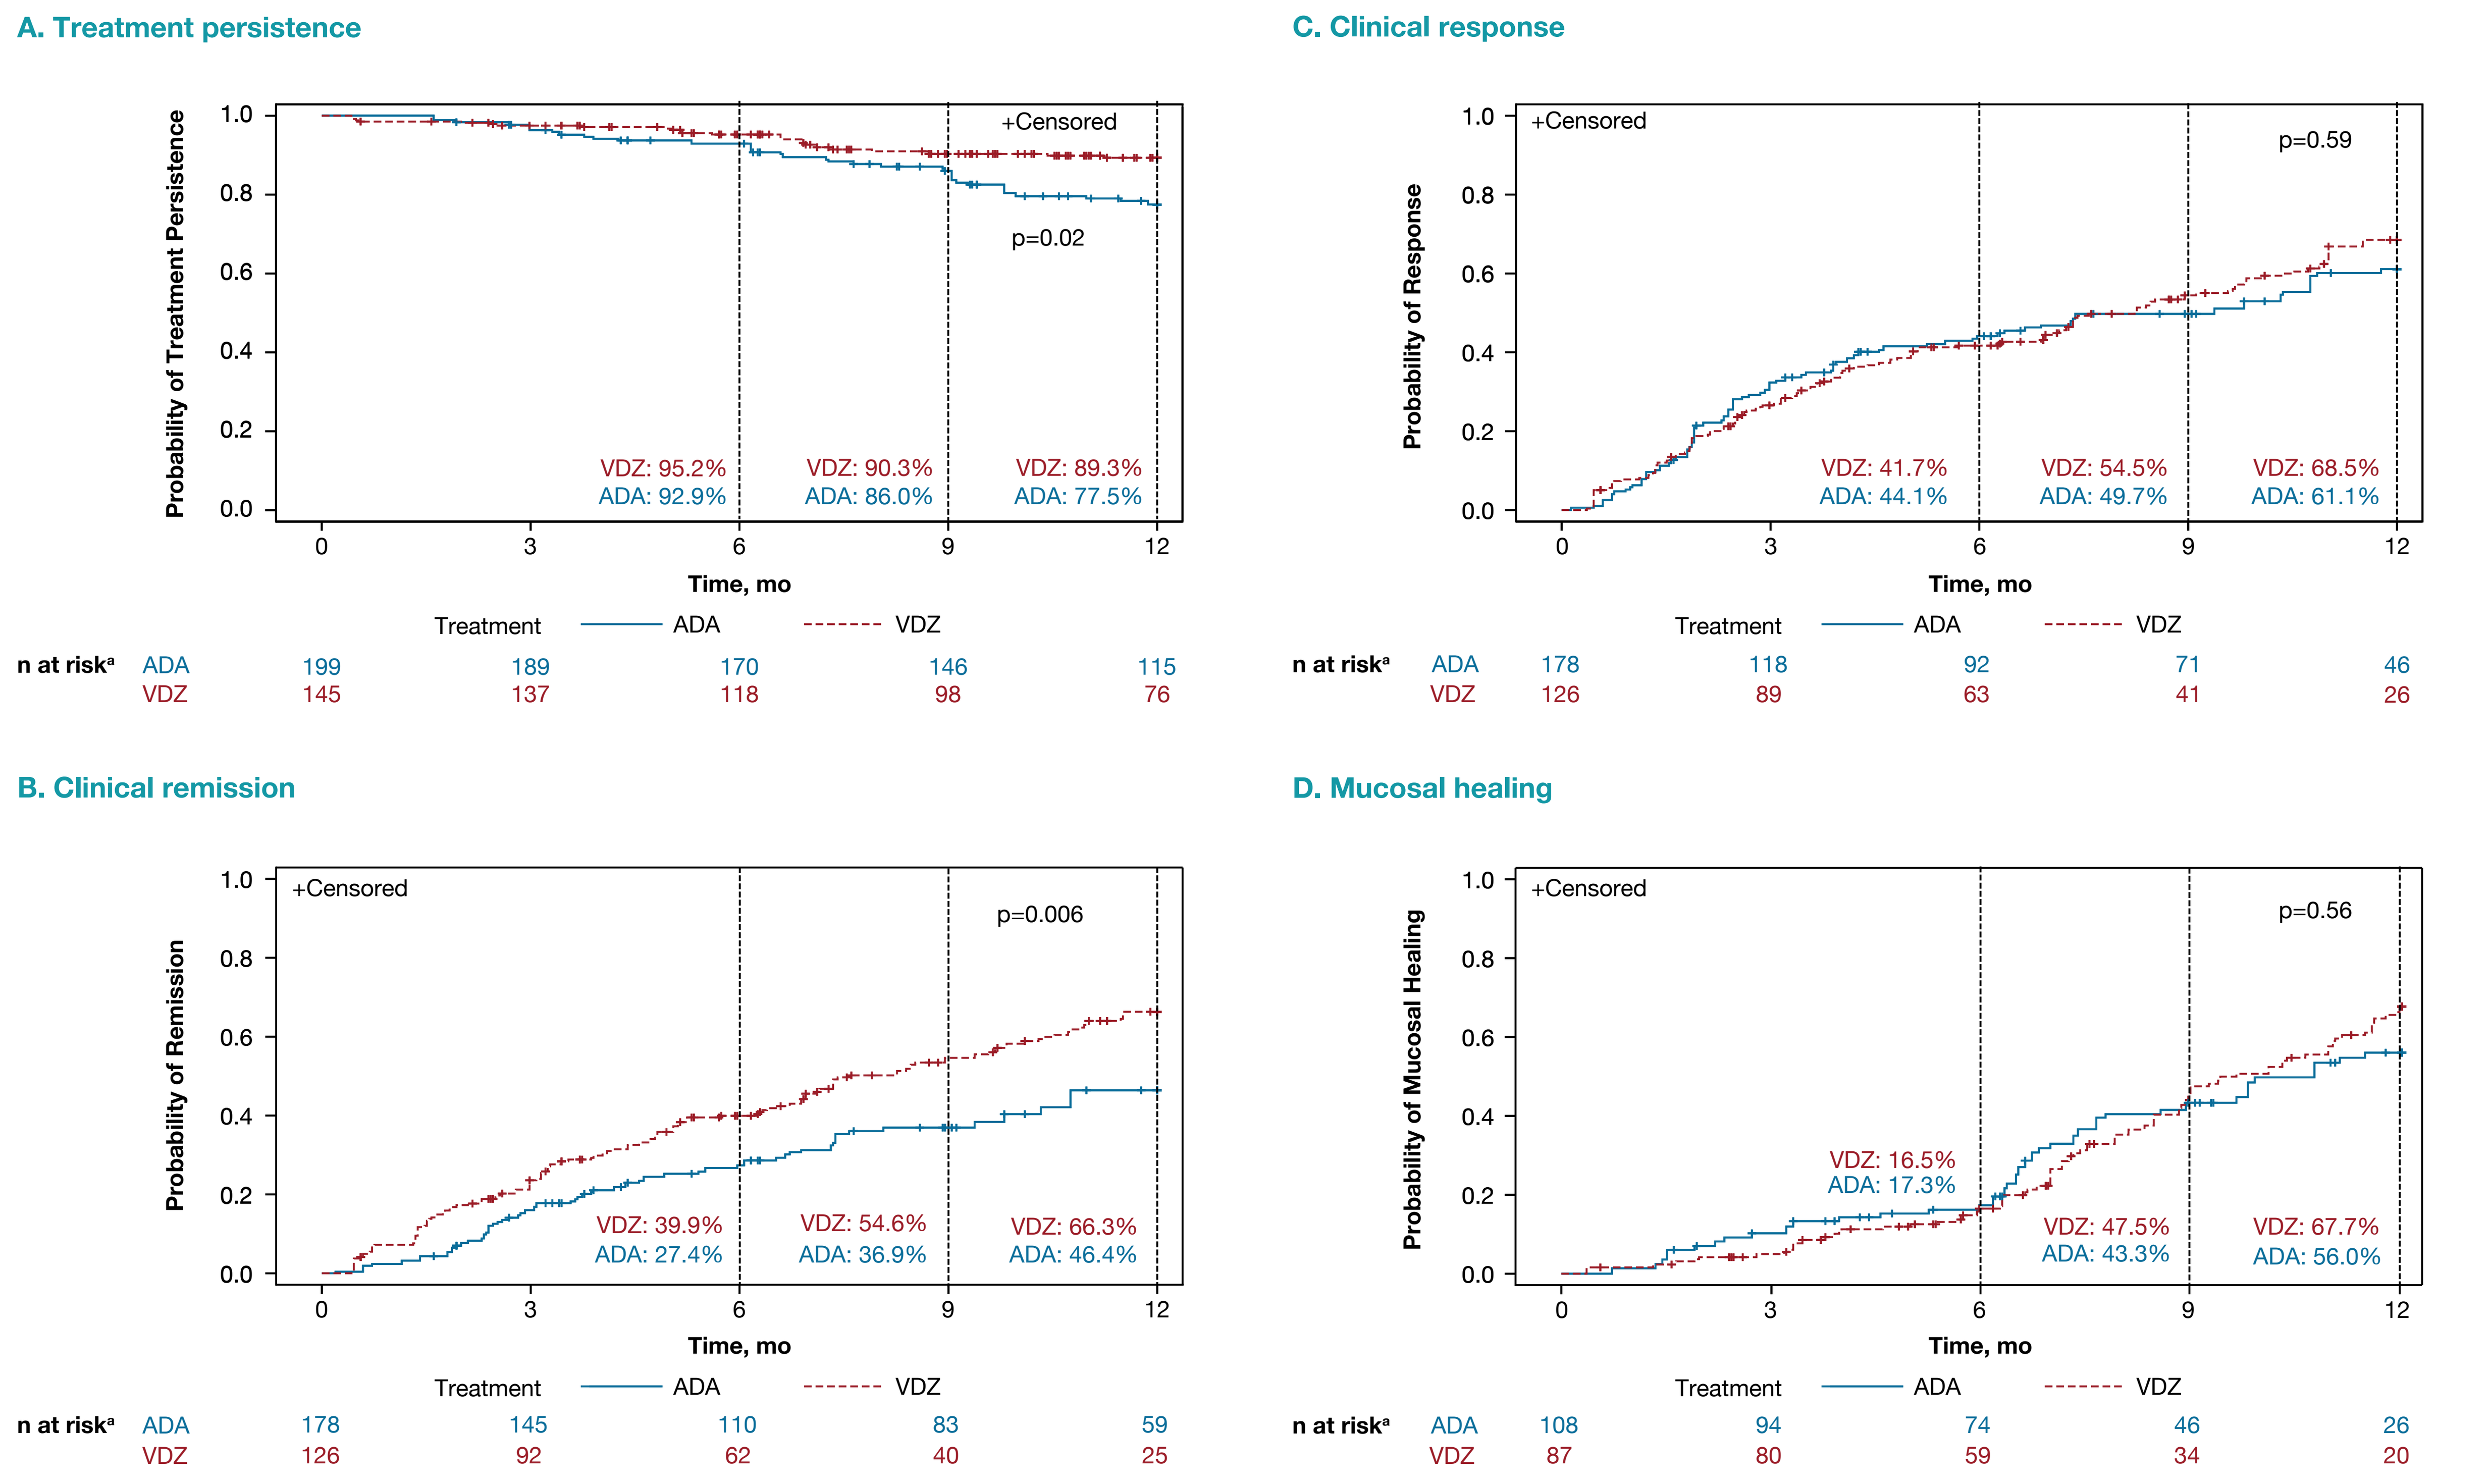
A. Treatment persistence
Probability of Treatment Persistence
+Censored
p=0.02
VDZ: 95.2%
ADA: 92.9%
VDZ: 90.3%
ADA: 86.0%
VDZ: 89.3%
ADA: 77.5%
Time, mo
Treatment ADA VDZ
n at risk[a] ADA 199 189 170 146 115
VDZ 145 137 118 98 76
C. Clinical response
Probability of Response
+Censored
p=0.59
VDZ: 41.7%
ADA: 44.1%
VDZ: 54.5%
ADA: 49.7%
VDZ: 68.5%
ADA: 61.1%
n at risk[a] ADA 178 118 92 71 46
VDZ 126 89 63 41 26
B. Clinical remission
Probability of Remission
+Censored
p=0.006
VDZ: 39.9%
ADA: 27.4%
VDZ: 54.6%
ADA: 36.9%
VDZ: 66.3%
ADA: 46.4%
n at risk[a] ADA 178 145 110 83 59
VDZ 126 92 62 40 25
D. Mucosal healing
Probability of Mucosal Healing
+Censored
p=0.56
VDZ: 16.5%
ADA: 17.3%
VDZ: 47.5%
ADA: 43.3%
VDZ: 67.7%
ADA: 56.0%
n at risk[a] ADA 108 94 74 46 26
VDZ 87 80 59 34 20

p values are log-rank from time-to-event analyses. Inverse probability weighting included the following covariates in the model: age, sex, disease location, disease duration (≤2 years, 2-5 years, 5-10 years, >10 years, and unknown), disease-related hospitalizations, disease severity, steroid dependency status, fistula status, and a composite biochemical marker (fecal calprotectin, C-reactive protein, and albumin).
ADA, adalimumab; CD, Crohn's disease; VDZ, vedolizumab.
[a]The sum of the patient weights for each group of patients still receiving treatment and who have clinical outcomes that can still be assessed.

Figure 2. CD-related exacerbations, surgeries, SAEs, and SIs in biologic-naïve patients with CD treated with VDZ or ADA over 12 months

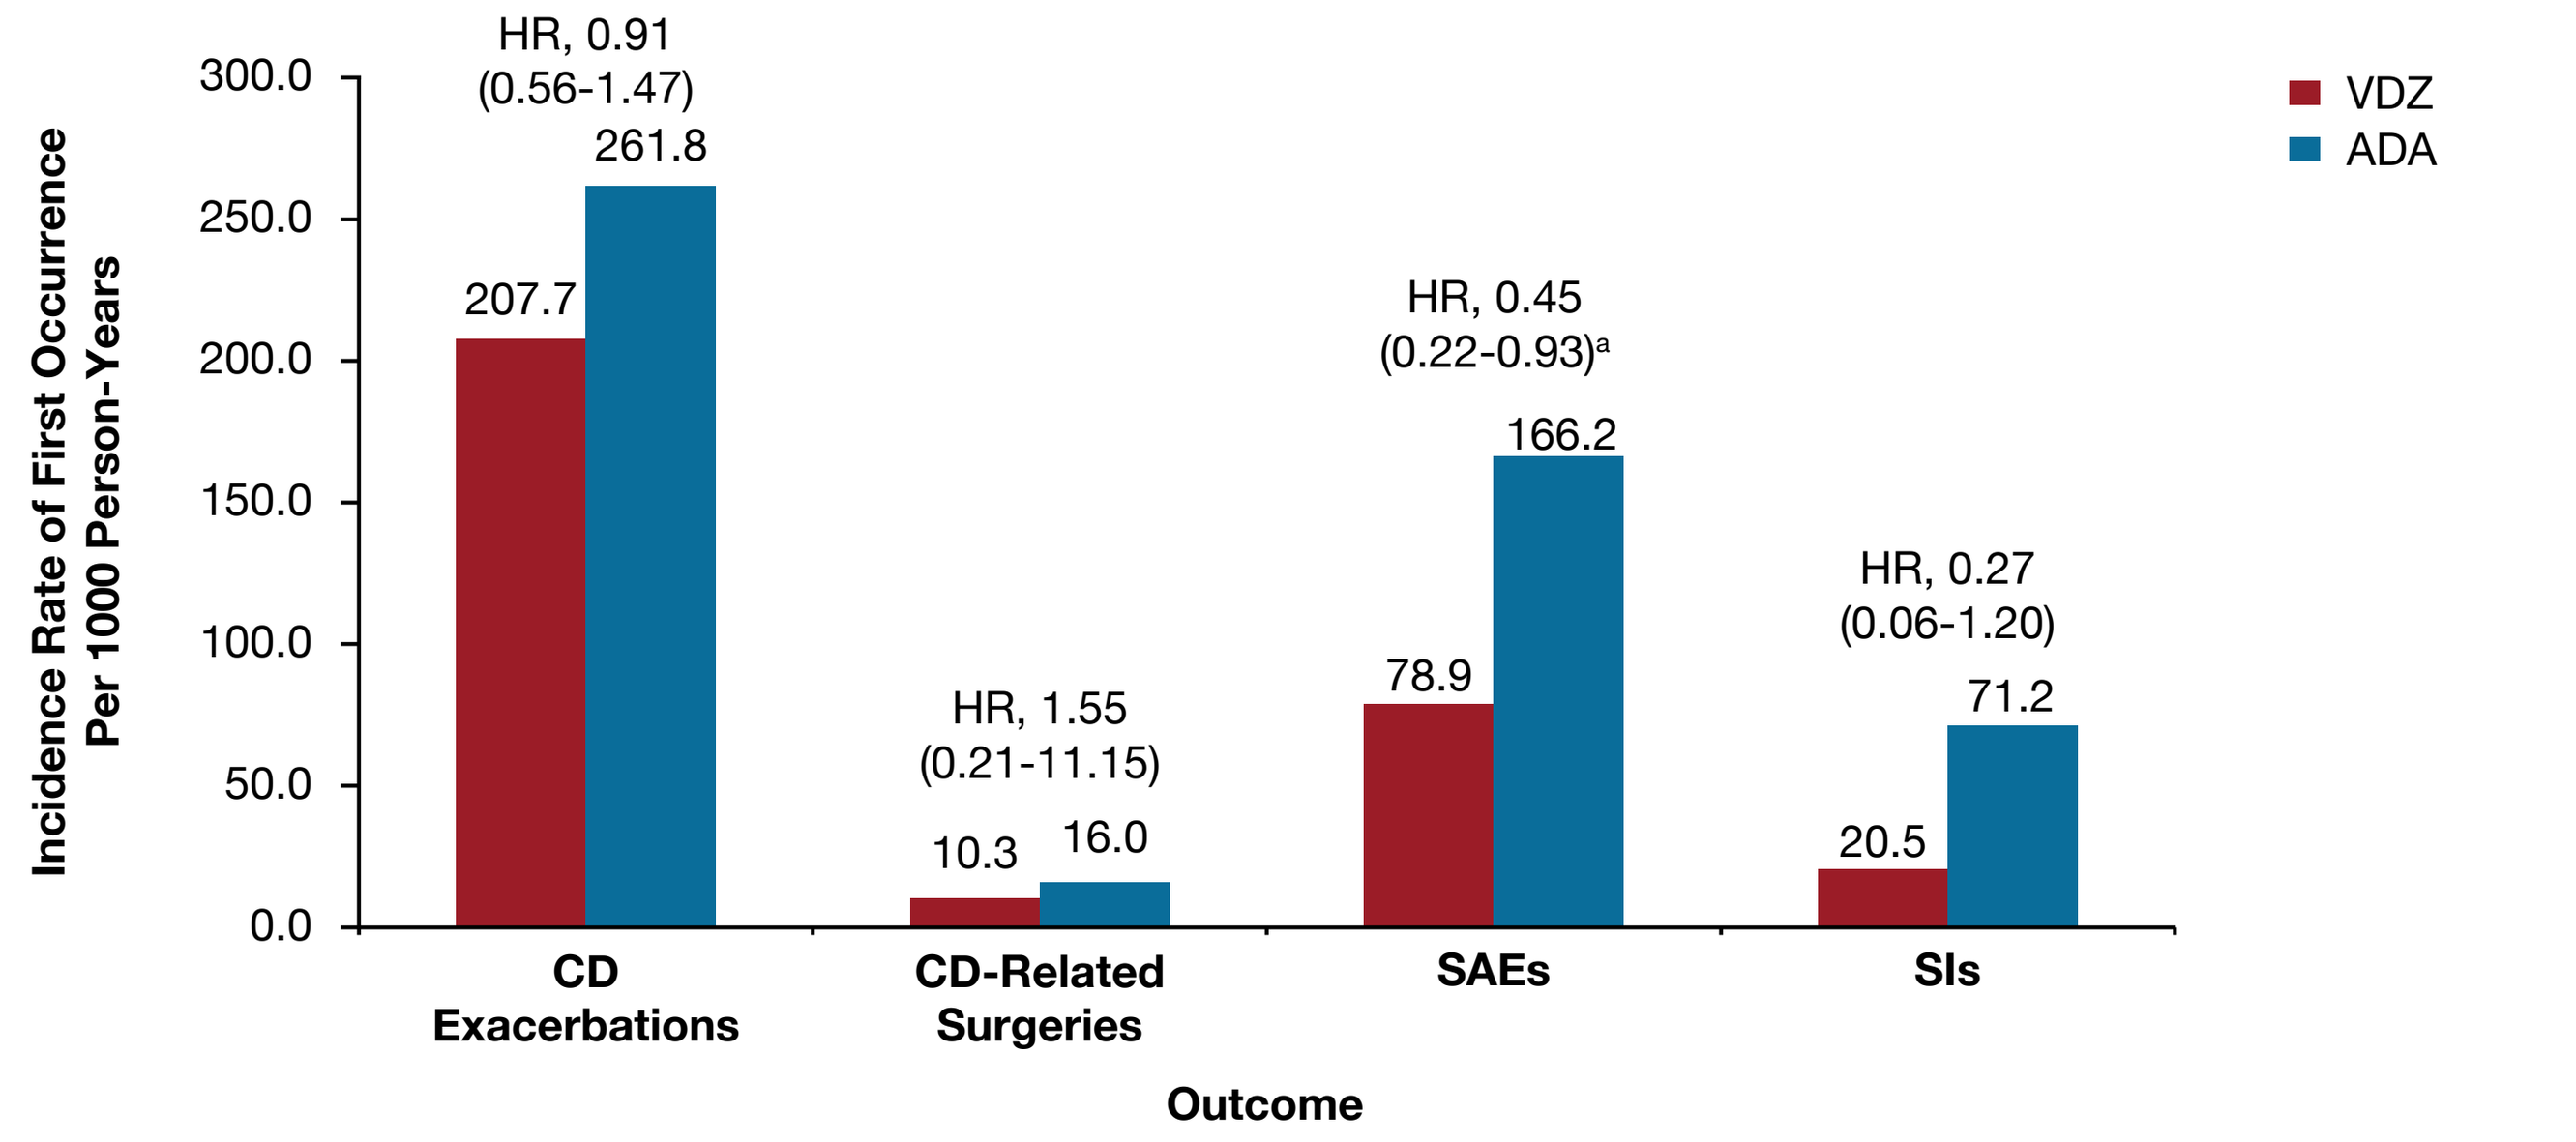
Incidence Rate of First Occurrence Per 1000 Person-Years
VDZ
ADA
HR, 0.91 (0.56-1.47)
207.7 261.8
CD Exacerbations
HR, 1.55 (0.21-11.15)
10.3 16.0
CD-Related Surgeries
HR, 0.45 (0.22-0.93)[a]
78.9 166.2
SAEs
HR, 0.27 (0.06-1.20)
20.5 71.2
SIs
Outcome

Data are unadjusted incidence rates and adjusted HRs (95% CI). p values are from time-to-event analyses.
ADA, adalimumab; CD, Crohn's disease; HR, hazard ratio; SAE, serious adverse event; SI, serious infection; VDZ, vedolizumab.
[a]Denotes statistically significant differences between cohorts.

Conclusions

- In a real-world setting, biologic-naïve patients with CD treated with vedolizumab had equivalent rates of response and mucosal healing but demonstrated a greater likelihood of continuing with treatment and achieving clinical remission versus patients treated with adalimumab
- Patients were less likely to experience SAEs with vedolizumab than with adalimumab

Presented at the American College of Gastroenterology Annual Scientific Meeting • October 21-26, 2022 • Charlotte, NC, USA

References
1. Cushing K, Higgins PDR. *JAMA.* 2021;325(1):69-80.
2. Entyvio [EPAR – product information - summary of product characteristics]. Takeda Pharma A/S; Vallensbaek Strand, Denmark; 2020.
3. Bressler B, et al. *J Crohns Colitis.* 2021;15(10):1694-1706.

Acknowledgments
This study was sponsored by Takeda. Medical writing support was provided by Megan Schladetsch, PhD, and Vardit Dror, PhD, of Excel Medical Affairs, and funded by Takeda.

Disclosures
AY: Advisory board: Arena, Bristol Myers Squibb, Prometheus, Takeda. **BB:** Advisor/speaker: AbbVie, Bristol Myers Squibb, Ferring, Janssen, Merck, Novartis, Pfizer, Takeda; advisor: Alimentiv, Allergan, Amgen, AMT, Bristol Myers Squibb, Celgene, Fresenius Kabi, Genentech, Gilead, Iterative Scopes, Merck, Microbiome Insights, Mylan, Pendopharm, Protagonist; research support: AbbVie, Alvine, Amgen, Boehringer Ingelheim, Bristol Myers Squibb, Celgene, Genentech, GlaxoSmithKline, Janssen, Merck, Qu Biologic; stock options: Qu Biologic. **NRB** and **MB:** Employees of Thermo Fisher Scientific. **SA** and **PK:** Employees of Takeda and hold stock/stock options in Takeda. **GJM:** Research grants: AbbVie, Genesis, Merck Sharp & Dohme, Takeda; advisor/speaker: AbbVie, Aenorasis, Dr Falk, Ferring, Hospira, Janssen, Merck Sharp & Dohme, MYLAN, Pfizer, Takeda, Vianex.

Copies of this poster and supplemental content obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ACG and the authors of this poster.

Scan this QR code to download a copy of the poster and any available supplemental content.