Trends in Hospitalizations and Outpatient Visits with Early or Delayed Antibiotic Treatment Initiation in Nontuberculous Mycobacterial Lung Disease

Kevin Winthrop[1], Catherine Waweru[2], Mariam Hassan[2], Sara Burns[3], Anjan Chatterjee[2]

[1]OHSU-PSU School of Public Health, Portland, OR, USA; [2]Insmed Incorporated, Bridgewater Township, NJ, USA; [3]Panalgo, Boston, MA, USA

Kevin Winthrop; winthrop@ohsu.edu
Catherine Waweru; Catherine.Waweru@insmed.com
Mariam Hassan; Mariam.Hassan@Insmed.com
Sara Burns; saramariaburns@gmail.com
Anjan Chatterjee; Anjan.Chatterjee@Insmed.com

BACKGROUND

- Nontuberculous mycobacterial lung disease (NTMLD) is a rare, progressive lung infection and a growing health concern.[1,2]
- NTMLD is characterized by nonspecific symptoms (such as chronic cough and dyspnea) and common respiratory comorbidities (such as chronic obstructive pulmonary disease and bronchiectasis).[3]
 - Treatment initiation with antibiotics is recommended in patients who meet the diagnostic criteria of NTMLD; the 2020 treatment guidelines for patients diagnosed with NTMLD recommend antibiotic treatment initiation rather than "watchful waiting" for most patients.[4]
- There are limited data on the consequences of delayed treatment of NTMLD and its impact on healthcare resource utilization (HCRU). The objective of this study was to assess HCRU in patients with NTMLD receiving early or delayed antibiotic treatment.

METHODS

Data Source

- This was a retrospective, observational, cohort study based on claims data from the MarketScan® Commercial Claims and Encounters database between 2014 and 2020.

Study Population

- Patient inclusion criteria were as follows:
 - ≥2 NTMLD medical claims (using the *International Classification of Diseases, Ninth Revision, Clinical Modification [ICD-9-CM]* code 031.0 or *Tenth Revision [ICD-10-CM]* code A31.0); ≥30 days apart and within 12 months of each other. The date of the first NTMLD claim was defined as the index date.
 - Continuous enrollment spanning 12 months pre-index date (referred to hereafter as baseline) to ≥24 months post-index date.
 - No diagnosis of tuberculosis anytime from baseline through follow-up.
 - Initiated antibiotic treatment for NTMLD after index date.

Treatment Definitions

- NTMLD treatment was defined as receiving initial treatment with antibiotics from ≥2 drug classes listed in **Table 1**, each for ≥28 days and within 30 days of each other.
- Patients receiving macrolide-based treatment were defined as patients receiving initial treatment with macrolide (azithromycin and/or clarithromycin) plus ≥1 of the following overlapping agents (for ≥28 days and within 30 days): rifamycin (rifampin or rifabutin), ethambutol, fluoroquinolone (ciprofloxacin, levofloxacin, or moxifloxacin), or intravenous/inhaled amikacin.
- The mean time to treatment initiation from index date was used to define early- and delayed-treatment groups.

Study Outcomes

- Hospitalizations (all cause and respiratory related) and outpatient visits (emergency room [ER] or non-ER visits) at baseline, year 1 and year 2 after index date.
 - Hospitalizations with ICD-9/10-CM codes for pulmonary comorbidities (ICD-9-CM 460-519 or ICD-10-CM J00-J99) in any position were considered as respiratory related.

Analysis

- Patient characteristics, including age at index date, sex, and baseline comorbidities were compared between treatment groups using X^2 tests for categorical variables and Student's t-test for continuous variables.
- For each treatment group, all-cause respiratory disease–related hospitalizations, and outpatient visits were compared at baseline and during the first 2 years post-index date.
 - To account for the nonindependence of this pre-post analysis, McNemar X^2 tests were used to compare the proportion of patients with hospitalizations at year 1 and year 2 post-index versus baseline. Similarly, Wilcoxon signed rank tests were used to compare the rates of hospitalization and outpatient visits in year 1 and year 2 post-index versus baseline. $P<0.05$ was considered statistically significant.

Table 1. Antibiotic Drug Classes Used to Define Treatment for NTMLD

Drug Class	Drug Name
Macrolide	azithromycin, clarithromycin
Ethambutol	ethambutol
Rifamycin	rifampin, rifabutin
Aminoglycoside	amikacin, streptomycin
Fluoroquinolone	ciprofloxacin, moxifloxacin, levofloxacin
Carbapenem	imipenem, meropenem
Oxazolidinone	linezolid, tedizolid
Glycylcycline	tigecycline
Cephalosporin	cefoxitin
Tetracycline	omadacycline
Diarylquinoline	bedaquiline
Lipophilic riminophenazine	clofazimine

NTMLD, nontuberculous mycobacterial lung disease

RESULTS

Study Population

- A total of 481 patients met the definition of NTMLD-treated. Of these, 445 patients (93%) received macrolide-based treatment. A summary of the initial antibiotic regimens received by the NTMLD-treated cohort is provided in **Table 2.**
- Mean (standard deviation [SD]) time from index date to antibiotic treatment initiation was 3.28 (7.05) months. Early-treatment patients were, therefore, defined as patients whose mean time to treatment initiation from index date was ≤3 months; while delayed-treatment patients initiated treatment >3 months post-index date.
- A total of 364 (76%) and 117 (24%) patients comprised the early- and delayed-treatment groups, respectively (**Figure 1**).
- The mean (SD) time to treatment in the early-and delayed-treatment groups was 0.62 (0.73) and 11.55 (10.63) months, respectively.

Table 2. Initial Antibiotic Regimen Received in the NTMLD-Treatment Cohort

Antibiotic regimen, n (%)	Patients Receiving Treatment (N=481)
Macrolide + ethambutol + rifamycin	269 (55.9)
Macrolide + ethambutol	56 (11.6)
Macrolide + rifamycin	41 (8.5)
Macrolide + fluoroquinolone	32 (6.7)
Macrolide + rifamycin + fluoroquinolone +/- ethambutol	10 (2.1)
Macrolide + ethambutol + fluoroquinolone	6 (1.2)
Macrolide + other[a]	31 (6.4)
Non-macrolide-containing regimen[b]	36 (7.5)

[a]Other: oxazolidinone (n=16); amikacin (n=12); diarylquinoline (n=1); oxazolidinone + amikacin (n=1); diarylquinoline + amikacin (n=1). [b]Non-macrolide-containing regimen: ethambutol + rifamycin (n=24); fluoroquinolone + oxazolidinone (n=5); fluoroquinolone + rifamycin (n=2); ethambutol + fluoroquinolone (n=2); ethambutol + fluoroquinolone + rifamycin (n=2); amikacin + oxazolidinone (n=1). NTMLD, nontuberculous mycobacterial lung disease.

Figure 1. Patient Disposition

All patients from Oct 2014 to June 2020
N=19,345
↓
NTMLD medical claims between July 2015 and June 2018
n=11,811
↓
Second NTMLD medical claim within ≤12 months of the first
n=6,161
↓
No NTMLD diagnosis 12 months prior to first NTMLD claim (index date)
n=4,233
↓
Continuous enrollment 12 months pre-index date
n=2,941
↓
Age at index date >18 years
n=2,831
↓
No tuberculosis diagnosis pre- and post-index date
n=2,340
↓
2 years continuous enrollment post-index
n= 1,126
↓
NTMLD-treated patients
n=481

Early treatment for NTMLD n=364 | Delayed treatment for NTMLD n=117

NTMLD, nontuberculous mycobacterial lung disease.

Baseline Demographics and Clinical Characteristics

- Compared with the delayed-treatment group, the early-treatment group was significantly younger (mean, 58.9 vs 62.0 years) and had a significantly lower proportion of patients who were female (57.7% vs 78.6%) (**Table 3**).
- The delayed-treatment group had a higher proportion of patients with bronchiectasis (41.0% vs 32.4%), asthma (32.5% vs 25.0%), and emphysema (16.2% vs 12.4%) (**Table 3**).

Hospitalization Burden in Patients with Early or Delayed Treatment for NTMLD

All-cause and respiratory-related hospitalizations

- In both treatment groups, ~30% of treated patients experienced a hospitalization at baseline (all cause; **Figure 2A**), with over a fifth of patients experiencing a respiratory-related hospitalization (**Figure 2B**).

Table 3. Characteristics of NTMLD-Treated Patients by Treatment Group

Characteristic	Early Treatment[a] (n=364)	Delayed Treatment[b] (n=117)	*P* value[f]
Mean age at index date[c], years (SD)	58.9 (12.8)	62.0 (13.6)	0.03
Age ≥65 years, n (%)	97 (26.7)	46 (39.3)	0.01
Female, n (%)	210 (57.7)	92 (78.6)	<0.0001
Mean baseline CCI, score (SD)[d]	1.85 (1.87)	1.97 (1.93)	0.56
Baseline immunosuppressant use, n (%)[d,e]	187 (51.4)	63 (53.9)	0.72
Baseline pulmonary comorbidities, n (%)[d]			
Cough	219 (60.2)	74 (63.3)	0.63
Pneumonia	143 (39.3)	45 (38.5)	0.96
COPD	134 (36.8)	44 (37.6)	0.96
Bronchiectasis	118 (32.4)	48 (41.0)	0.11
Asthma	91 (25.0)	38 (32.5)	0.14
Emphysema	45 (12.4)	19 (16.2)	0.36
Hemoptysis	32 (8.8)	14 (12.0)	0.40
Baseline non-pulmonary comorbidities, n (%)[d]			
Hypertension	152 (41.8)	51 (43.6)	0.81
Cancer	66 (18.1)	22 (18.8)	0.97
Cardiovascular	62 (17.0)	21 (18.0)	0.93
Diabetes	44 (12.1)	19 (16.2)	0.32

[a]Time to treatment initiation from index date was ≤3 months. [b]Time to treatment initiation from index date was >3 months. [c]Defined as the date of the first NTMLD diagnosis. [d]12 months prior to index date. [e]Immunosuppressant use included the following: methotrexate, leflunomide, sulfasalazine, tacrolimus, cyclosporine, cyclophosphamide, azathioprine, infliximab, etanercept, golimumab, certolizumab, adalimumab, tofacitinib, baricitinib, upadicitinib, rituximab, abatacept, ixekizumab, secukinumab, corticosteroids (oral and inhaled), anakinra, brodalumab, ustekinumab. [f]X^2 test used to compare categorical variables; Student's t-test used to compare continuous variables. CCI, Charlson Comorbidity Index; COPD, chronic obstructive pulmonary disease; NTMLD, nontuberculous mycobacterial lung disease; SD, standard deviation.

- In the early-treatment group, the proportion of patients with all-cause hospitalizations decreased in years 1 and 2 post-index date, while in the delayed-treatment group, there was an increase in the proportion of patients with all-cause hospitalizations in year 1 followed by a decrease in year 2.
 - The early-treatment group showed a statistically significant reduction in the proportion of hospitalized patients from baseline at year 2 (33.2% vs 21.7%; $P<0.05$), whereas no statistically significant differences from baseline in the proportions of hospitalized patients were observed in the delayed-treatment group (**Figure 2A**).
- The mean number of all-cause and respiratory-related hospitalizations per patient at baseline were higher in the early- than delayed-treatment group (**Figure 2C** and **2D**).
- The mean number of all-cause hospitalizations per patient in the early-treatment group decreased in years 1 and year 2 post-index date; conversely, in the delayed-treatment group, there was an increase in the mean number of hospitalizations in year 1 followed by a decrease in year 2.
 - The early-treatment group showed a statistically significant reduction in the mean (SD) number of all-cause hospitalizations at year 2 from baseline (0.50 [0.94] vs 0.35 [0.80]; $P<0.05$), while the delayed-treatment group showed a statistically significant increase in the mean (SD) number of all-cause hospitalizations at year 1 from baseline (0.40 [0.67] vs 0.61 [1.11]; $P<0.05$) (**Figure 2C**).
- Similar trends were observed for the proportion of patients with respiratory-related hospitalizations (**Figure 2B**) and mean number of respiratory-related hospitalizations per patient (**Figure 2D**).

Outpatient visits

ER visits

- Approximately one-third of early- and delayed-treatment patients experienced an ER visit (**Figure 3A**) at baseline.
- In the early-treatment group, the proportion of patients with ER visits decreased at year 1 and year 2 post-index date compared with baseline, reaching statistical significance at year 2 (31.0% vs 24.7%; P<0.05); a decrease at year 1 and year 2 post-index versus baseline also was observed in the delayed-treatment group, but this was not statistically significant. (**Figure 3A**).
- At baseline, mean number of ER visits per patient were similar between the early- and delayed-treatment group. Mean (SD) number of ER visits per patient decreased significantly in the early-treatment group from baseline to year 1 (0.51 [1.12] vs 0.40 [0.89]; $P<0.05$); decreases also were observed at year 2 versus baseline but did not reach statistical significance (**Figure 3B**).
- No significant differences in the mean number of ER visits per patient from baseline to years 1 and 2 post-index were observed in the delayed-treatment group.

Non-ER visits

- A majority of patients overall (>99.2%) had a non-ER outpatient visit at baseline and years 1 and 2 post-index date. For both treatment groups, no differences were observed in the proportion of patients with non-ER visits at years 1 and 2 post-index date from baseline.
- At baseline, mean number of non-ER visits per patient were similar between the early- and delayed-treatment group. For both the treatment groups, the mean (SD) number of non-ER outpatient visits per patient was statistically significantly higher at year 1 compared with baseline (early treatment group: 24.27 [17.06] vs 29.53 [20.50]; delayed-treatment group: 25.13 [18.67] vs 32.48 [21.40]). In the delayed-treatment group, the mean number of non-ER visits per patient also was numerically higher at year 2 versus baseline but did not reach statistical significance (**Figure 3C**).

Figure 2. Proportion of Patients With All-Cause (panel A) and Respiratory-Related[a] (panel B) Hospitalizations; Mean Number of All-Cause (panel C) and Respiratory-Related (panel D) Hospitalizations per Patient

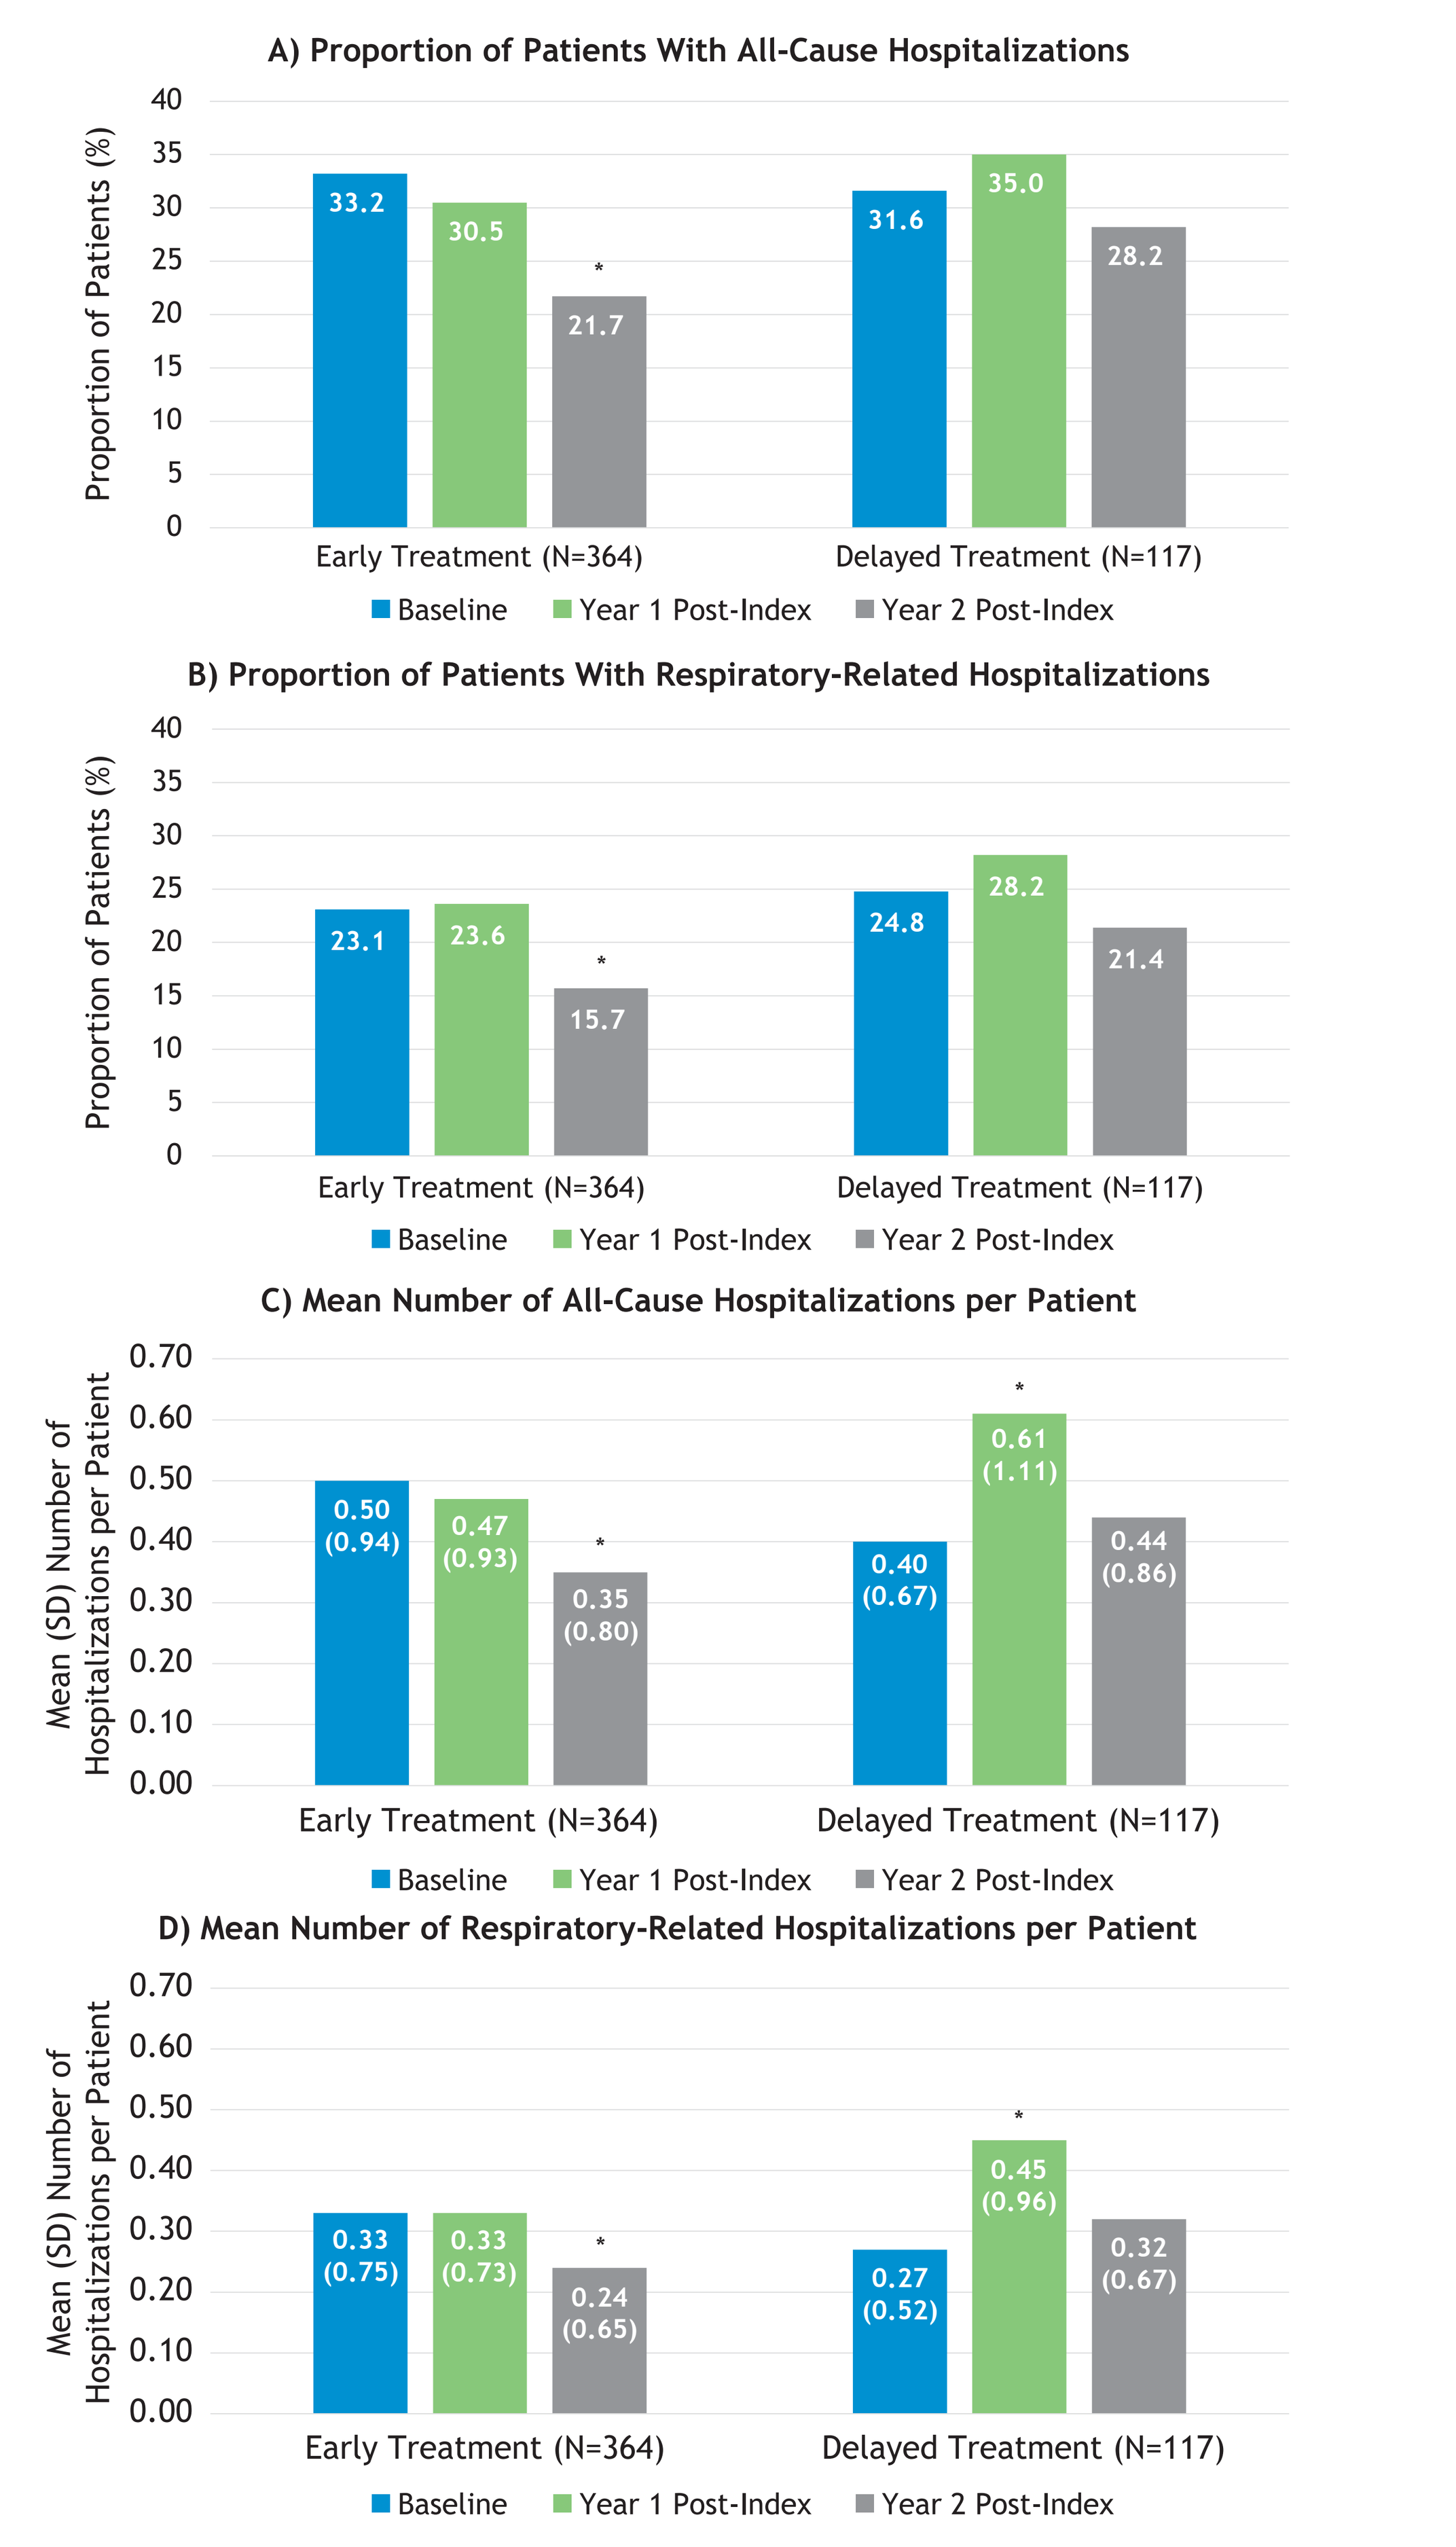

A) Proportion of Patients With All-Cause Hospitalizations
Proportion of Patients (%)
33.2
30.5
21.7
31.6
35.0
28.2
B) Proportion of Patients With Respiratory-Related Hospitalizations
23.1
23.6
15.7
24.8
28.2
21.4
C) Mean Number of All-Cause Hospitalizations per Patient
Mean (SD) Number of Hospitalizations per Patient
0.50 (0.94)
0.47 (0.93)
0.35 (0.80)
0.40 (0.67)
0.61 (1.11)
0.44 (0.86)
D) Mean Number of Respiratory-Related Hospitalizations per Patient
0.33 (0.75)
0.33 (0.73)
0.24 (0.65)
0.27 (0.52)
0.45 (0.96)
0.32 (0.67)
Early Treatment (N=364)
Delayed Treatment (N=117)
Baseline
Year 1 Post-Index
Year 2 Post-Index

*$P<0.05$. McNemar X^2 test used to compare proportions of hospitalizations at year 1 and year 2 versus baseline (panels A and B); Wilcoxon signed rank test used to compare mean number of hospitalizations at year 1 and year 2 versus baseline (panels C and D). [a]Hospitalizations with any claim for a respiratory event. Index date: date of first NTMLD diagnosis. NTMLD, nontuberculous mycobacterial lung disease; SD, standard deviation.

FUNDING:
This work was supported by funding from Insmed Inc.

ACKNOWLEDGMENTS:
Medical writing support was provided by Tracey McManus, PhD, of Curo Consulting, a division of Envision Pharma Group, and funded by Insmed Inc.

REFERENCES:

1. Adjemian J, et al. *Am J Respir Crit Care Med*. 2012;185:881-6. 2. Marras TK, et al, *J Manag Care Spec Pharm*. 2018;24:964-74. 3. Stout JE, et al, *Int J Infect Dis*. 2016;45:123-34. 4. Daley CL, et al. *Clin Infect Dis*. 2020;71:e1-e36.doi:10.1093/cid/ciaa241.

Figure 3. Proportion of Patients With ER Visits (panel A); Mean number of ER Visits per Patient (panel B); and Mean Number of Non-ER Visits per Patient (panel C)

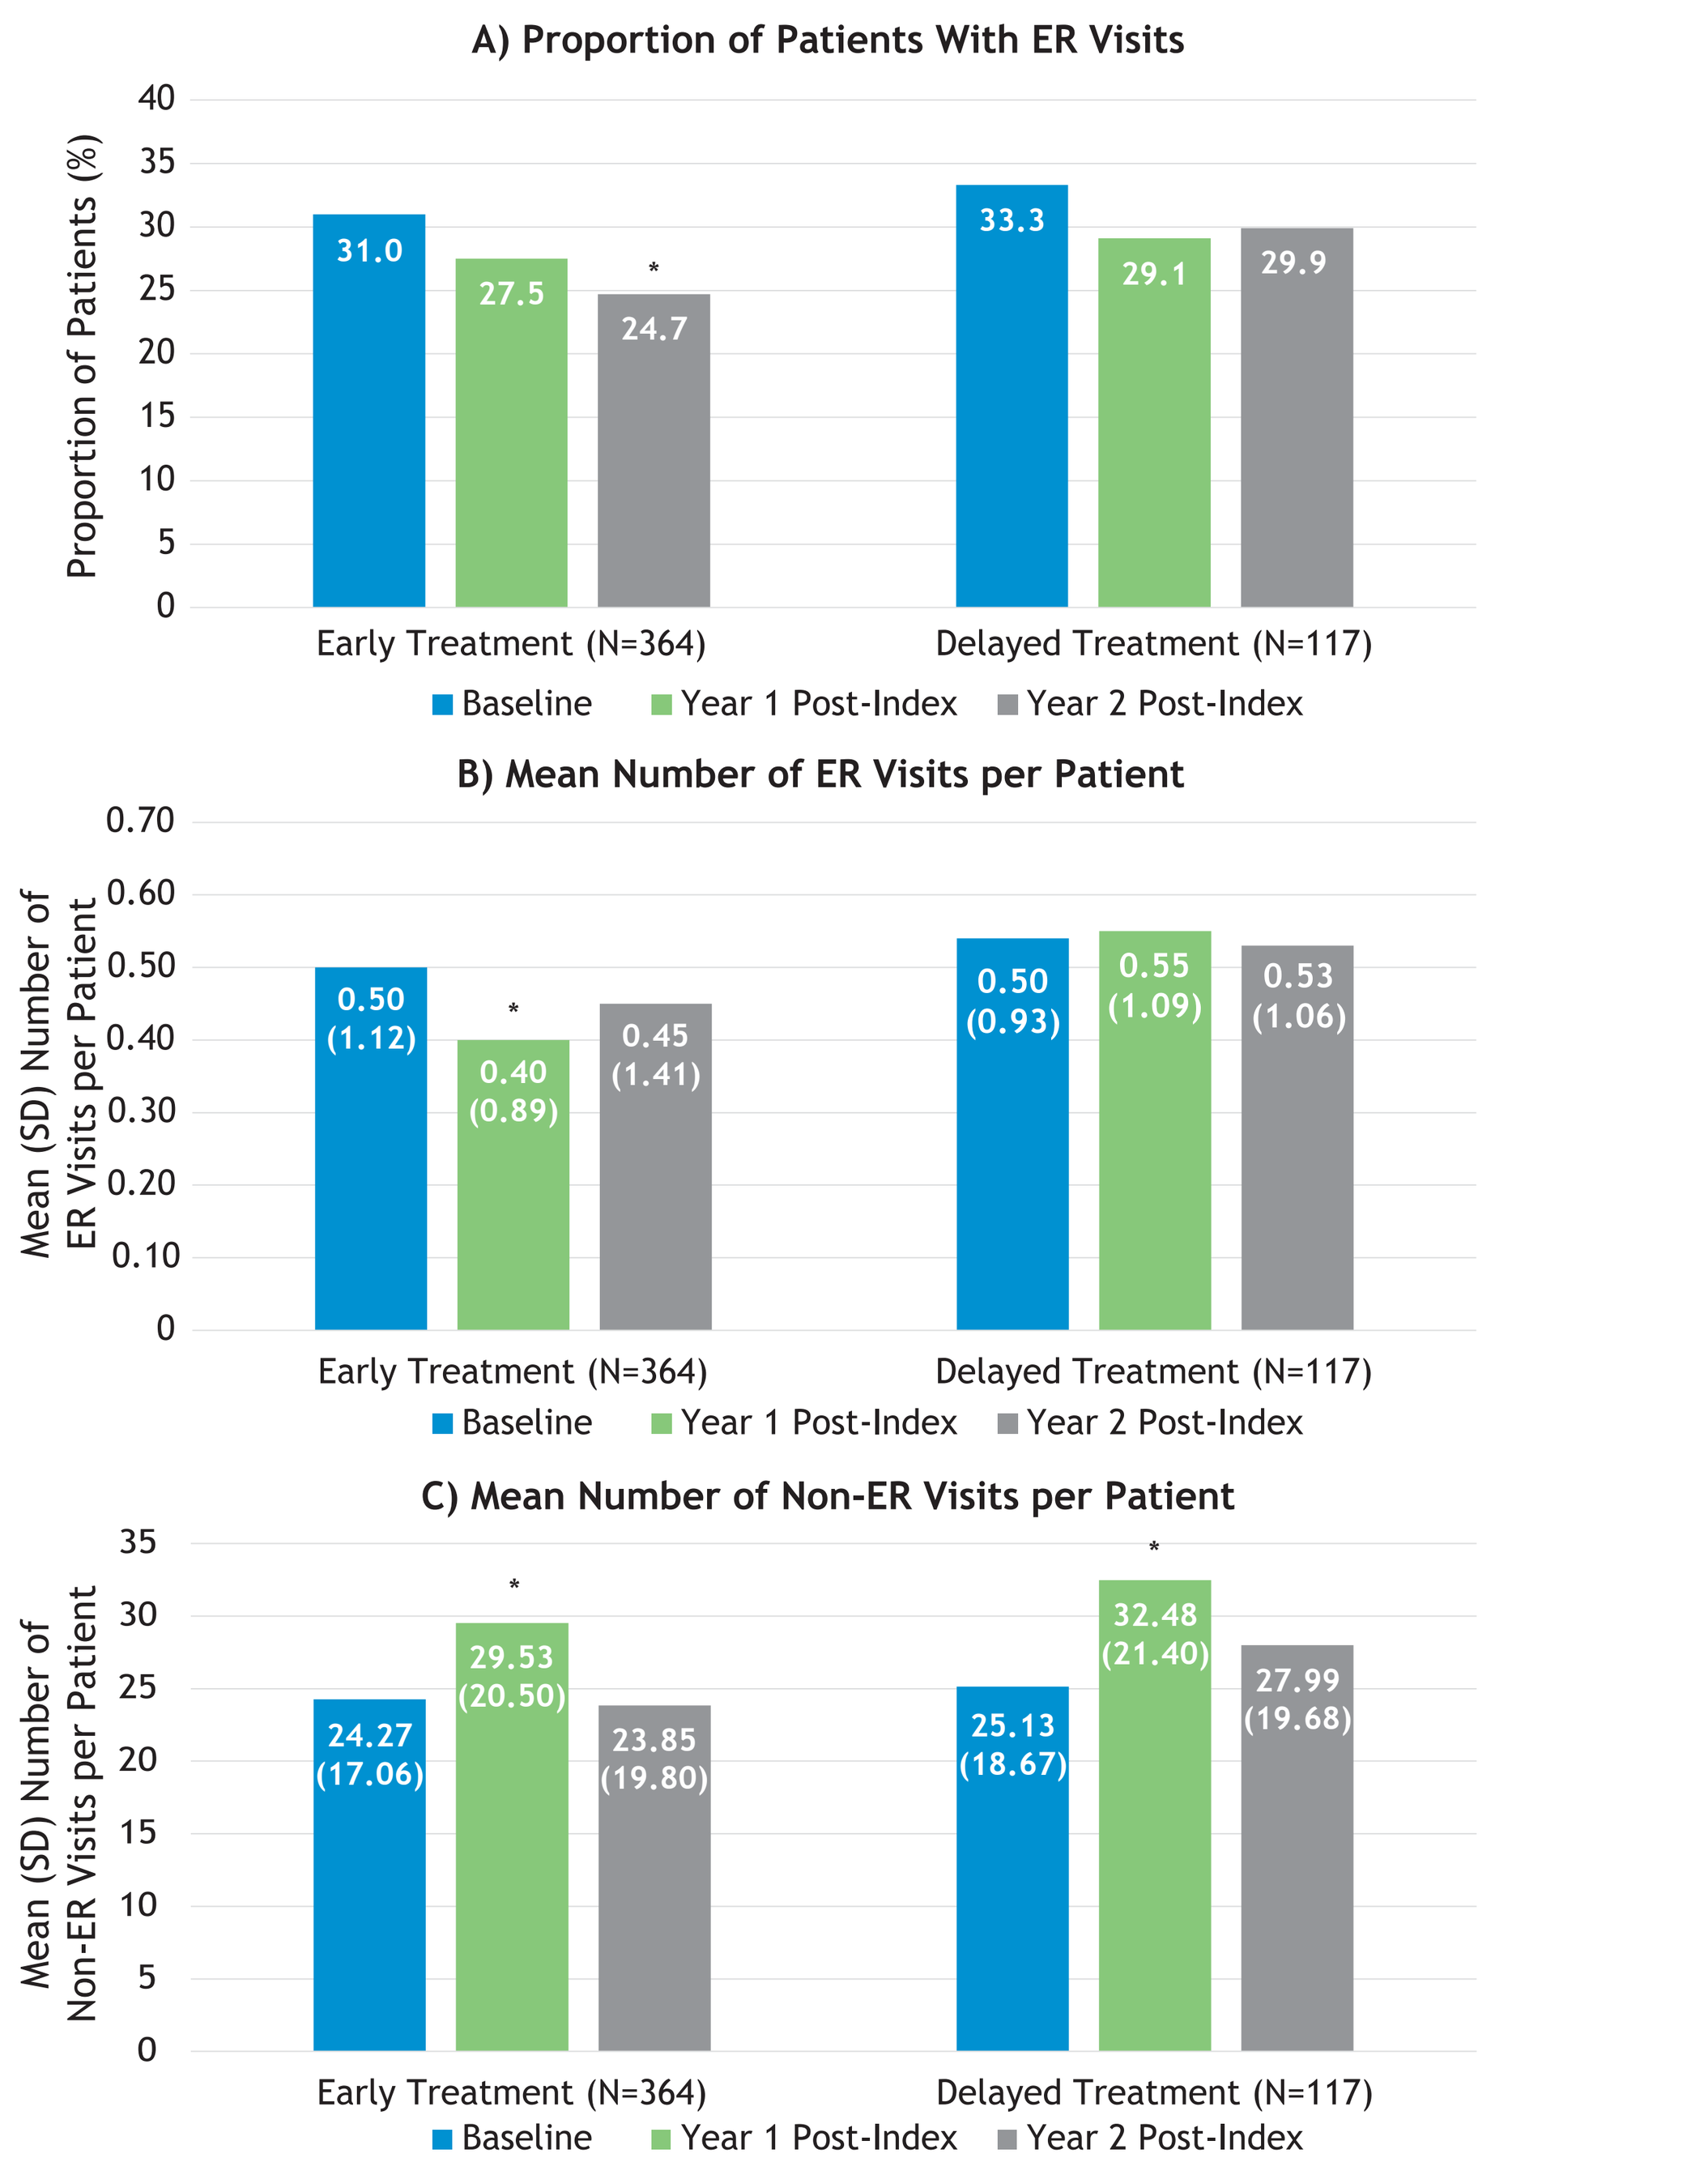

A) Proportion of Patients With ER Visits
Proportion of Patients (%)
31.0
27.5
24.7
33.3
29.1
29.9
B) Mean Number of ER Visits per Patient
Mean (SD) Number of ER Visits per Patient
0.50 (1.12)
0.40 (0.89)
0.45 (1.41)
0.50 (0.93)
0.55 (1.09)
0.53 (1.06)
C) Mean Number of Non-ER Visits per Patient
Mean (SD) Number of Non-ER Visits per Patient
24.27 (17.06)
29.53 (20.50)
23.85 (19.80)
25.13 (18.67)
32.48 (21.40)
27.99 (19.68)
Early Treatment (N=364)
Delayed Treatment (N=117)
Baseline
Year 1 Post-Index
Year 2 Post-Index

*$P<0.05$. McNemar X^2 test used to compare proportions of ER visits at year 1 and year 2 versus baseline (panel A); Wilcoxon signed rank test used to compare mean number of ER visits at year 1 and year 2 versus baseline (panels B and C). Index date: date of first NTMLD diagnosis. ER, emergency room; NTMLD, nontuberculous mycobacterial lung disease; SD, standard deviation.

LIMITATIONS

- Because this was a retrospective claims analysis, data on sputum conversion, clinical symptoms, quality of life, and treatment adherence were lacking. However, HCRU is considered a good indicator of outcomes in patients diagnosed with NTMLD.
- The study population was comprised of commercially-insured patients with continuous enrollment only, and so may not be generalizable to patients without insurance coverage or with other types of insurance.

CONCLUSIONS

- In the first 2 years following NTMLD diagnosis, significant reductions in all-cause and respiratory-related hospitalizations and ER visits were observed in patients who received early antibiotic therapy.
- In patients with delayed-treatment initiation, no statistically significant reductions in all-cause and respiratory-related hospitalization burden were observed. Notably, statistically significant increases in the proportion of patients with hospitalizations were observed at year 1 versus baseline. In addition, little-to-no improvements in ER visits from baseline were observed in years 1 and 2 post-index date.
- Findings suggest that delaying antibiotic treatment in patients diagnosed with NTMLD does not result in significant improvements in HCRU over time.

DISCLOSURES:

Kevin Winthrop has received grant/research support from and is an advisor/consultant to AN2, Insmed Inc., Paratek, and Red Hill Biopharma, and is also an advisor/consultant to Spero Therapeutics. Catherine Waweru, Mariam Hassan, and Anjan Chatterjee are employees of, and hold stock/bonds options in, Insmed Inc. Sara Burns was an employee of Panalgo at the time of the analysis and conducted the formal data analysis for this study.

Please scan this QR (Quick Response) code with your smartphone app to view an electronic version of this poster. If you do not have a smartphone, access the poster via the internet at: https://bit.ly/3SH3G9q
Copies of this poster obtained through QR are for personal use only and may not be reproduced without written permission of the authors.

Poster presented at IDWeek 2022, October 19-23, 2022