Poster No. 593
IDWeek 2022
Washington, D.C.
October 19–23, 2022

Population Pharmacokinetic Model Development for Epetraborole and MAC Lung Disease Patients Using Data from Phase 1 and 2 Studies

H. Ganesan[1], M.C. Safir[1], S.M. Bhavnani[1], K.M. Krause[2], C.M. Rubino[1]
[1]Institute for Clinical Pharmacodynamics, Inc., Schenectady, NY; [2]AN2 Therapeutics,Menlo Park, CA

AN2Therapeutics
Menlo Park, CA 94027
www.an2therapeutics.com

ABSTRACT

Background: Epetraborole (EBO), an orally available bacterial leucyl transfer RNA synthetase inhibitor with potent activity against nontuberculous mycobacteria, is under clinical development for treatment of MAC lung disease. A population pharmacokinetic (PK) model describing the disposition of EBO after oral (PO) and intravenous (IV) administration was developed to support EBO PK-PD analyses and dose selection for patients with MAC lung disease.

Methods: Model development was conducted using NONMEN (v.7.4.3). Models were attempted for oral absorption, systemic compartments, and linear vs. non-linear elimination. Model evaluation involved goodness-of fit plots and prediction-corrected visual predictive plots, which describe the ability of model-based simulations to capture the observed data. Included were data from 5 Phase 1 (3 IV and 2 PO) and 2 Phase 2 studies (IV only) **(Table 1**).

Results: The pooled dataset included 2637 EBO PK samples from 138 subjects/patients. A robust fit to observed data across studies was obtained using a three-compartment model with linear elimination (**Table 2**). The impact of body weight on PK was included using an allometric scaling approach to accommodate observed lower body weight in MAC lung disease patients. PO dosing was modeled using an absolute bioavailability term (F) and transit compartments with separate absorption rates for fed and fasted administration. Interindividual variability (IIV) in systemic clearance was low (7.9%), but IIV in F (32.7%) contributed to slightly higher variability in PK for PO vs. IV administration. Moderate shrinkage was observed for the IIV in the model parameters. This was considered acceptable given inclusion of patients with limited PK data in the model and the objective to facilitate simulations designed to inform dose selection. The prediction-corrected visual predictive check plots for the data obtained from a recently completed Phase 1b study evaluating 28-day oral dosing regimens (NCT04892631) are provided in **Figure 1**.

Conclusion: This model is useful for describing expected PK in MAC lung disease patients and was used to conduct simulations for the advancement of the oral EBO 500 mg q24 dosing regimen into clinical studies in patients with MAC lung disease.

INTRODUCTION

- Epetraborole (EBO, previously known as GSK2251052 and AN3365) is an orally available benzoxaborole, a boron-heterocyclic antimicrobial class that inhibits leucyl transfer RNA synthetase (LeuRS). LeuRS is an essential enzyme for protein synthesis, and its inhibition stops bacterial growth [1, 2].
- EBO has potent activity against nontuberculous mycobacteria [1, 3, 4].
- EBO has been found to concentrate in alveolar macrophages [5] and is under clinical development for treatment of *Mycobacterium avium* complex (MAC) lung disease.
- The aim of this analysis was to develop a population pharmacokinetic (PK) model describing the disposition of EBO after oral (PO) and intravenous (IV) administration to support EBO pharmacokinetic-pharmacodynamic (PK-PD) analyses and dose selection for patients with MAC lung disease.

METHODS

- Seven studies were used to construct the population PK model. This included five Phase 1 studies, three of which utilized intravenous (IV) administration and two of which utilized oral (PO) administration, and two Phase 2 studies employing IV administration. A brief overview of the studies included are presented in **Table 1**.
- Model development was conducted using NONMEM (v.7.4.3). Models were attempted for oral absorption, systemic compartments, and linear versus non-linear elimination.
- Model evaluation involved goodness-of-fit plots and prediction-corrected visual predictive plots, which describe the ability of model-based simulations to capture the observed data.

METHODS

Table 1. Summary of studies utilized in the population PK analysis

Study Description	Dosing
First-in-human, SAD/MAD	IV only SAD: 200 – 3000 mg MAD: 500 – 1600 mg BID
Phase 1, ELF study, single and multiple doses	IV only 1500 mg (BID with sampling on Day 3 in MAD)
Phase 1, Mass balance	IV only 1500 mg
Phase 1, SAD/MAD, food-effect	PO only SAD: 500, 2000, 4000 mg MAD: 2000, 3000 mg BID for 10 days
Phase 2, cUTI/AP	IV only 750 or 1500 mg BID
Phase 2, cIAI	IV only 750 or 1500 mg BID
Phase 1b, MAD[a]	PO only 250, 500, 750 mg QD and 500 mg QOD

AP, acute pyelonephritis; BID, twice daily; cIAI, complicated intraabdominal infection; cUTI, complicated urinary tract infection; ELF, epithelial lining fluid; IV, intravenous; MAD, multiple ascending dose; PO, by mouth; SAD, single ascending dose; QD, once daily; QOD, once every other day.
a. Study was ongoing during model development; only data from the first four cohorts from that study have been included in the analysis.

RESULTS

- The pooled dataset included 2,637 EBO PK samples from 138 subjects/patients.
- A robust fit to observed data across studies was obtained using a three-compartment model with linear elimination.
- The impact of body weight on PK was included using an allometric scaling approach to accommodate observed lower body weight in MAC lung disease patients.
- PO administration was modeled using an absolute bioavailability term (F) and transit compartments with separate absorption rates for fed and fasted administration.
- Final population PK parameter estimates are provided in **Table 2**. Overall, the model parameters are estimated with acceptable precision.
- Interindividual variability (IIV) in systemic clearance was low (7.9%), but IIV in F (32.7%) contributed to slightly higher variability in PK for PO vs IV administration.
- Moderate shrinkage was observed for the IIV in the model parameters. This was considered acceptable given the inclusion of patients with limited PK data in the model and the objective to facilitate simulations designed to inform dose selection.
- Goodness-of-fit plots (**Figure 1**) indicated that the model provided a precise and unbiased fit to the observed plasma data, regardless of route of administration or dose.
- The prediction–corrected visual predictive check plots for the pooled data and the available data from the Phase 1b study evaluating 28-day oral dosing regimens (NCT04892641) are provided in **Figure 2** and **Figure 3**, respectively.

RESULTS

Table 2. Population PK parameter estimates for the application of the population PK model to the pooled EBO plasma concentration-time data

Parameter	Final Estimate	%SEM	Magnitude of IIV, IOV, or RV	Shrinkage
CL (L/h/70 kg)	15.3	4.91	7.9	44.9
Cld (L/h/70 kg)	23	7.18	—	—
Cld2 (L/h/70 kg)	43.3	6.68	—	—
Vc (L/70 kg)	15.6	6	37.2	30.1
Vp (L/70 kg)	140	3.02	8.68	43.3
Vp2 (L/70 kg)	33.2	9.62	18.9	52.5
Ka, fast (1/h)	2.89	5.33	29.5	42.4
Ka, fed (1/h)	1.34	9.18		
F	0.565	3.07	32.7	46.9
K_{14} (1/h)	0.266	17.2	57.80	57.8
K_{40} (1/h)	0.661	14.7	—	
IOV in Ka	0.0343	26.1	18.5	55.3 (occasion 1) 58.6 (occasion 2)
RV_{plasma}	0.0587	1.72	24.2%	5.04
RV_{ELF}	0.0656	43.2	25.6%	15.7

CL, systemic clearance; CLd/CLd2, distributional clearance for compartments 1 and 2, respectively; F, absolute bioavailability; IIV, interindividual variability; IOV, interoccasion variability; K14, rate constant from central compartment to ELF compartment; K40, rate constant out of the ELF compartment; Ka, fast/Ka, fed, absorption rate constants for fasting and fed states, respectively; %SEM, percent standard error of the mean; RV, residual variability; RV_{elf}, residual variability for ELF; RV_{plasma}, residual variability for plasma; Vc, volume of distribution for the central compartment; Vp/Vp2, volume of distribution for peripheral compartments 1 and 2, respectively.

Figure 1. Goodness-of-fit plots for the application of the population PK model to the pooled EBO concentration-time data

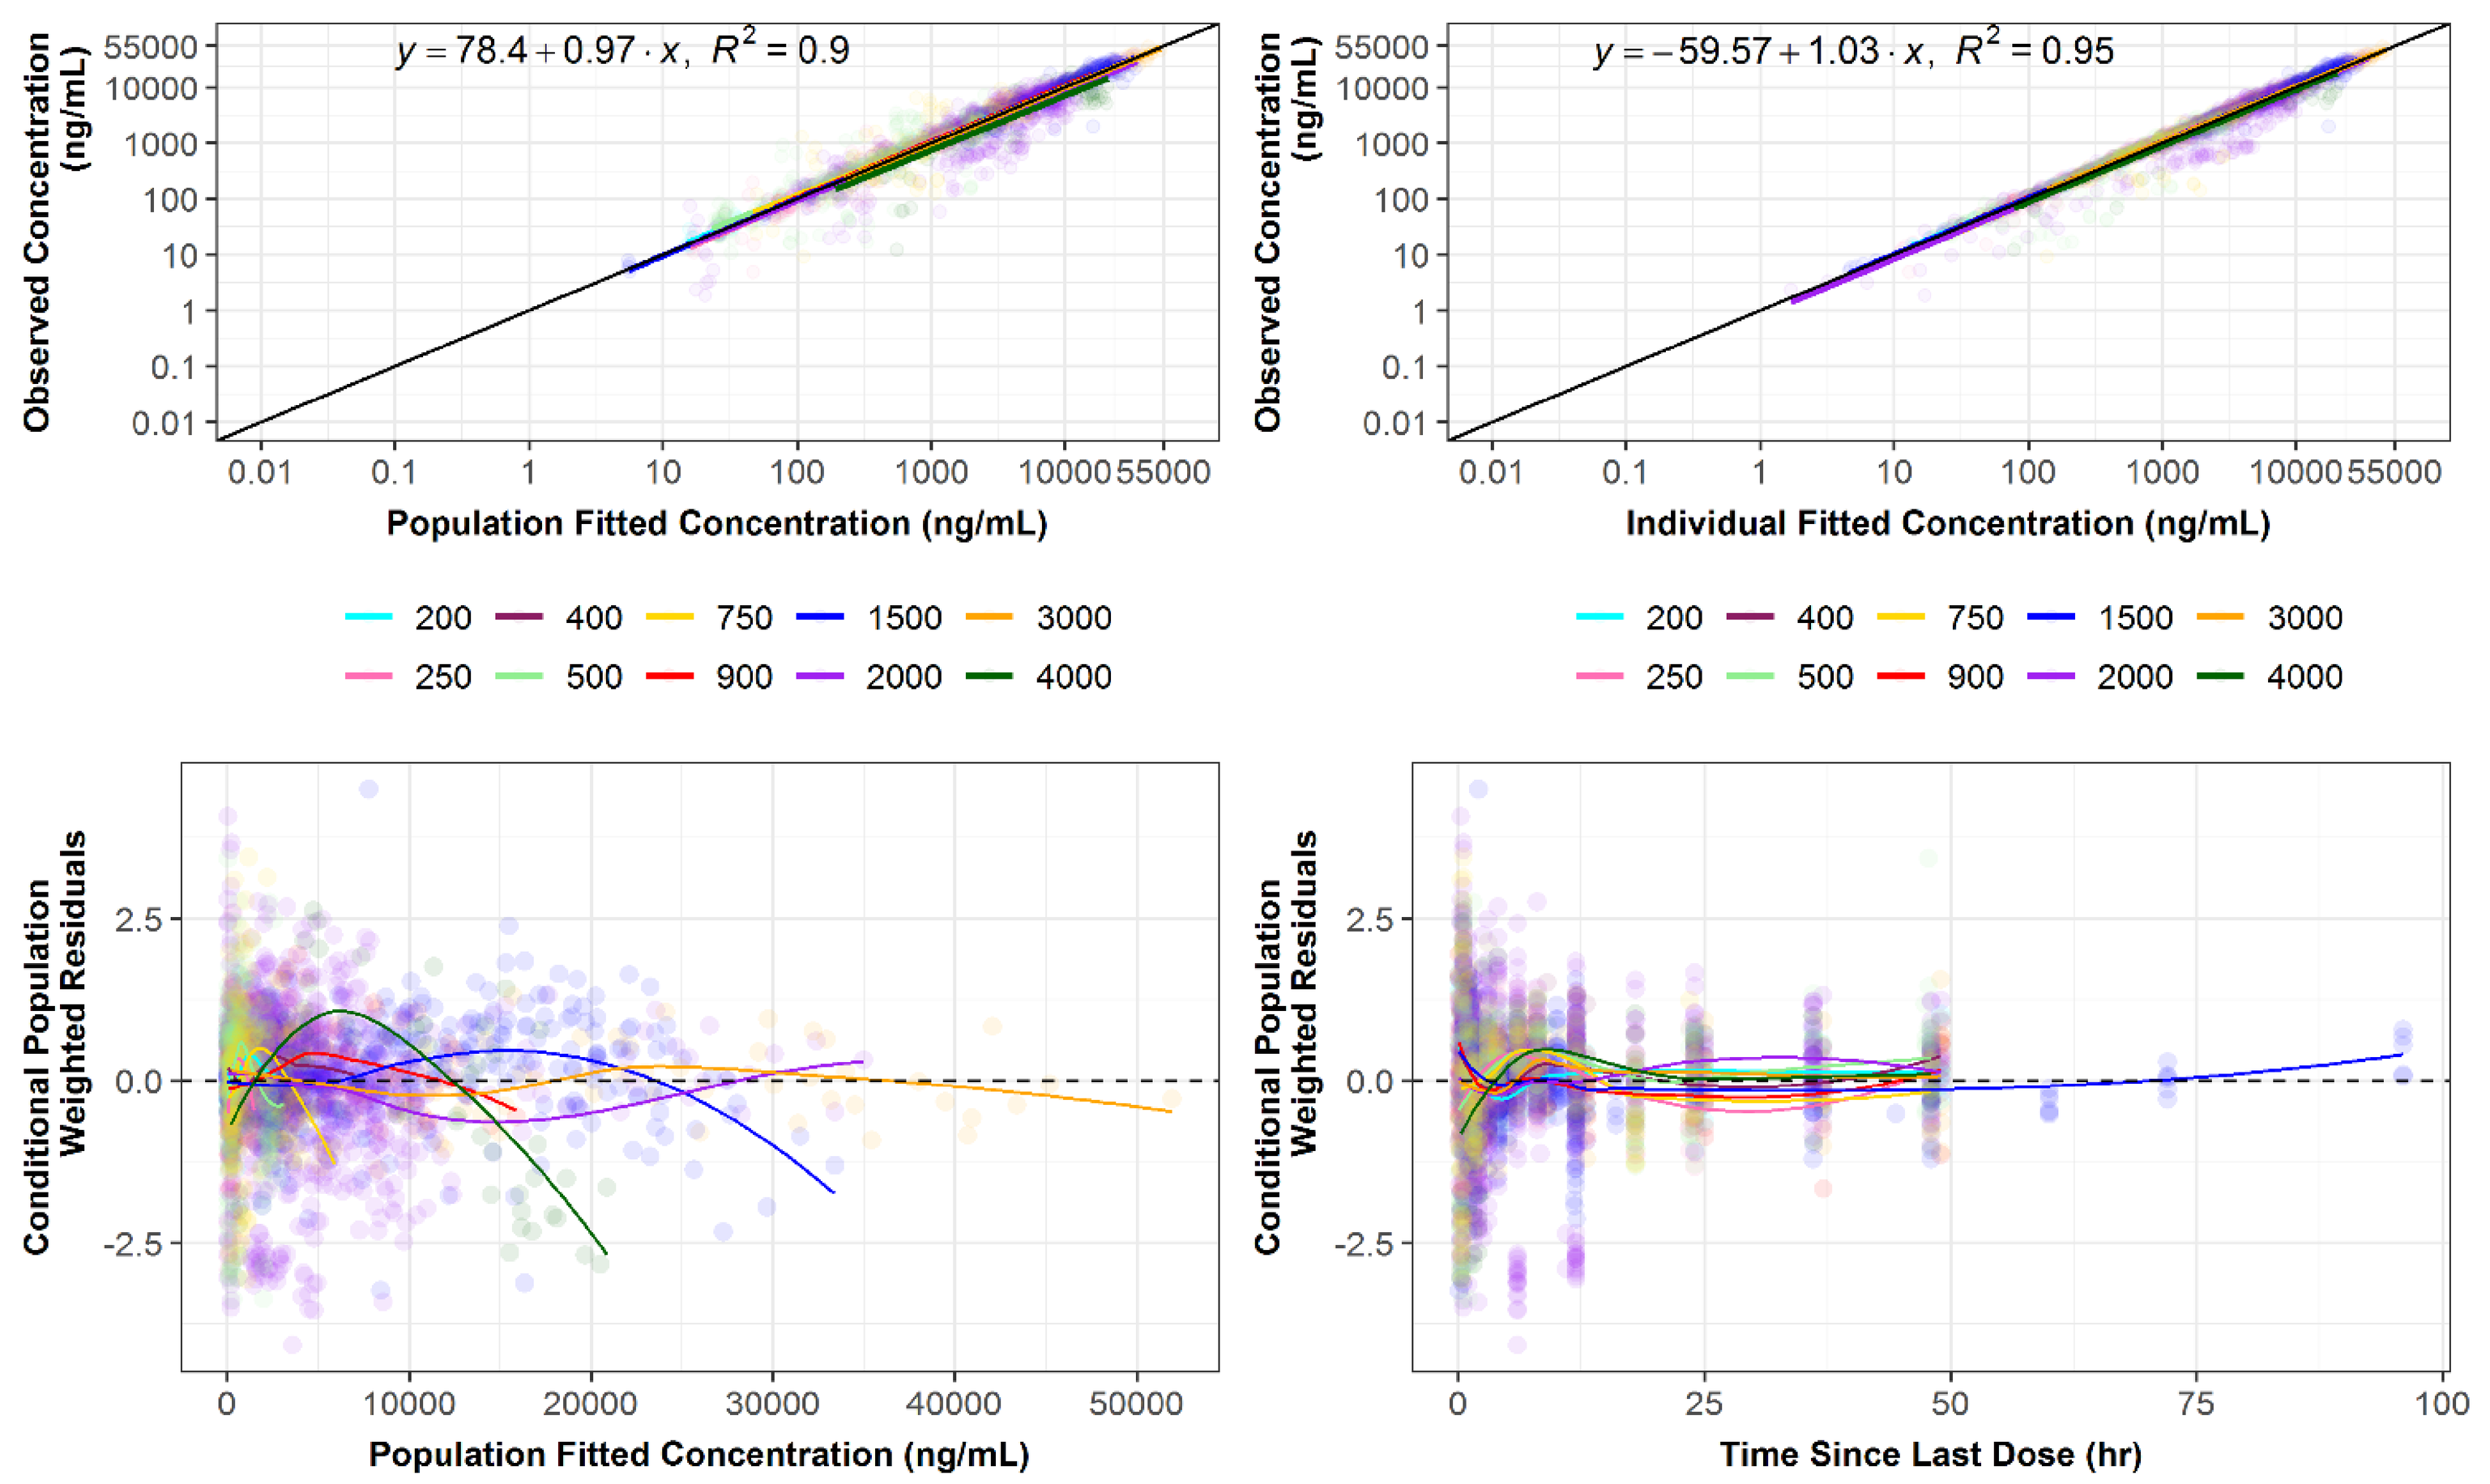
y = 78.4 + 0.97 · x, R² = 0.9
y = −59.57 + 1.03 · x, R² = 0.95
Observed Concentration (ng/mL)
Population Fitted Concentration (ng/mL)
Individual Fitted Concentration (ng/mL)
200 400 750 1500 3000
250 500 900 2000 4000
Conditional Population Weighted Residuals
Time Since Last Dose (hr)

Figure 2. Prediction-corrected visual predictive check plots for the application of the population PK model to the pooled EBO plasma concentration-time data (all data)

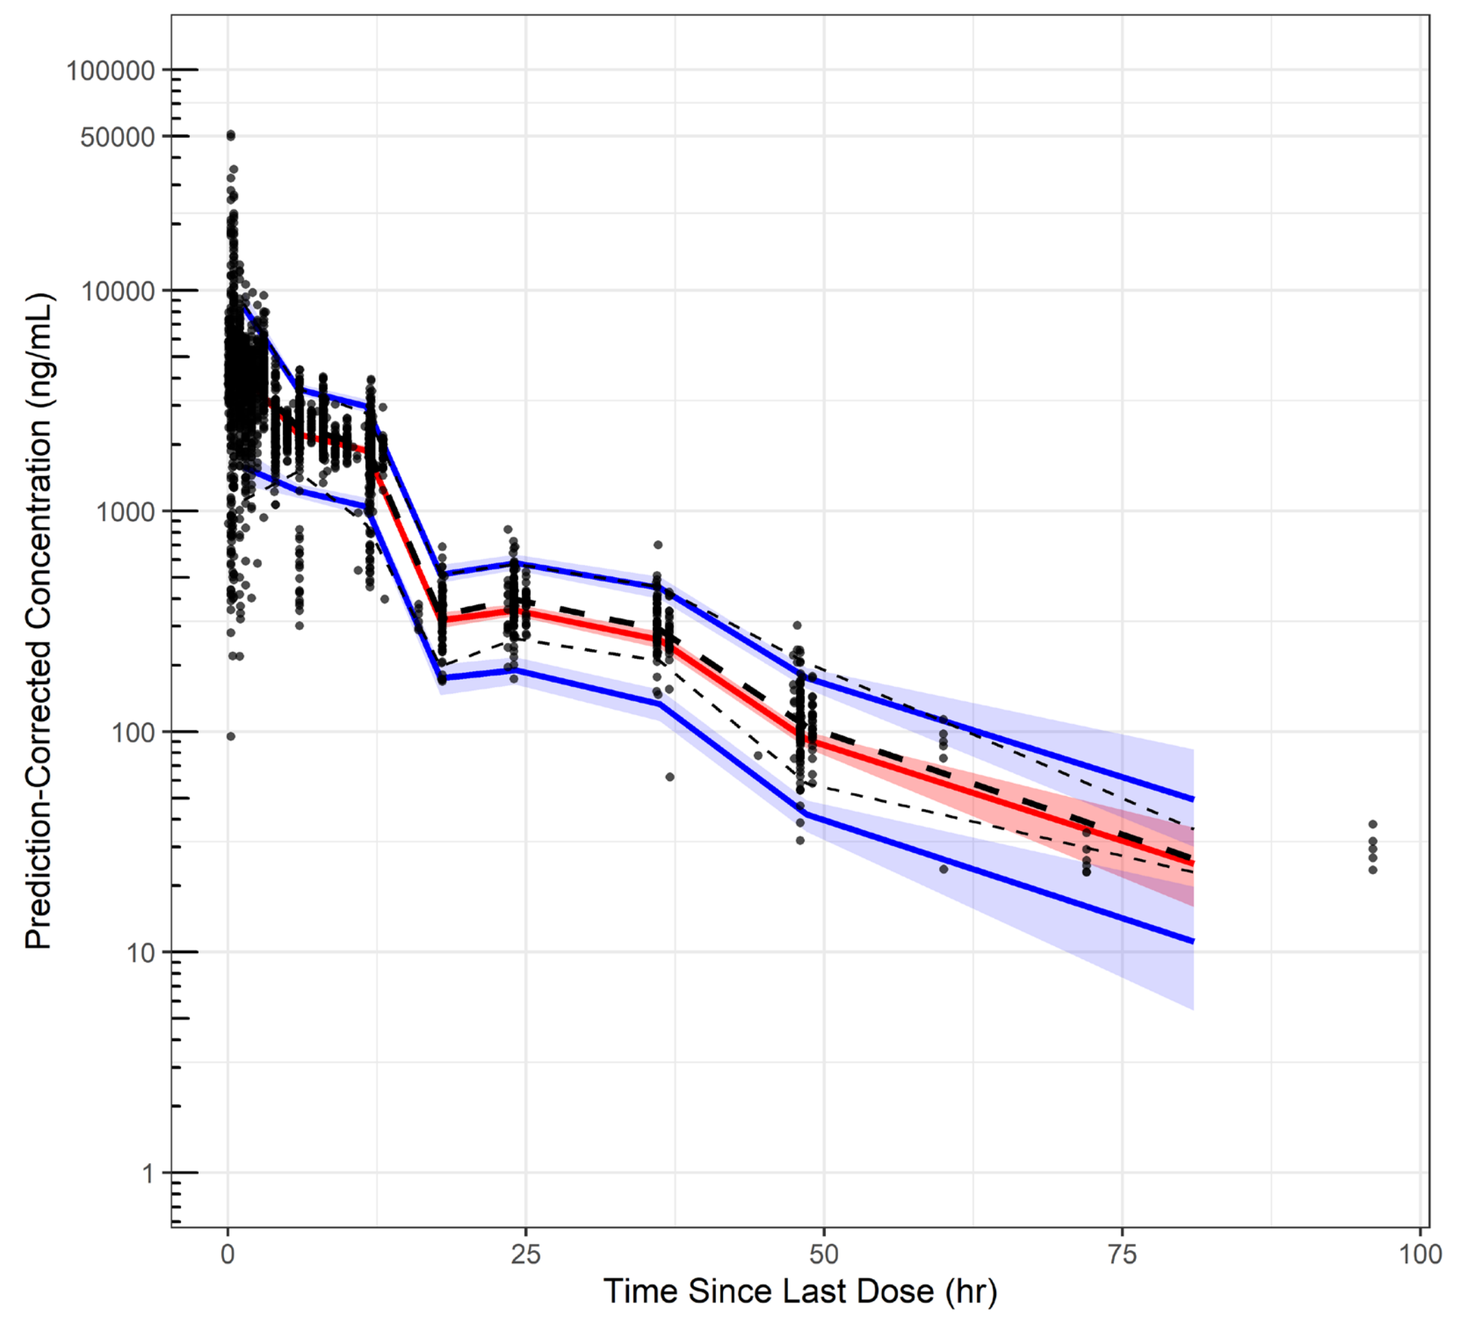
Prediction-Corrected Concentration (ng/mL)
Time Since Last Dose (hr)

Figure 3. Prediction-corrected visual predictive check plots for the application of the population PK model to the EBO plasma concentration-time data from the Phase 1b study conducted by AN2 (MAD EBO PO tablets only)

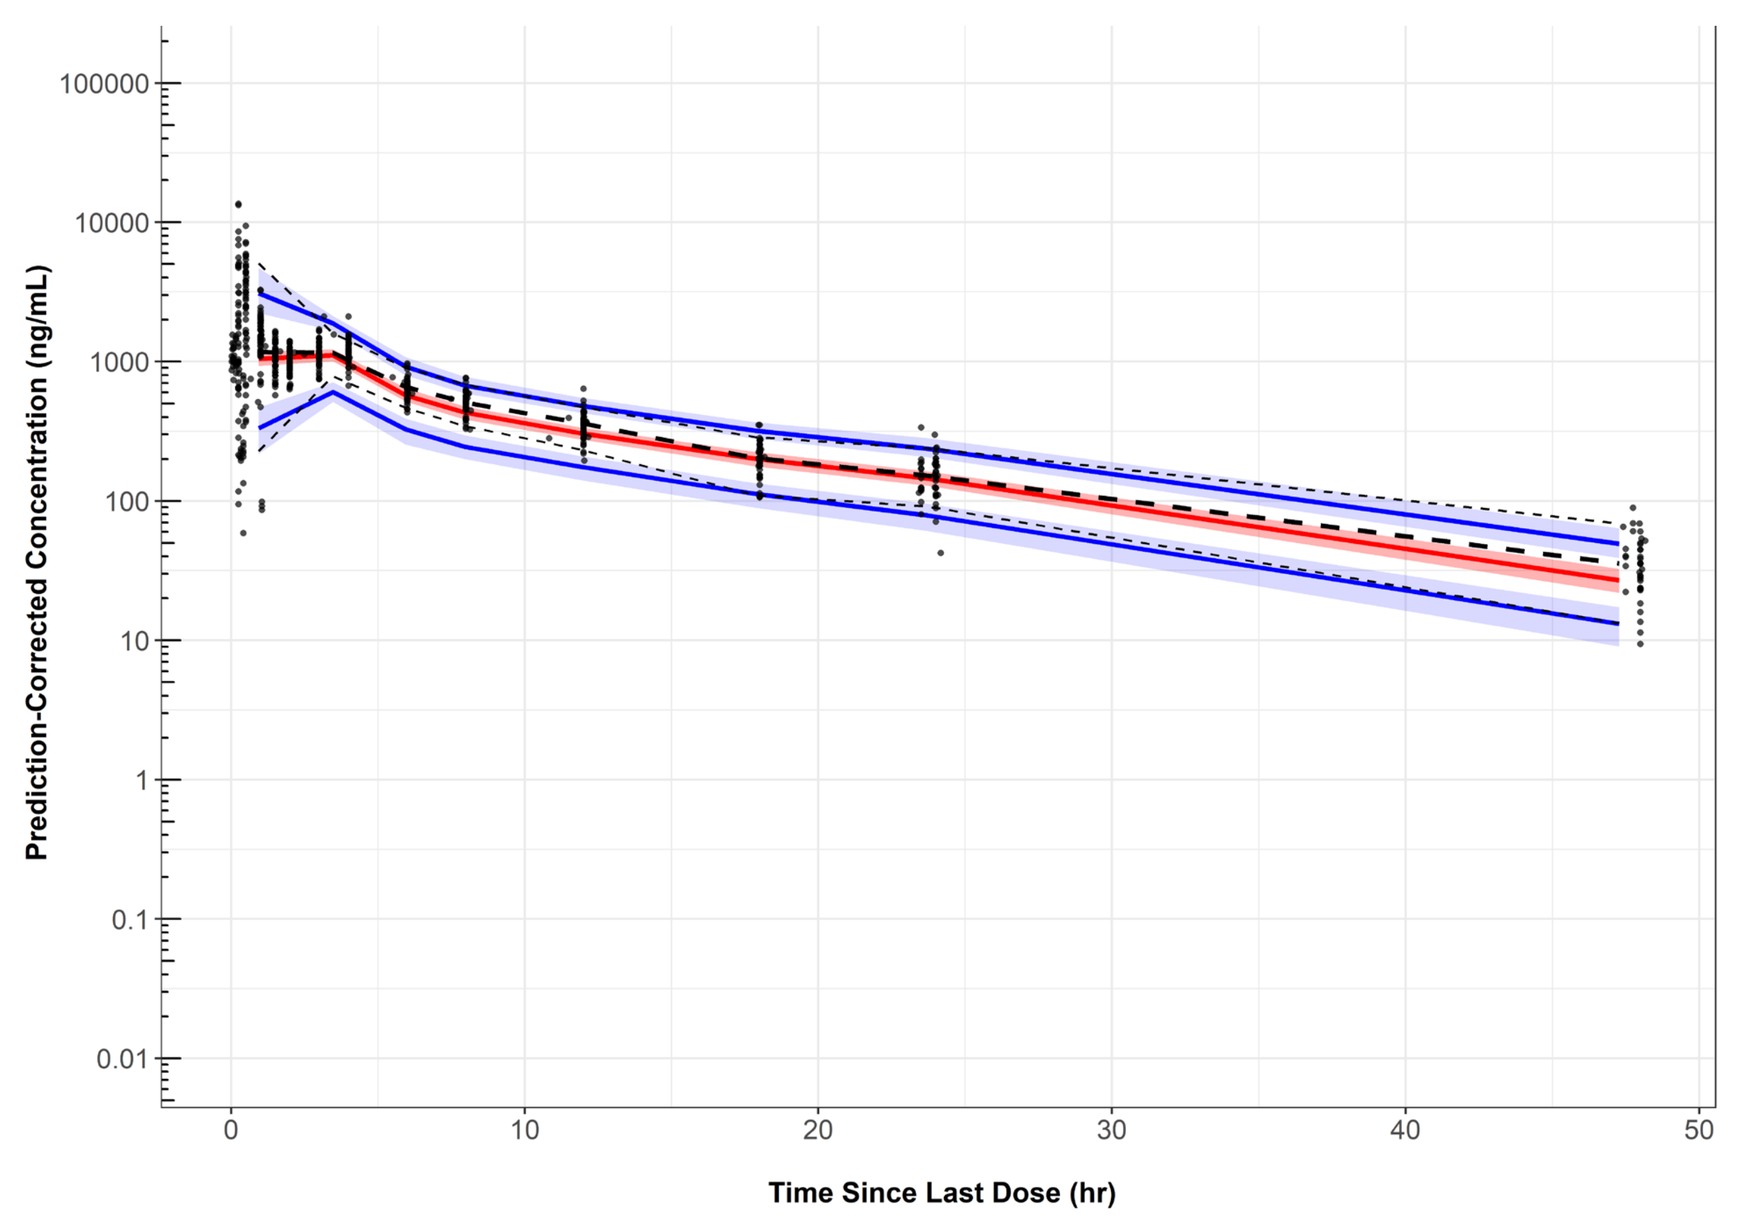
Prediction-Corrected Concentration (ng/mL)
Time Since Last Dose (hr)

Circles represent prediction-corrected plasma concentrations while the black lines represent the median and 5th and 95th percentiles of the observed data. The solid line represents the median model predicted concentration values. The solid blue lines represent the 5th and the 95th model predicted concentration values. Shaded areas represent the 95% model predicted confidence interval around the 5th, median, and 95th model predictors.

- While there is a slight tendency to underpredict the lowest observed concentrations (i.e., the lower 5th percentile of the prediction intervals are below the 5th percentile of the observed data), the plots indicate that model-based simulations generally capture the observed data well such that the model can be considered qualified for the conduct of PK-PD target attainment simulations.
- The population PK model will be updated as new data become available to identify predictors of variability in PK such as body size or renal function.

CONCLUSIONS

This model is useful for describing expected PK in MAC lung disease patients and was used to conduct simulations for the advancement of the oral EBO 500 mg q24h dosing regimen into clinical studies in patients with MAC lung disease.

ACKNOWLEDGEMENTS

This study was funded by AN2 Therapeutics, Menlo Park, CA.

REFERENCES

1. Ganapathy US, Gengenbacher M, Dick T. Epetraborole is active against *Mycobacterium abscessus*. Antimicrob Agents Chemother 2021;65:e01156-21.
2. Hernandez V, Crepin T, Palencia A, *et al.* Discovery of a novel class of boron-based antibacterials with activity against Gram-negative bacteria. Antimicrob Agents Chemother 2013;57:1394-1403.
3. Sullivan JR, Lupien A, Kalthoff E, *et al.* Efficacy of epetraborole against *Mycobacterium abscessus* is increased with norvaline. PLoS Pathog 2021;17(10):e1009965.
4. Kim T, Hanh BTB, Heo B, *et al*. A screening of the MMV pandemic response box reveals epetraborole as a new potent inhibitor against *Mycobacterium abscessus*. Int J Mol Sci 2021;22:5936.
5. Tenero D, Bowers G, Rodvold KA, *et al.* Intrapulmonary pharmacokinetics of GSK2251052 in healthy volunteers. Antimicrob Agents Chemother 2013;57; 3334-3339.

Contact: Christopher M. Rubino, Pharm.D., Institute for Clinical Pharmacodynamics, Inc., Email: crubino@icpd.com