Activity of Gepotidacin Tested Against Molecularly Characterized *Escherichia coli* Isolates Resistant to Commonly Used Oral Therapies for UTI in the US (2019–2020)

RE Mendes[1], SJR Arends[1], JH Kimbrough [1], V Kantro[1], D Butler[2], N Scangarella-Oman[2], J Streit[1], M Castanheira[1]

[1] JMI Laboratories, North Liberty, Iowa, USA; [2] GlaxoSmithKline, Collegeville, PA

Introduction

- Gepotidacin is a novel, bactericidal, first-in-class triazaacenaphthylene antibiotic that inhibits bacterial DNA replication by a distinct mechanism of action, which confers activity against most strains of target pathogens, such as *Escherichia coli*, *Staphylococcus saprophyticus*, and *Neisseria gonorrhoeae*, including those resistant to current antibiotics.
- This study evaluates the epidemiology of community-acquired UTI *E. coli* included in the Gepotidacin Global Surveillance Study as part of the SENTRY Antimicrobial Surveillance Program.
- In addition, the activity of gepotidacin and comparators were investigated against various subsets, including those with characterized β-lactam resistance mechanisms.

Materials and Methods

Bacterial organisms

- A total of 1,993 *E. coli* collected from 45 US sites located in 9 Census Regions were included as part of the Gepotidacin Global Surveillance Program during 2019–2020.

Susceptibility testing

- Isolates were tested for susceptibility by broth microdilution and agar dilution following Clinical and Laboratory Standards Institute (CLSI) M07 (2018) guidelines.
- Frozen-form broth microdilution panels were manufactured by JMI Laboratories (North Liberty, Iowa, USA) and contained cation-adjusted Mueller-Hinton broth. Agar dilution plates were used for testing fosfomycin and included glucose-6-phosphate (25 mg/L).
- *E. coli* with MIC results ≥2 mg/L for aztreonam and/or ceftazidime and/or ceftriaxone were defined as presumptive ESBL producers and subjected to genome sequencing followed by β-lactamase gene screening and epidemiology typing (MLST, O:H, and *fimH*).

Screening of β-lactamase genes

- Selected isolates had total genomic DNA extracted and sequenced on MiSeq Sequencer platforms at JMI Laboratories.
- FASTQ format sequencing files for each sample set were assembled independently using *de novo* assembler SPAdes 3.11.0. An in-house software was applied to align the assembled sequences against a comprehensive in-house database containing known β-lactamase genes.

Epidemiology typing

- Isolates that met the criteria for the screening of ESBL genes were subjected to MLST and O:H typing. Isolates associated with ST131 were also subjected to *fimH* typing.

Results

- A total of 84.4% (1,682/1,993) *E. coli* isolates did not meet the criteria for screening of β-lactamase genes (ESBL negative) (Table 1). Among these isolates, 15.9% (267/1,682) were not susceptible to the fluoroquinolones ciprofloxacin and/or levofloxacin, whereas 26.2% (440/1,682) were not susceptible to trimethoprim-sulfamethoxazole (SXT).
- A presumptive ESBL phenotype was noted in 15.6% (311/1,993) of *E. coli*, with the highest rates observed in the Middle Atlantic (49.7%), East South Central (21.3%), and Pacific (19.6%) regions. Other regions had rates of 6.2–15.9% (Figure 1).
- The *E. coli* isolates with a presumptive ESBL phenotype were mostly not susceptible to the fluoroquinolones (78.8%), SXT (61.7%), amoxicillin-clavulanate (52.9%), and the oral cephalosporins (99.7%) (Table 2).
- Most ESBL isolates carried CTX-M alleles alone (81.4%; 253/311), whereas 9.6% (30/311) had plasmid AmpC genes, including 1 strain from Washington state that carried both $\mathit{bla}_{\mathrm{CMY\text{-}2}}$ and $\mathit{bla}_{\mathrm{DHA\text{-}1}}$.
- Four *E. coli* isolates carried combinations of CTX-M and CMY-encoding genes, and 1 strain from Texas had an NDM-5 gene. A β-lactamase gene could not be detected in 21 (6.8%) of the 311 screened isolates.
- Approximately half (56.3%; 175/311) of the presumptively ESBL isolates belonged to CC131, of which 70.9% (124/175) were O25b:H4 and carried *fimH*30.
- Gepotidacin inhibited 98.8% and 96.1% of non-ESBL and ESBL isolates, respectively, at ≤4 mg/L, as well as all but 1 ESBL isolate belonging to the CC131 lineage (Table 1).
- Several antimicrobial agents tested showed activity against *E. coli* isolates that did not meet the criteria for β-lactamase gene screening; however, ampicillin, amoxicillin-clavulanate, ciprofloxacin, and SXT had suboptimal (i.e. <90% susceptible) susceptibility rates (55.7–86.5% susceptible) (Table 2).
- Gepotidacin ($MIC_{50/90}$, 2/4 mg/L) and fosfomycin ($MIC_{50/90}$, 0.5/2 mg/L; 98.4–98.9% susceptible) showed the lowest MIC results against the presumptive ESBL and CTX-M producers as well as the CC131 and non-CC131 subsets. Nitrofurantoin (90.3–95.6% susceptible) was also active *in vitro* against these subsets, despite MIC results ($MIC_{50/90}$, 16/32 mg/L) at least 8-fold higher than gepotidacin and fosfomycin (Table 2).

- **High rates of *E. coli* that were not susceptible to commonly used oral agents (e.g., FQs and SXT) were observed, plus a presumptive ESBL phenotype further compromised the activity of oral agents, including oral cephalosporins.**
- **Gepotidacin had potent and stable *in vitro* activity against various subsets, including the resistant CC131 O25b:H4 clone.**
- **These data support the further clinical development of gepotidacin as a treatment option for uUTI caused by *E. coli*, including resistant isolates against which other oral treatment options are limited.**

Table 1. Distribution of gepotidacin MICs against phenotypic and genotypic subsets of *E. coli*

E. coli phenotype/genotype (No. isolates)	Number (cumulative %) of isolates inhibited by gepotidacin at MIC (mg/L) of:										MIC (mg/L)	
	≤0.06	0.12	0.25	0.5	1	2	4	8	16	32	50%	90%
Non-ESBL (1,682)	1 (0.1)	4 (0.3)	14 (1.1)	76 (5.6)	548 (38.2)	910 (92.3)	109 (98.8)	19 (99.9)	1 (100)		2	2
FQ-NS (267)		2 (0.7)	12 (5.2)	35 (18.4)	88 (51.3)	96 (87.3)	24 (96.3)	9 (99.6)	1 (100)		1	4
SXT-NS (440)		2 (0.5)	5 (1.6)	35 (9.5)	162 (46.4)	195 (90.7)	29 (97.3)	11 (99.8)	1 (100)		2	2
ESBL (311)	2 (0.6)	2 (1.3)	3 (2.3)	28 (11.3)	103 (44.4)	124 (84.2)	37 (96.1)	7 (98.4)	4 (99.7)	1 (100)	2	4
FQ-NS (245)	2 (0.8)	2 (1.6)	3 (2.9)	20 (11.0)	85 (45.7)	94 (84.1)	29 (95.9)	5 (98.0)	4 (99.6)	1 (100)	2	4
SXT-NS (192)		3 (1.6)	3 (3.1)	17 (12.0)	65 (45.8)	74 (84.4)	20 (94.8)	7 (98.4)	2 (99.5)	1 (100)	2	4
CTX-M[a] (253)		2 (1.6)	3 (2.8)	26 (13.0)	91 (49.0)	96 (87.0)	26 (97.2)	5 (99.2)	1 (99.6)	1 (100.0)	2	4
CMY/DHA[b] (30)					6 (20.0)	15 (70.0)	7 (93.3)	1 (96.7)	1 (100.0)		2	4
CC131[c] (175)		1 (0.6)	0 (0.6)	16 (9.7)	62 (45.1)	76 (88.6)	19 (99.4)	1 (100.0)			2	4
O25b:H4/*fimH*30 (124)				8 (6.5)	42 (40.3)	58 (87.1)	15 (99.2)	1 (100.0)			2	4
O16:H5/*fimH*41 (26)				8 (30.8)	10 (69.2)	7 (96.2)	1 (100.0)				2	2
Non-CC131 (136)	2 (1.5)	1 (2.2)	3 (4.4)	12 (13.2)	41 (43.4)	48 (78.7)	18 (91.9)	6 (96.3)	4 (99.3)	1 (100.0)	2	4

ESBL, extended-spectrum -β-lactamase; FQ, fluoroquinolones (not susceptible to ciprofloxacin and/or levofloxacin); SXT, trimethoprim-sulfamethoxazole; NS, not susceptible; CC, clonal complex.
[a] Includes 149 $\mathit{bla}_{\mathrm{CTX\text{-}M\text{-}15}}$, 69 $\mathit{bla}_{\mathrm{CTX\text{-}M\text{-}27}}$, 24 $\mathit{bla}_{\mathrm{CTX\text{-}M\text{-}14}}$, and 11 isolates with other $\mathit{bla}_{\mathrm{CTX\text{-}M}}$ alleles.
[b] Includes 22 $\mathit{bla}_{\mathrm{CMY}}$, 7 $\mathit{bla}_{\mathrm{DHA}}$, and 1 isolate with both $\mathit{bla}_{\mathrm{CMY}}$ and $\mathit{bla}_{\mathrm{DHA}}$ alleles.
[c] Includes clonal complex 131, which is represented by ST131 (164 isolates), ST131-like (5), ST2279 (3), and ST8671 (3).

Figure 1. Proportions of presumptive ESBL-producing *E. coli* causing UTI in the 9 US Census Regions. The proportion of these isolates belonging to the CC131 clone are also shown.

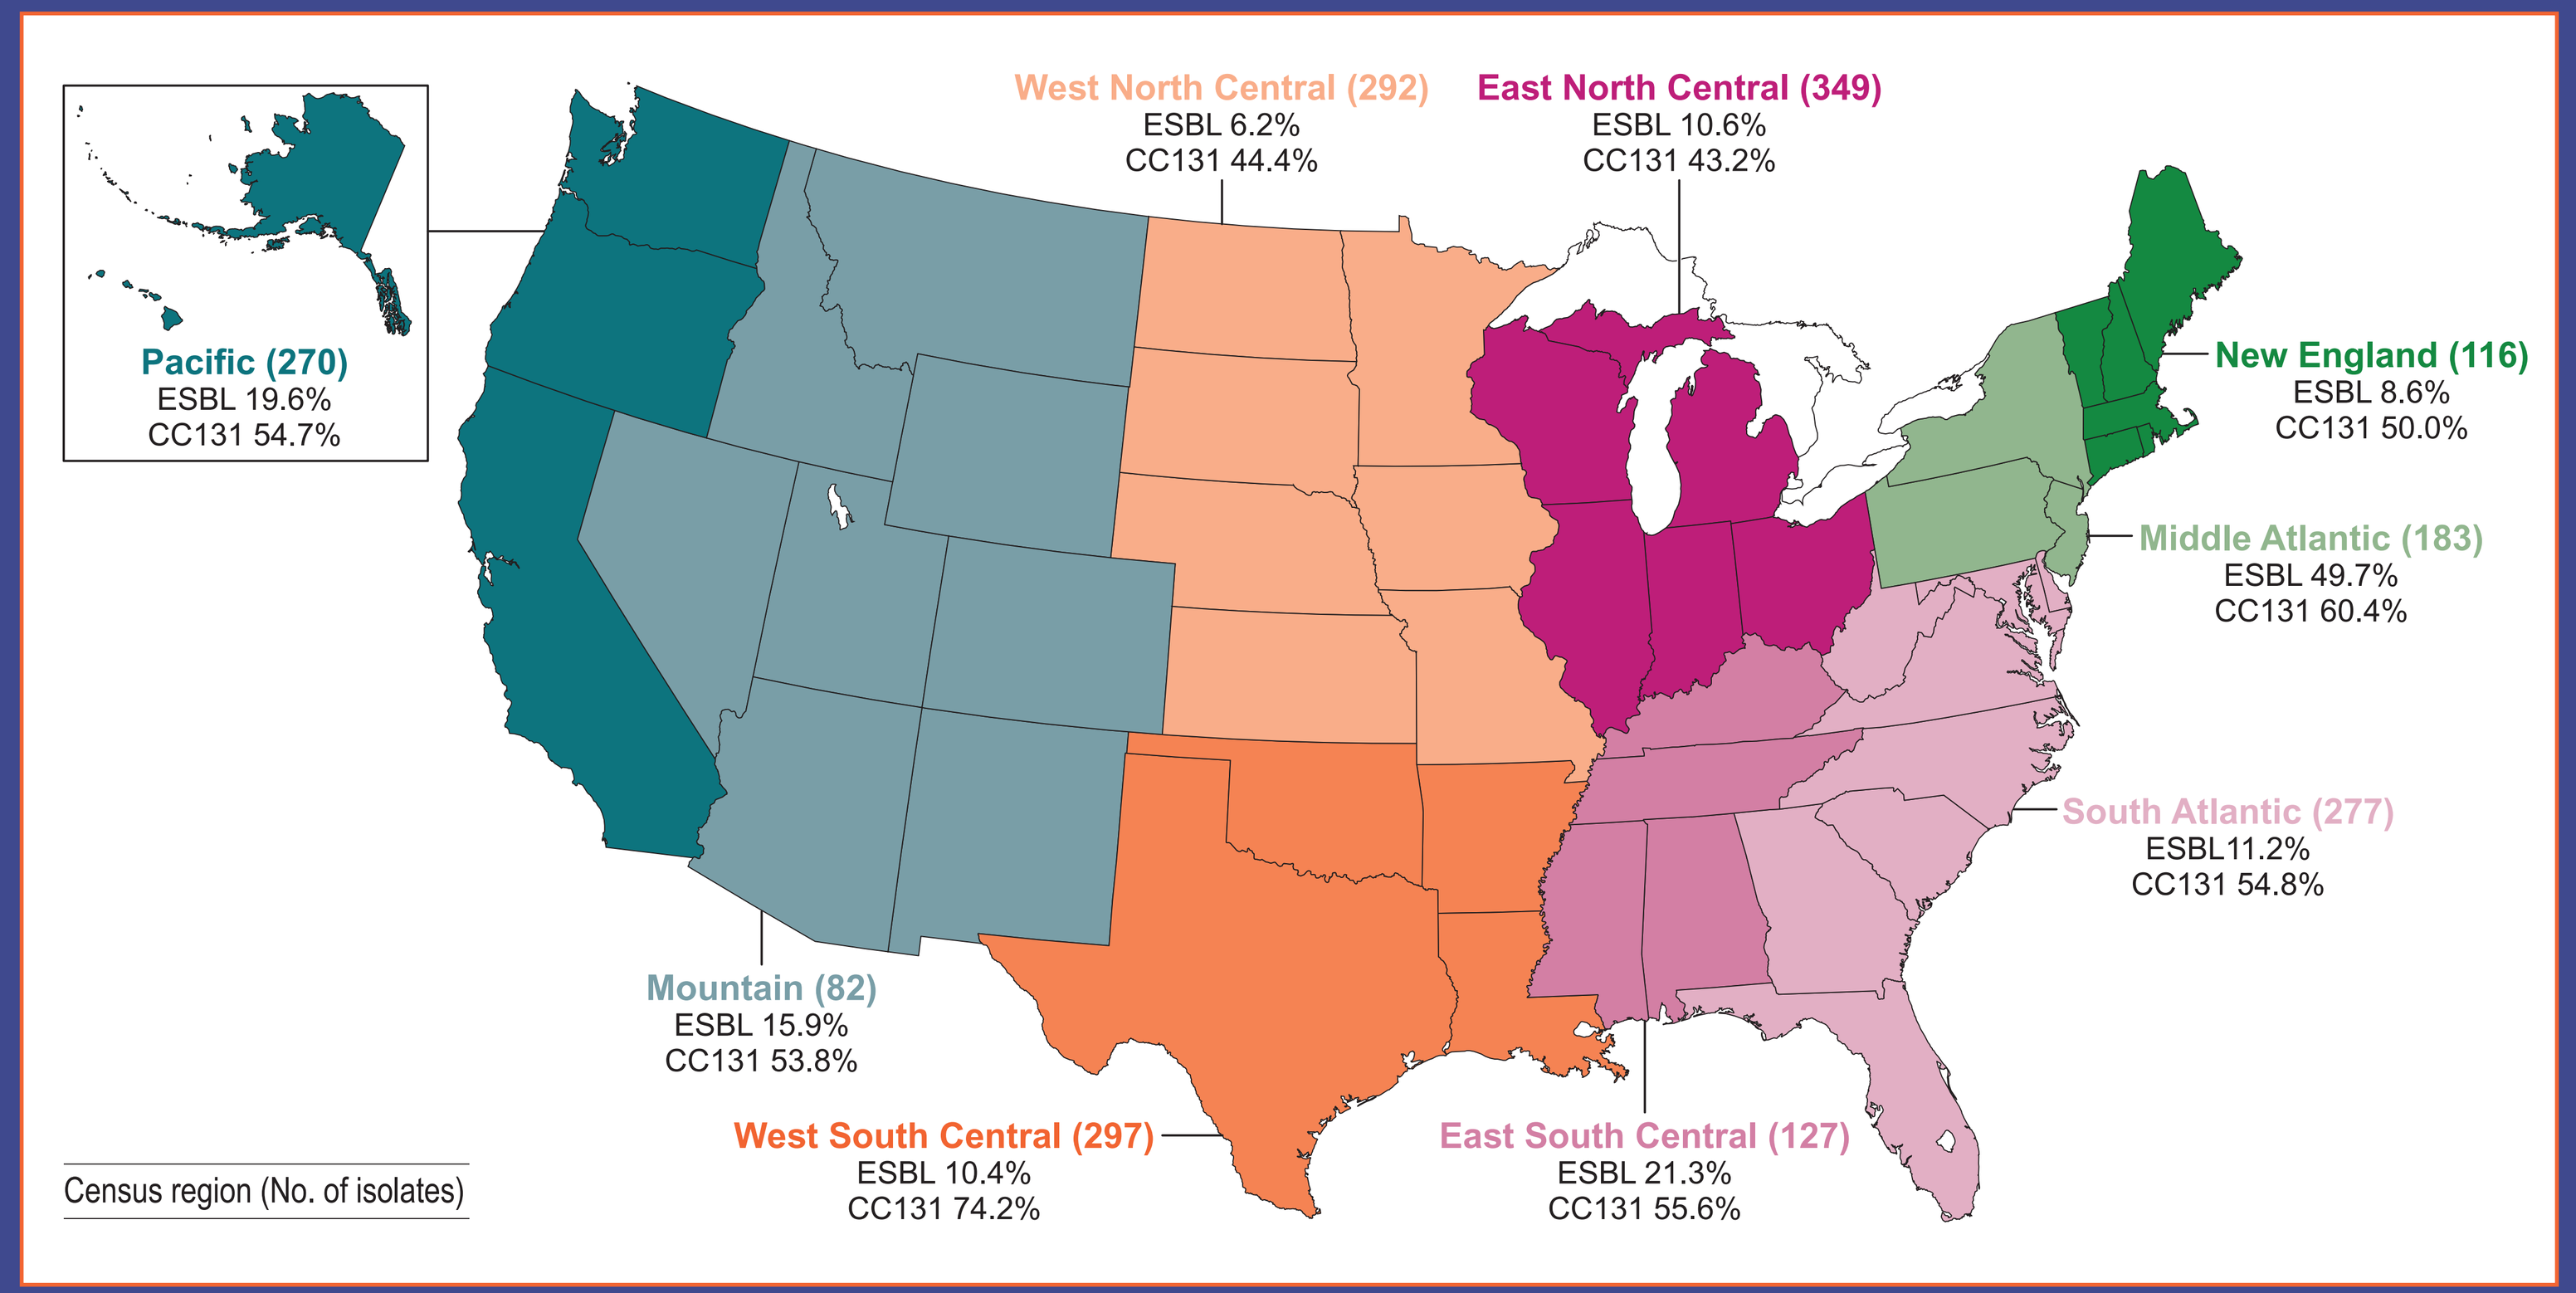
West North Central (292)
ESBL 6.2%
CC131 44.4%
East North Central (349)
ESBL 10.6%
CC131 43.2%
Pacific (270)
ESBL 19.6%
CC131 54.7%
New England (116)
ESBL 8.6%
CC131 50.0%
Middle Atlantic (183)
ESBL 49.7%
CC131 60.4%
South Atlantic (277)
ESBL11.2%
CC131 54.8%
Mountain (82)
ESBL 15.9%
CC131 53.8%
West South Central (297)
ESBL 10.4%
CC131 74.2%
East South Central (127)
ESBL 21.3%
CC131 55.6%
Census region (No. of isolates)

SCAN ME

Poster #P1675
IDWeek 2022, October 19–23, 2022,
Washington, D.C.

Table 2. Activity of gepotidacin and comparator agents against various subsets of *E. coli*

Antimicrobial agent	MIC (mg/L)			CLSI[a]		
	50%	90%	Range	%S	%I	%R
Non-ESBL (1,682)						
Gepotidacin	2	2	0.06 to 16			
Ampicillin	8	>64	≤1 to >64	55.7	0.8	43.5
A/C	4	16	0.5 to >32	86.5	11.1	2.4
Cefazolin	2	8	≤0.5 to >32	96.5 [b]		3.5
Ciprofloxacin	0.015	>4	≤0.002 to >4	84.5	1.7	13.9
Nitrofurantoin	16	32	≤2 to >128	98.3	0.9	0.8
SXT	≤0.12	>4	≤0.12 to >4	73.8		26.2
Fosfomycin	0.5	1	0.25 to >256	99.8	0.2	0.1
Aztreonam	≤0.06	0.12	≤0.06 to 1	100.0	0.0	0.0
Ceftriaxone	0.12	0.25	≤0.03 to 1	100.0	0.0	0.0
Ceftazidime	0.12	0.5	0.03 to 1	100.0	0.0	0.0
ESBL (311)						
Gepotidacin	2	4	≤0.03 to 32			
Ampicillin	>64	>64	8 to >64	0.3	0.0	99.7
A/C	16	32	2 to >32	47.1	31.0	21.9
Cefazolin	>32	>32	16 to >32	0.3 [b]		99.7
Ciprofloxacin	>4	>4	0.004 to >4	21.0	3.9	75.2
Nitrofurantoin	16	32	≤2 to >128	92.6	3.5	3.9
SXT	>4	>4	≤0.12 to >4	38.3		61.7
Fosfomycin	0.5	2	0.25 to >256	98.7	0.0	1.3
Aztreonam	>16	>16	0.12 to >16	18.3	17.0	64.6
Ceftriaxone	>8	>8	0.12 to >8	6.8	1.3	92.0
Ceftazidime	16	>32	0.25 to >32	28.7	15.2	56.1
CTX-M (253)[c]						
Gepotidacin	2	4	≤0.03 to 32			
Ampicillin	>64	>64	>64	0.0	0.0	100.0
A/C	8	16	2 to >32	56.3	37.3	6.3
Cefazolin	>32	>32	32 to >32	0.0 [b]		100.0
Ciprofloxacin	>4	>4	0.004 to >4	12.7	3.2	84.1
Nitrofurantoin	16	32	≤2 to >128	92.5	3.6	4.0
SXT	>4	>4	≤0.12 to >4	34.8		65.2
Fosfomycin	0.5	2	0.25 to >256	98.4	0.0	1.6
Aztreonam	>16	>16	0.5 to >16	10.3	16.2	73.5
Ceftriaxone	>8	>8	>8	0.0	0.0	100.0
Ceftazidime	16	>32	0.25 to >32	27.7	16.6	55.7
CC131 (175)[c]						
Gepotidacin	2	4	0.12 to 8			
Ampicillin	>64	>64	>64	0.0	0.0	100.0
A/C	16	16	4 to >32	48.3	42.5	9.2
Cefazolin	>32	>32	32 to >32	0.0 [b]		100.0
Ciprofloxacin	>4	>4	0.015 to >4	4.6	2.9	92.5
Nitrofurantoin	16	32	≤2 to 128	90.3	4.0	5.7
SXT	>4	>4	≤0.12 to >4	36.0		64.0
Fosfomycin	0.5	2	0.25 to >256	98.9	0.0	1.1
Aztreonam	>16	>16	0.25 to >16	8.0	18.3	73.7
Ceftriaxone	>8	>8	0.12 to >8	0.6	1.1	98.3
Ceftazidime	16	>32	0.5 to >32	21.7	16.6	61.7

ESBL, extended spectrum-β-lactamase; A/C, amoxicillin-clavulanate (2:1); SXT, trimethoprim-sulfamethoxazole
[a] Criteria as published by CLSI (2022).
[b] Using breakpoints as a surrogate test to predict susceptibility results to oral cephalosporins for treating uncomplicated UTI.
[c] See footnotes on Table 1 for additional information.

Acknowledgements

This study at JMI Laboratories was supported by GlaxoSmithKline. JMI Laboratories received compensation fees for services in relation to preparing the poster. This project has been funded in whole or in part with Federal funds from the Office of the Assistant Secretary for Preparedness and Response, Biomedical Advanced Research and Development Authority, under OTA Agreement No. HHSO100201300011C.

References

1. Clinical and Laboratory Standards Institute (2018). Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically. *M07 11th Edition*. Wayne, PA, USA.
2. Clinical and Laboratory Standards Institute (2022). Performance standards for antimicrobial susceptibility testing. *M100 32nd Edition*. Wayne, PA, USA.
3. Mendes RE, Jones RN, Woosley LN, Cattoir V, Castanheira M (2019). Application of next-generation sequencing for characterization of surveillance and clinical trial isolates: Analysis of the distribution of β-lactamase resistance genes and lineage background in the United States. *Open Forum Infect Dis* 6: S69-S78.
4. Taylor SN, Morris DH, Avery AK, Workowski KA, Batteiger BE, Tiffany CA, Perry CR, Raychaudhuri A, Scangarella-Oman NE, Hossain M, Dumont EF (2018). Gepotidacin for the treatment of uncomplicated urogenital gonorrhea: A phase 2, randomized, dose-ranging, single-oral dose evaluation, *Clin Infect Dis*, 67: 504–512.

Contact

Rodrigo E. Mendes, Ph.D.
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, Iowa 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: rodrigo-mendes@jmilabs.com