IDWeek 2022 | Poster #P430

Oritavancin Activity Against Gram-Positive Pathogens Causing Bloodstream Infections in Hematology/Oncology and Transplant Units in US Medical Centers (2010–2019)

Cecilia G. Carvalhaes, Dee Shortridge, Helio S. Sader, Rodrigo E. Mendes
JMI Laboratories, North Liberty, Iowa, USA

Introduction

- Bacterial bloodstream infections (BSI) complicate the course of immunocompromised patients, significantly contributing to morbidity and mortality.
- Rising antimicrobial resistance rates may impact the efficacy of empirical and target antibiotic treatment in high-risk patients undergoing hematologic, oncologic, and/or transplant treatment.
- Appropriate antimicrobial management is crucial for patients with suspected or confirmed BSI.
- We evaluated the *in vitro* activity of oritavancin and comparators against Gram-positive isolates causing BSI in patients from hematology/oncology and transplant units (HTU) in US medical centers.

Methods

- A total of 1,217 Gram-positive isolates causing bloodstream infections in HTU patients were consecutively collected during 2010–2019 as part of the SENTRY Antimicrobial Surveillance Program.
- A single isolate per patient was collected in 33 US medical centers located in all 9 US Census Divisions.
- Only isolates determined to be significant by local criteria as the reported probable cause of infection were included in the program.
- Bacterial identification was performed by MALDI-TOF (Bruker Daltonics, Billerica, MA, USA) and/or standard microbiological testing methods.
- Antimicrobial susceptibility testing was performed using CLSI broth microdilution methodology in a central laboratory (JMI Laboratories).
- CLSI M100 (2022) breakpoints were applied for comparator agents.
- For *in vitro* comparisons, the oritavancin CLSI susceptible breakpoints for *S. aureus* (≤0.12 mg/L) and vancomycin-susceptible *E. faecalis* (≤0.12 mg/L) were applied to all *Staphylococcus* spp. and *Enterococcus* spp. isolates, respectively.
- The oritavancin CLSI susceptible breakpoints were applied for β-hemolitic streptococci (BHS; ≤0.25 mg/L) and Viridans group streptococci (VGS; ≤0.25mg/L).
- Oritavanin and comparators were also evaluated against MRSA, MRCoNS, and VRE resistant subsets, as well as, a subset of susceptible dose-dependent *E. faecium* isolates displaying high daptomycin MIC values (2–4 mg/L).

Table 1. Activity of oritavancin and comparators against Gram-positive pathogens causing BSI in HTU patients in US medical centers

Organism group (no. of isolates)	Oritavancin			Vancomycin			Linezolid			Daptomycin		
	MIC_{50}	MIC_{90}	%S	MIC_{50}	MIC_{90}	%S	MIC_{50}	MIC_{90}	%S	MIC_{50}	MIC_{90}	%S
E. faecalis (174)	0.015	0.03	98.9[a]	1	2	97.1	1	2	100.0	1	1	98.3
E. faecium (246)	0.03	0.12	96.7[a]	>16	>16	27.2	1	2	100.0	2	2	99.2[b]
VRE (179)	0.03	0.12	95.5[a]	>16	>16	0.0	1	2	100.0	2	2	98.9[b]
Dapto MIC 2-4 mg/L (136)	0.03	0.12	95.6[a]	>16	>16	31.6	1	2	100.0	2	4	100.0[b]
S. aureus (434)	0.03	0.06	99.3	1	1	100.0	1	2	100.0	0.25	0.5	100.0
MRSA (158)	0.03	0.06	99.4	1	1	100.0	1	1	100.0	0.25	0.5	100.0
CoNS (144)	0.03	0.12	96.5[c]	1	2	100.0	0.5	1	100.0	0.25	0.5	100.0
MR-CoNS (110)	0.06	0.12	95.5[c]	2	2	100.0	0.5	1	100.0	0.5	0.5	100.0
BHS (42)	0.03	0.25	95.2	0.5	0.5	100.0	1	1	100.0	0.12	0.25	100.0
VHS (117)	0.015	0.25	96.6	0.5	0.5	100.0	1	1	99.1	0.25	0.5	100.0

[a] Oritavancin breakpoint published for vancomycin-susceptible *E. faecalis* (≤0.12 mg/L) was applied to all *Enterococcus* isolates (CLSI, 2022).
[b] Susceptible dose-dependent.
[c] Oritavancin breakpoint published for *S. aureus* (≤0.12 mg/L) was applied to all CoNS isolates (CLSI, 2022).

Results

- *Enterococcus* spp. (overall, 36.1%; *E. faecium*, 20.2%; *E. faecalis*, 14.3%) and *S. aureus* (35.7%; 36.4% methicillin-resistant [MRSA]) were the most common organism groups, followed by coagulase-negative *Staphylococcus* (CoNS; 11.8%; 76.4% MR), VGS (9.6%), and BHS (3.5%; Figures 1 and 2).

Activity against enterococci

- Oritavancin inhibited 96.7% and 98.9% of *E. faecium* and *E. faecalis* at ≤0.12 mg/L, respectively (susceptible breakpoint for vancomycin-susceptible *E. faecalis*; Figure 3).
- Ampicillin (100.0% susceptible [S]), linezolid (100.0%S), daptomycin (98.3%S), and vancomycin (97.1%S) were also active against *E. faecalis* (Figure 3).
- Oritavancin (96.7% inhibited at ≤0.12 mg/L) and linezolid (100.0%S) remained active against *E. faecium* isolates, while only 27.2% were susceptible to vancomycin and 99.2% were daptomycin susceptible dose-dependent (Figure 3).
- Vancomycin-resistant *Enterococcus* (VRE) phenotype was noted in 72.8% of *E. faecium*, and oritavancin inhibited 95.5% of these isolates at ≤0.12 mg/L (Figures 2 and 3).
- Oritavancin inhibited 95.6% of *E. faecium* isolates displaying elevated daptomycin MIC (2–4 mg/L) values, which were noted in 55.3% of all *E. faecium* isolates (Figures 2 and 3).

Activity against staphylococci

- Oritavancin displayed equivalent $MIC_{50/90}$ (0.03/0.06 mg/L) values against MSSA (99.3%S) and MRSA (99.4%S; Table 1 and Figure 4).
- Oritavancin inhibited 95.5% of MRCoNS at ≤0.12 mg/L (*S. aureus* susceptible breakpoint).
- Vancomycin, daptomycin, and linezolid remained active against MRSA and MRCoNS (Figure 4).

Activity against streptococci

- Oritavancin was also active against BHS ($MIC_{50/90}$, 0.03/0.25 mg/L; 95.2%S) and VGS ($MIC_{50/90}$, 0.015/0.25 mg/L; 96.6%S), as was vancomycin (100.0%S), daptomycin (100.0%S), and linezolid (100.0%/99.1%S, respectively; Table 1 and Figure 4).

Figure 1. Distribution of Gram-positive pathogens causing BSI in HTU patients in US medical centers (2010–2019)

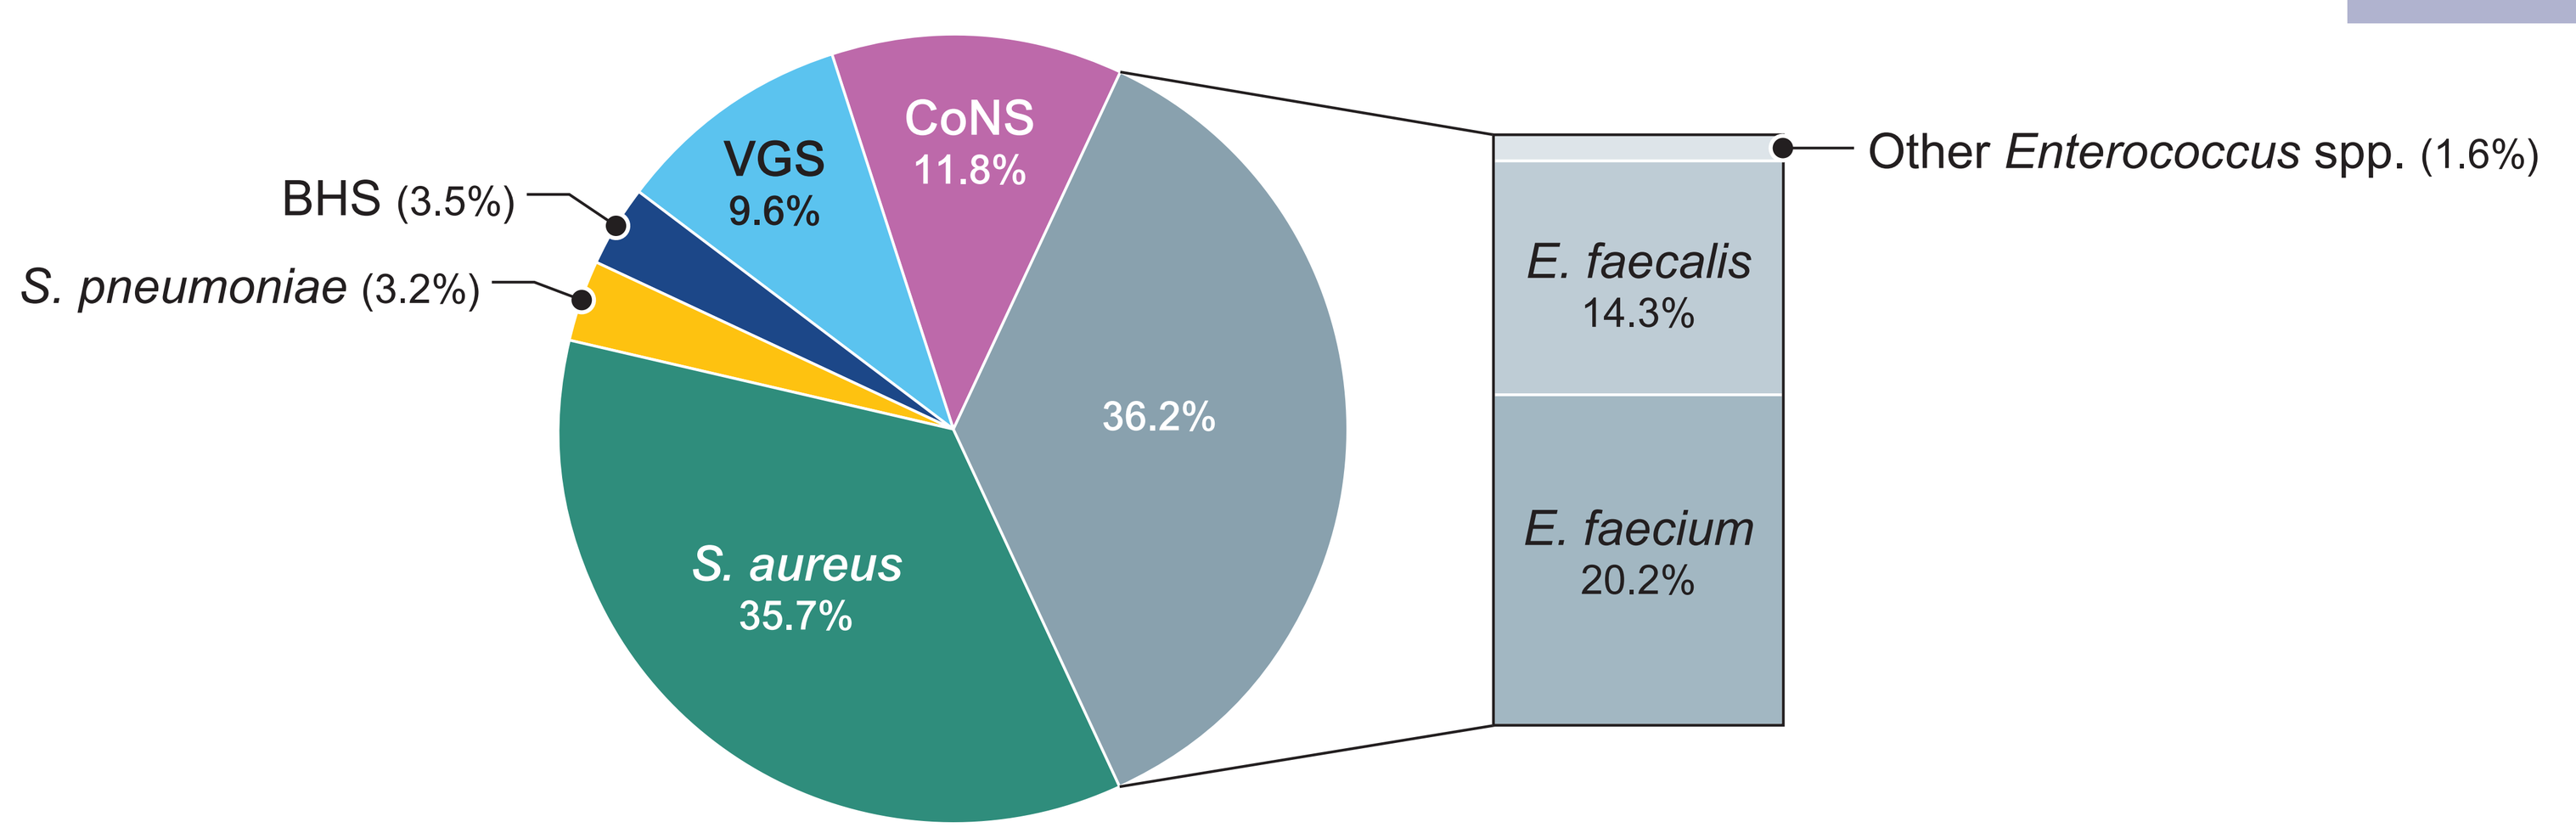

BHS (3.5%)
S. pneumoniae (3.2%)
VGS 9.6%
CoNS 11.8%
36.2%
S. aureus 35.7%
Other Enterococcus spp. (1.6%)
E. faecalis 14.3%
E. faecium 20.2%

Figure 2. Resistant phenotype rates observed in Gram-positive pathogens causing BSI in HTU patients in US medical centers (2017–2019)

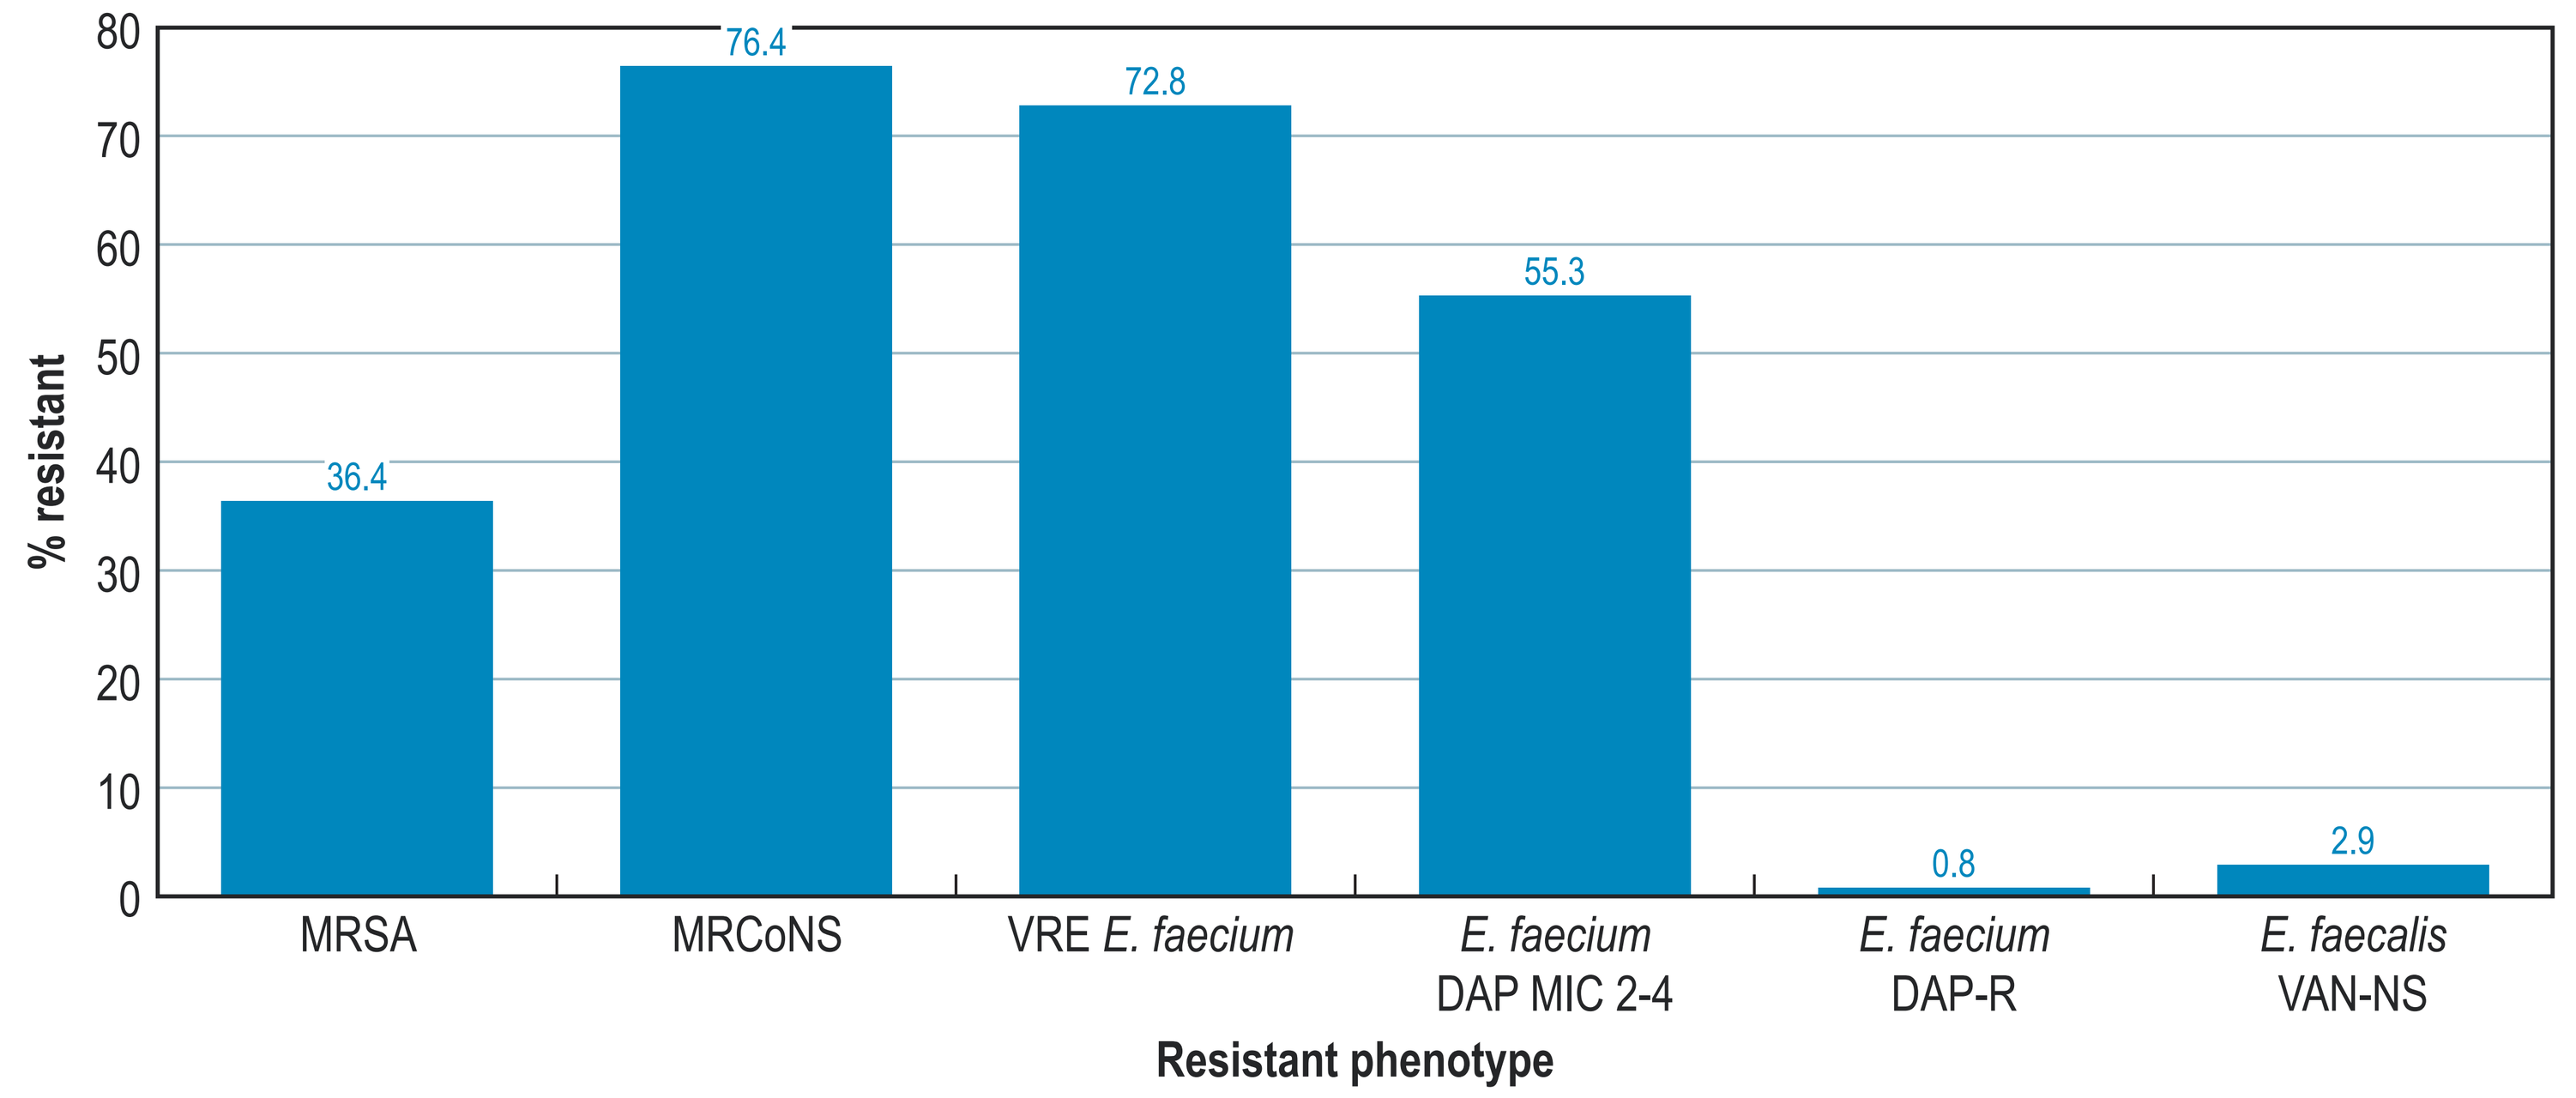

% resistant
36.4
76.4
72.8
55.3
0.8
2.9
MRSA
MRCoNS
VRE E. faecium
E. faecium DAP MIC 2-4
E. faecium DAP-R
E. faecalis VAN-NS
Resistant phenotype

Abbreviations: MRSA, methicillin-resistant *S. aureus*; MRCoNS, methicillin-resistant coagulase-negative *Staphylococcus* spp.; VRE, vancomycin-resistant enterococci; DAP, daptomycin; DAP-R, daptomycin-resistant; VAN-NS, vancomycin-nonsusceptible.

Figure 3. Oritavancin and comparators' susceptibility rates against *Enterococcus* spp. and resistant phenotypes

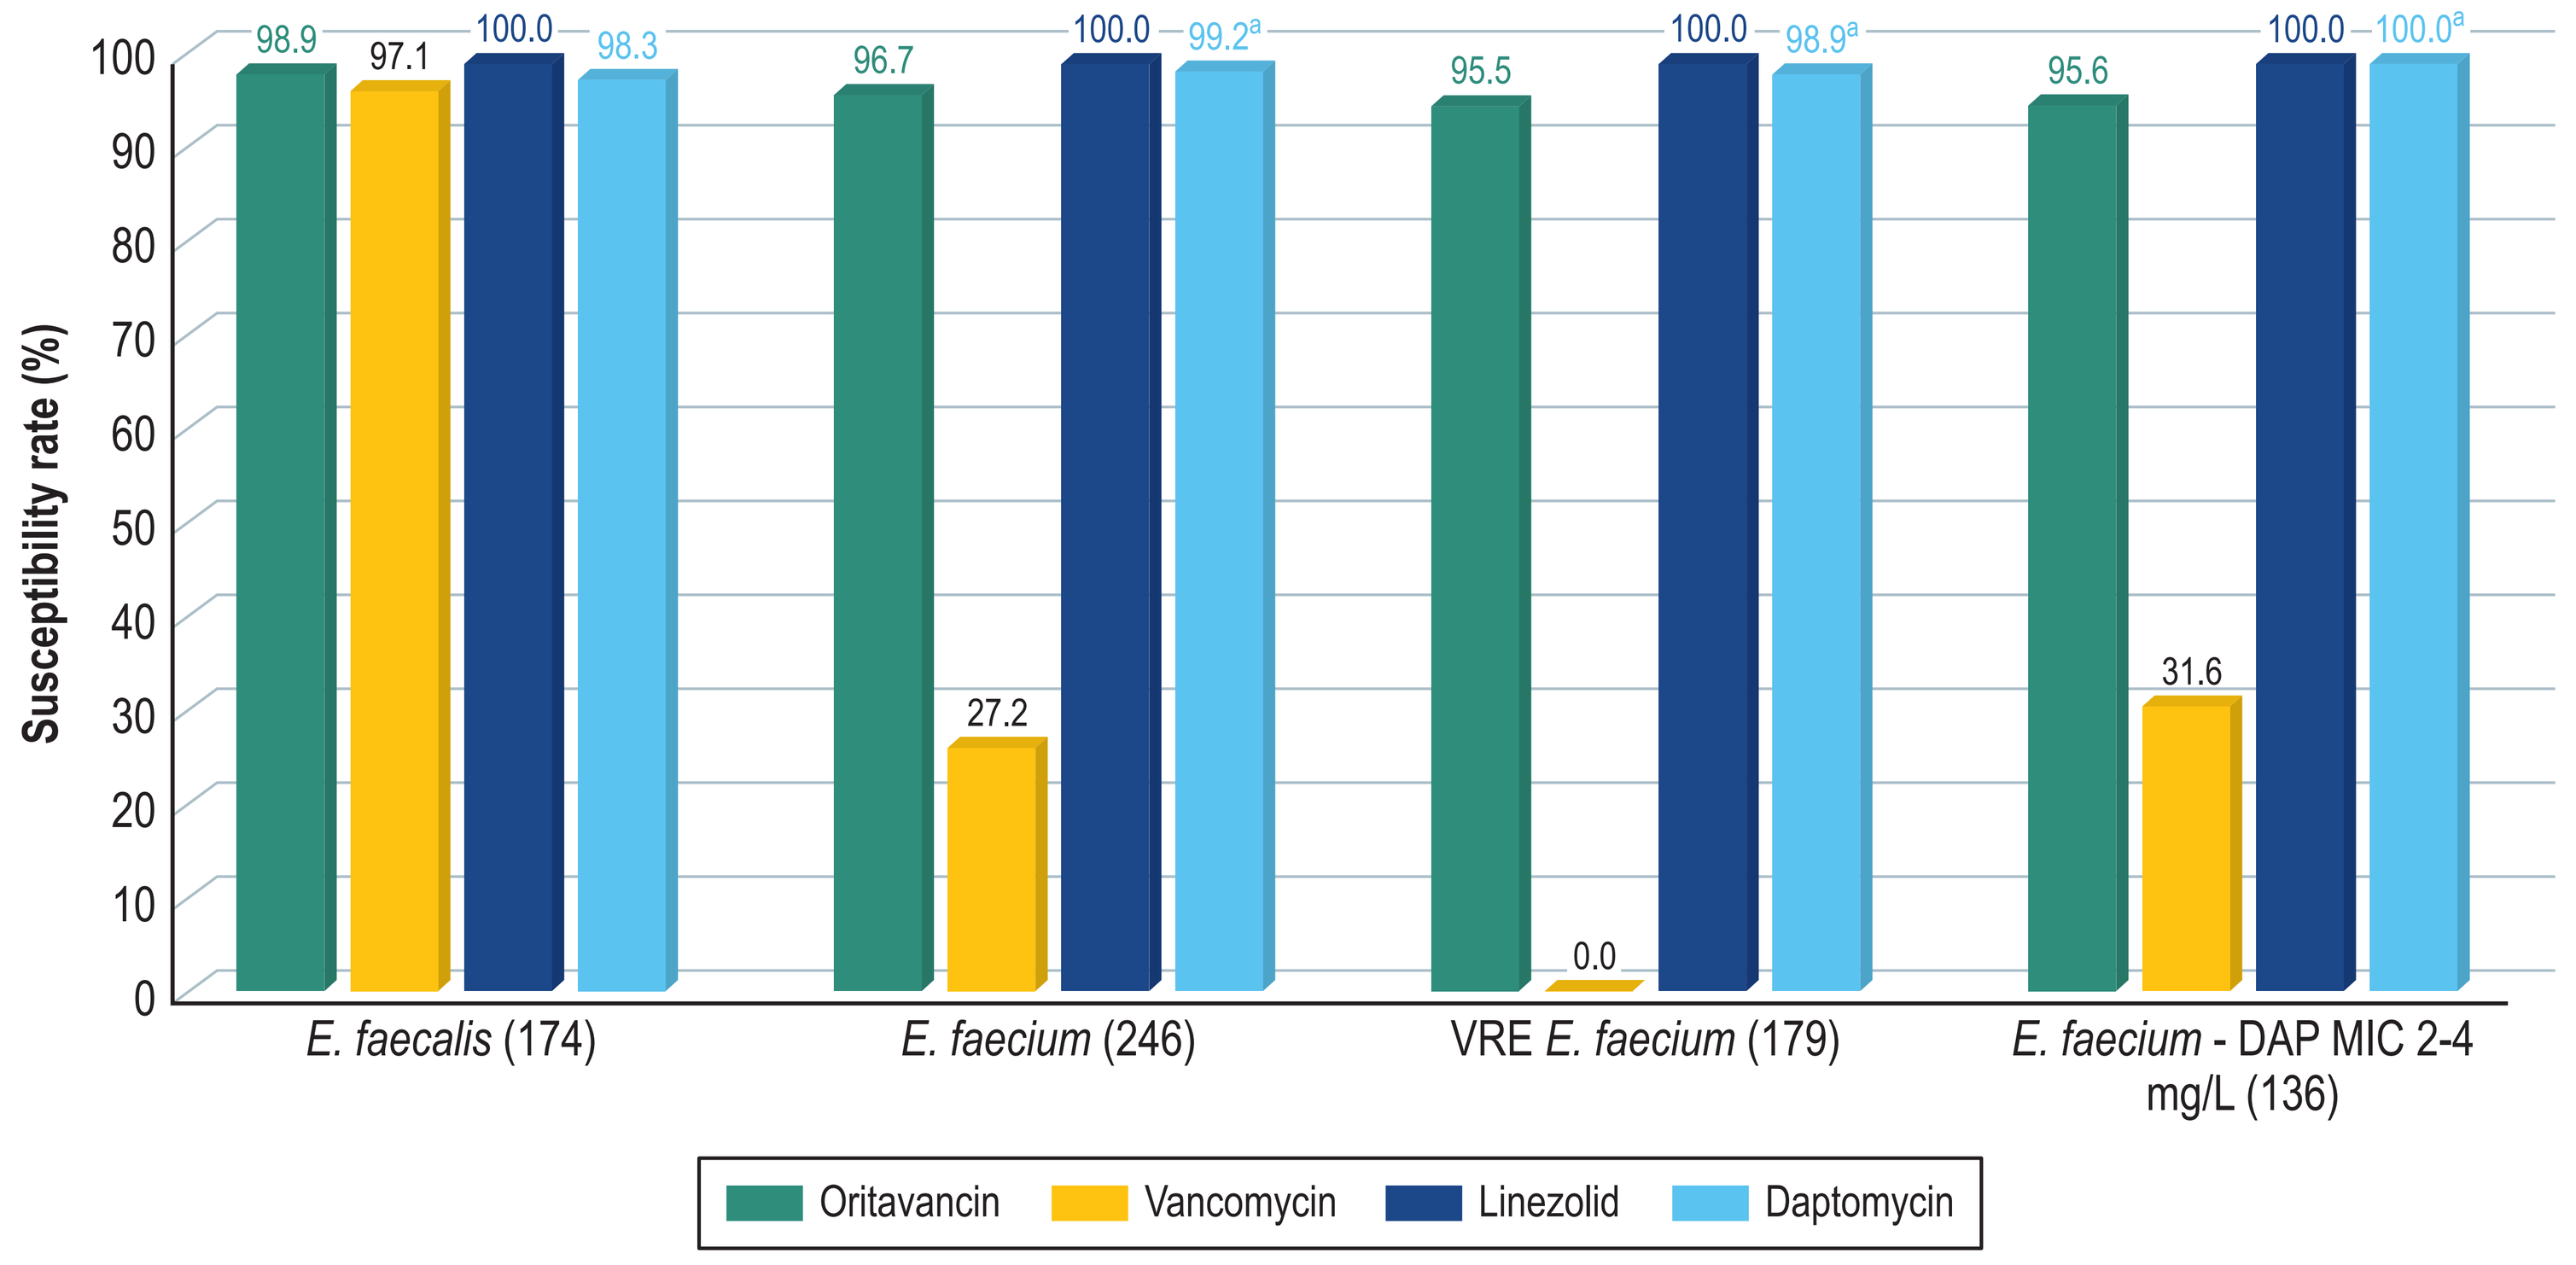

Susceptibility rate (%)
98.9 97.1 100.0 98.3
96.7 27.2 100.0 99.2a
95.5 0.0 100.0 98.9a
95.6 31.6 100.0 100.0a
E. faecalis (174)
E. faecium (246)
VRE E. faecium (179)
E. faecium - DAP MIC 2-4 mg/L (136)
Oritavancin
Vancomycin
Linezolid
Daptomycin

Abbreviation: VRE, vancomycin-resistant enterococci.
[a] Susceptible dose-dependent.
The oritavancin breakpoint published for vancomycin-susceptible *E. faecalis* (≤0.12 mg/L) was applied to all *Enterococcus* isolates per CLSI M100 (2022) criteria.

Figure 4. Susceptibility rates of oritavancin and comparators against staphylococci and streptococci causing BSI in HTU patients in US medical centers

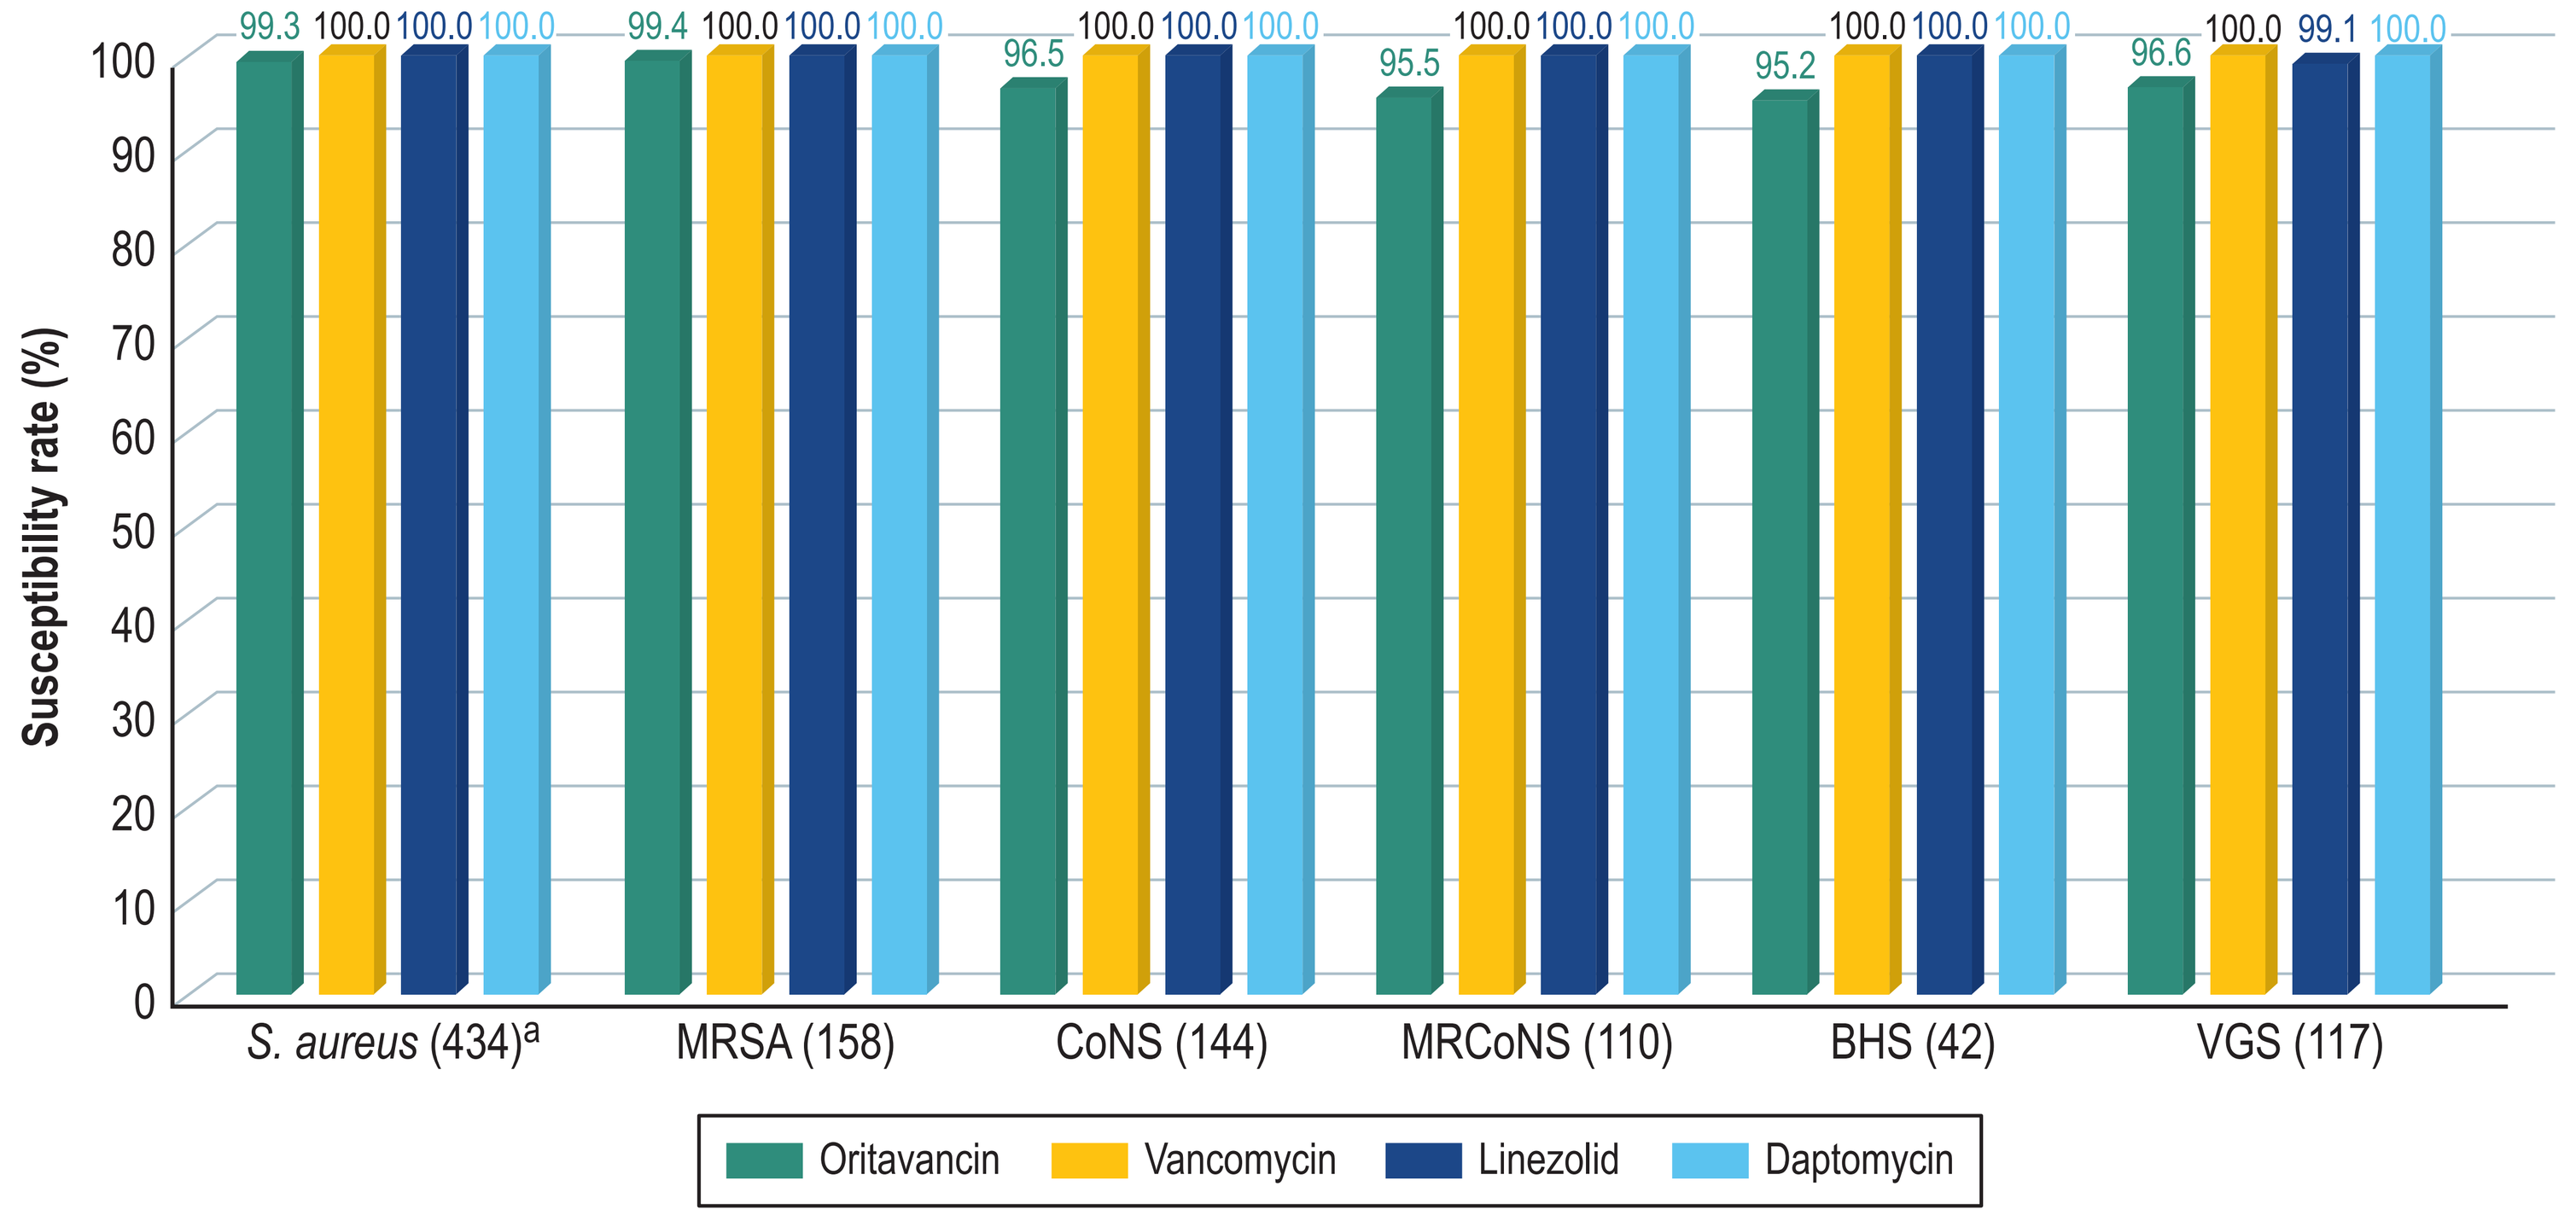

Susceptibility rate (%)
99.3 100.0 100.0 100.0
99.4 100.0 100.0 100.0
96.5 100.0 100.0 100.0
95.5 100.0 100.0 100.0
95.2 100.0 100.0 100.0
96.6 100.0 99.1 100.0
S. aureus (434)a
MRSA (158)
CoNS (144)
MRCoNS (110)
BHS (42)
VGS (117)
Oritavancin
Vancomycin
Linezolid
Daptomycin

Abbreviations: MRSA, methicillin-resistant *S. aureus*; CoNS, coagulase-negative *Staphylococcus* spp.; MRCoNS, methicillin-resistant coagulase-negative *Staphylococcus* spp.; BHS, β-hemolytic streptococci; VGS, Viridans group streptococci.
[a] The oritavancin susceptible breakpoint published for *S. aureus* (≤0.12 mg/L) was applied to all CoNS isolates per CLSI M100 (2022) criteria.

Conclusions

- Overall, 36.1% of the isolates were *Enterococcus* spp., 20.2% were *E. faecium*, 72.8% of *E. faecium* were resistant to vancomycin, and 55.3% displayed daptomycin MIC values of 2–4 mg/L.
- High resistance rates were noted among Gram-positive pathogens recovered from BSI in HTU patients in US, including MRSA (36.4%), MRCoNS (76.4%), and VRE in *E. faecium* (72.8%).
- Oritavancin was active against *S. aureus* and CoNS (>95.5% inhibited at ≤0.12 mg/L), including MRSA and MRCoNS.
- Oritavancin inhibited >96% of *E. faecalis* and *E. faecium* isolates at ≤0.12 mg/L, including >95% of isolates within resistant subsets, such as VRE *E. faecium* and *E. faecium* displaying elevated daptomycin MIC values (2–4 mg/L).

Funding

This study was supported by Melinta Therapeutics. CG Carvalhaes, D Shortridge, HS Sader, and RE Mendes are employees of JMI Laboratories, which was paid consultant to Melinta in connection with the development of this poster.

Acknowledgments

The authors thank the participant centers for providing isolates.

References

Carvalhaes CG, Sader HS, Streit JM, et al. Activity of Oritavancin against Gram-Positive Pathogens Causing Bloodstream Infections in the United States over 10 Years: Focus on Drug-Resistant Enterococcal Subsets (2010–2019). Antimicrob. Agents Chemother. 2022 Feb 15; 66(2) :e0166721.

CLSI. 2022. M100Ed32. Performance standards for antimicrobial susceptibility testing: 32nd informational supplement. Clinical and Laboratory Standards Institute, Wayne, PA.

Freifeld AG, Bow EJ, Sepkowitz KA, et al. Infectious Diseases Society of America. Clinical practice guideline for the use of antimicrobial agents in neutropenic patients with cancer: 2010 update by the Infectious Diseases Society of America. Clin. Infect. Dis. 2011 Feb 15; 52(4): e56-93.

Zimmer AJ, Stohs E, Meza J, Arnold C, et al. Bloodstream Infections in Hematologic Malignancy Patients with Fever and Neutropenia: Are Empirical Antibiotic Therapies in the United States Still Effective? Open Forum Infect. Dis. 2022 May 18; 9(7): ofac240.

Contact

Cecilia Carvalhaes, MD, Ph.D., D(ABMM)
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: cecilia-carvalhaes@jmilabs.com

SCAN ME

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2022_OritaVsGPs.pdf

No personal information is stored.

IDWeek 2022, October 19–23, 2022, Washington, D.C.